16

Impact of Oral Sebetralstat on Anxiety Associated with Hereditary Angioedema Attacks in the Phase 3 KONFIDENT Trial

Timothy J. Craig,[1] Emel Aygören-Pürsün,[2] Jonathan A. Bernstein,[3] Paula J. Busse,[4] Teresa Caballero,[5] Danny M. Cohn,[6] Mar Guilarte,[7] Henriette Farkas,[8] Douglas H. Jones,[9] Sorena Kiani-Alikhan,[10] Michael E. Manning,[11] Marcus Maurer,[12] Marc A. Riedl,[13] Sinisa Savic,[14] H. James Wedner,[15] Patrick F. K. Yong,[16] Andrea Zanichelli,[17,18] Erik Hansen,[19] James Hao,[19] Michael D. Smith,[19] Christopher M. Yea,[19] Paul K. Audhya,[19] William R. Lumry[20]

[1]Departments of Medicine, Pediatrics, Ob/Gyn MFM and Biomedical Sciences, Pennsylvania State University, Hershey, PA, USA; Vinmec International Hospital, Times City, Hanoi, Vietnam; [2]University Hospital Frankfurt, Goethe University Frankfurt, Frankfurt, Germany; [3]University of Cincinnati College of Medicine, and Bernstein Clinical Research Center, Cincinnati, OH, USA; [4]Department of Medicine, Icahn School of Medicine at Mount Sinai School of Medicine, New York, NY, USA; [5]Servicio de Alergia, Hospital La Paz Health Research Institute, Biomedical Research Network on Rare Diseases, Madrid, Spain; [6]Amsterdam University Medical Center, University of Amsterdam, Amsterdam, Netherlands; [7]Department of Allergy, Hospital Universitari Vall d'Hebron, Vall d'Hebron Research Institute (VHIR), Barcelona, Spain; [8]Hungarian Angioedema Center of Reference and Excellence, Semmelweis University, Budapest, Hungary; [9]Rocky Mountain Allergy, Asthma, and Immunology, Layton, UT, USA; [10]Division of Infection and Immunity, University College London, London, UK; [11]Medical Research of Arizona, Scottsdale, AZ, USA; and University of Arizona College of Medicine-Phoenix, Phoenix, AZ, USA; [12]Institute of Allergology, Charité-Universitätsmedizin Berlin, corporate member of Freie Universitätsmedizin Berlin and Humboldt-Universität zu Berlin, Berlin, Germany, and Fraunhofer Institute for Translational Medicine and Pharmacology ITMP, Immunology and Allergology, Berlin, Germany; [13]University of California, San Diego, La Jolla, CA, USA; [14]The Leeds Institute of Rheumatic and Musculoskeletal Medicine, University of Leeds, Leeds, UK; [15]John T. Milliken Department of Medicine, Washington University School of Medicine, St. Louis, MO, USA; [16]Department of Immunology, Frimley Health NHS Foundation Trust, Frimley, UK; [17]Operative Unit of Medicine, Angioedema Center, IRCCS Policlinico San Donato, San Donato Milanese, Milan, Italy; [18]Department of Biomedical Science for Health, University of Milan, Milan, Italy; [19]KalVista Pharmaceuticals, Salisbury, United Kingdom, and Cambridge, MA, USA; [20]AARA Research Center, Dallas, TX, USA

Introduction

- People living with hereditary angioedema with C1-inhibitor deficiency (HAE-C1INH) experience unpredictable, painful, and potentially life-threatening attacks that are associated with substantial psychological burden[1-3]
 - Due to the unpredictable onset and symptoms of HAE attacks, and the anxiety associated with parenteral treatments, including injection-site reactions or fear of needles, people living with HAE-C1INH reported delaying or withholding their on-demand treatment[4]
- Sebetralstat, an oral plasma kallikrein inhibitor for on-demand treatment of HAE attacks, met primary and key secondary endpoints in the phase 3, randomized, double-blind, placebo-controlled KONFIDENT (NCT05259917) trial and was well tolerated[5]
- The objective of this analysis of KONFIDENT was to assess the impact of sebetralstat on attack-related anxiety

Methods

Trial Design

- In KONFIDENT, participants with HAE-C1INH (≥12 years old) were randomly assigned to 1 of 6 sequences to treat ≤3 attacks with oral sebetralstat 300 mg, 600 mg, or placebo[5]
 - Participants reported the baseline attack severity at attack onset using the Patient Global Impression of Severity (PGI-S) scale, ranging from "None" to "Very Severe"
- Participants completed a modified Generalized Anxiety Numeric Rating Scale (GA-NRS; "How anxious do you feel right now?") from 0 (not at all anxious) to 10 (extremely anxious) at treatment administration and for 24 hours after to record levels of anxiety (**Figure 1**)

Figure 1. GA-NRS

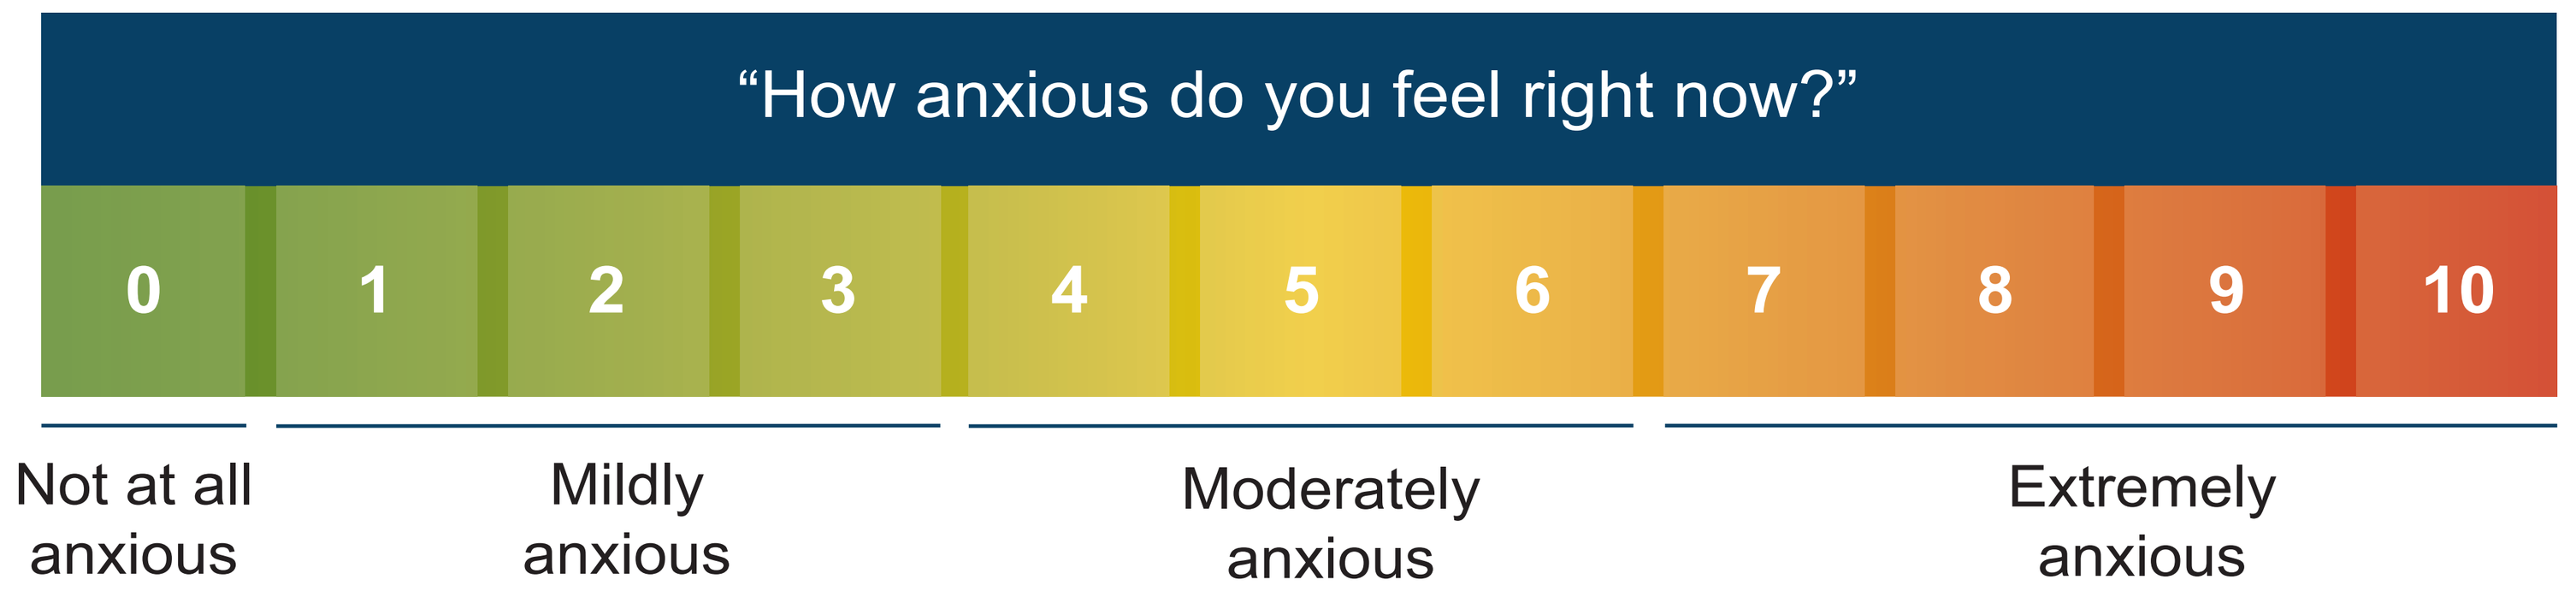
"How anxious do you feel right now?"
0 1 2 3 4 5 6 7 8 9 10
Not at all anxious
Mildly anxious
Moderately anxious
Extremely anxious

GA-NRS, Generalized Anxiety Numeric Rating Scale.

Prespecified Exploratory Endpoints

- Cumulative GA-NRS score, calculated as the area under the curve over 12 hours (AUC_{0-12}) or 24 hours (AUC_{0-24}) from treatment administration
- Least squares mean change from treatment administration at 4 hours and 12 hours post-treatment administration (**Figure 2**)
- Time to reductions of ≥2 points in GA-NRS score with a GA-NRS score of ≥2 at treatment administration

Figure 2. Anxiety Assessments

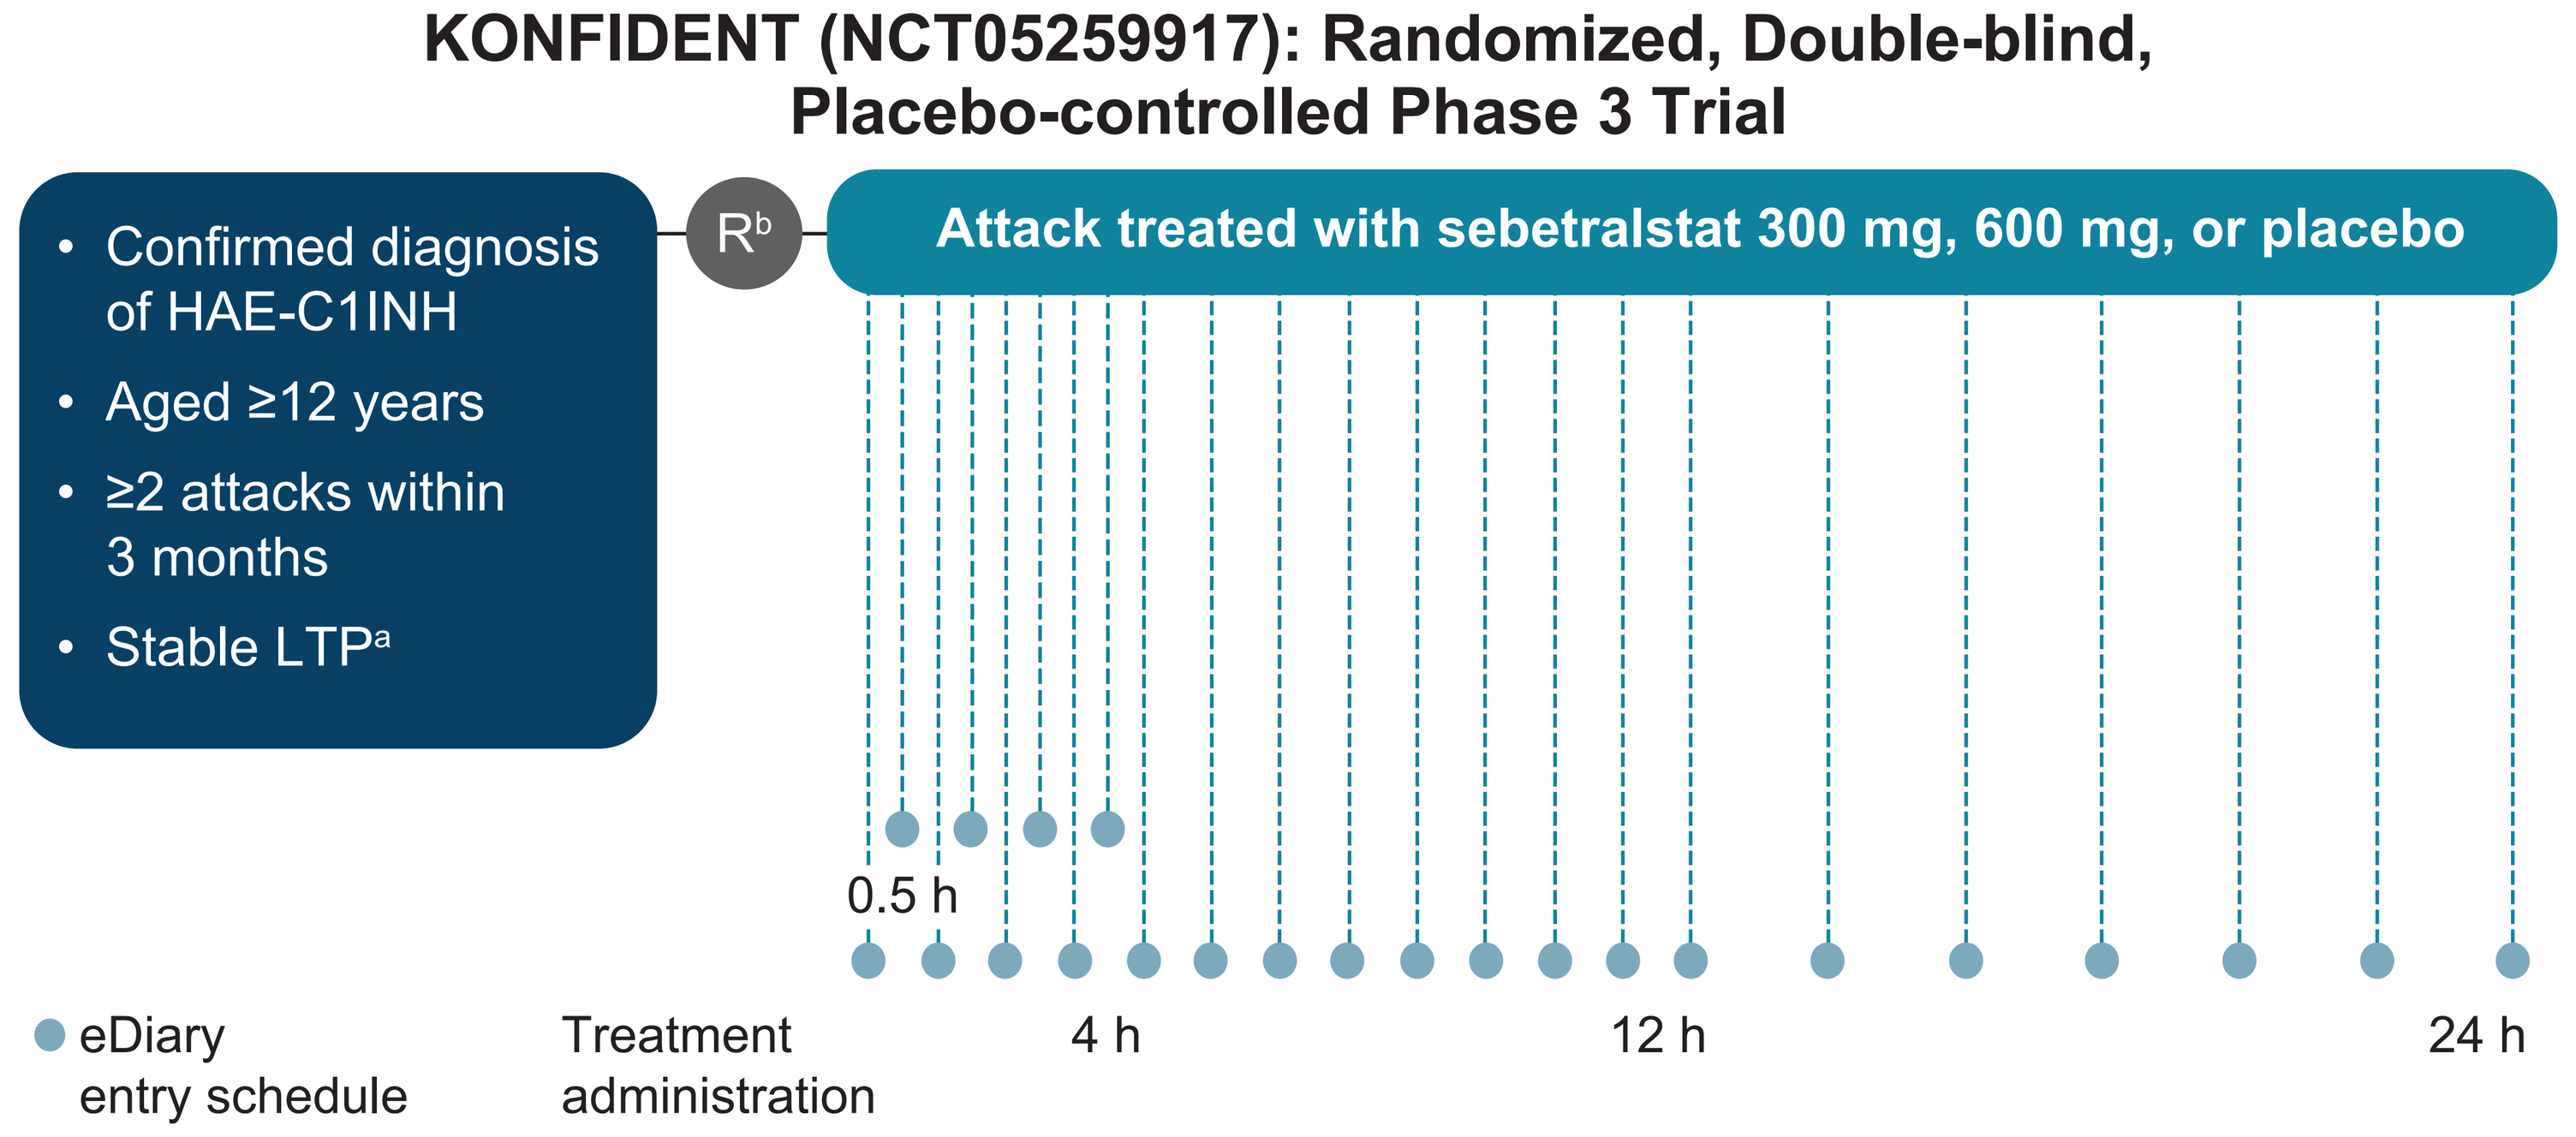
KONFIDENT (NCT05259917): Randomized, Double-blind, Placebo-controlled Phase 3 Trial
Confirmed diagnosis of HAE-C1INH
Aged ≥12 years
≥2 attacks within 3 months
Stable LTP[a]
R[b]
Attack treated with sebetralstat 300 mg, 600 mg, or placebo
0.5 h
4 h
12 h
24 h
eDiary entry schedule
Treatment administration

HAE-C1INH, hereditary angioedema due to C1 inhibitor deficiency; LTP, long-term prophylaxis, R, randomization.
[a]Participants receiving LTP were required to be on a stable dose for ≥3 months prior to screening.
[b]Participants were randomized 1:1:1:1:1:1 to a treatment sequence using a permuted-block method, stratified by the use of LTP at enrollment.

Statistical Considerations

- Outcomes were assessed in all attacks and attacks inducing moderate-to-extreme anxiety (GA-NRS score of 4-10 at treatment administration)

Results

Participants, Attacks, and Time to Treatment

Table 1. Participant Demographics and Disease Characteristics

	All participants[a] N=110
Age, median (IQR), years	39.5 (25.0-49.0)
Sex, female, n (%)	66 (60.0)
Race, n (%)	
White	92 (83.6)
Asian	10 (9.1)
Black or African American	1 (0.9)
Other or not reported	7 (6.4)
Time since HAE diagnosis, median (IQR), years	12 (7-22)
Medical history of anxiety, n (%)	8 (7.3)
Current treatment regimen, n (%)	
On-demand only	86 (78.2)
On-demand + LTP[b]	24 (21.8)

C1INH, C1-inhibitor; HAE, hereditary angioedema; IQR, interquartile range; LTP, long-term prophylaxis.
[a]Characteristics are reported for participants who experienced ≥1 treated attack.
[b]Of the 24 participants receiving LTP, 10 (42%) received berotralstat, 8 (33%) received lanadelumab, and 7 (29%) received C1INH.

- Factors associated with increased anxiety by Pearson Correlation included:
 - Female sex (vs male, *P*=0.0034)
 - Shorter time since HAE diagnosis (vs longer time, *P*=0.0014)
 - Moderate or severe/very severe baseline attack severity (moderate [vs mild, *P*=0.0030]; severe/very severe [vs mild, *P*<0.0001])

Table 2. Attack Characteristics

	All attacks[5] N=264	Attacks inducing moderate-to-extreme anxiety[a] n=115/261[b] (44%)
Baseline PGI-S category, n (%)[c]		
Mild[d]	115 (43.6)	38 (33.0)
Moderate	102 (38.6)	47 (40.9)
Severe/very severe	45 (17.0)	30 (26.1)
Baseline primary attack location, n (%)[c]		
Abdomen	114 (43.2)	57 (49.6)
Legs/feet	62 (23.5)	33 (28.7)
Arms/hands	76 (28.8)	30 (26.1)
Head/face/neck	29 (11.0)	12 (10.4)
Larynx/throat	8 (3.0)	4 (3.5)
Genitals	9 (3.4)	3 (2.6)
Time from onset to first administration, median (IQR), minutes	41 (6-140)	45 (6-155)

GA-NRS, Generalized Anxiety Numeric Rating Scale; IQR, interquartile range; PGI-S, Patient Global Impression of Severity.
[a]Moderate-to-extreme anxiety defined as a GA-NRS score of ≥4.
[b]Of 261 attacks for which there were GA-NRS ratings at treatment administration.
[c]The score on the PGI-S scale and attack location at baseline are missing for 2 attacks in the "all attacks" category.
[d]Includes 2 attacks in the "all attacks" category and 1 attack in the "attacks inducing moderate-to-extreme anxiety" category with a baseline PGI-S category of "None."

- Compared with placebo for all attacks, on-demand treatment with sebetralstat significantly reduced the AUC_{0-12} (300 mg, *P*=0.0040; 600 mg, *P*=0.0008) and AUC_{0-24} (300 mg, *P*=0.0220; 600 mg, *P*=0.0012; **Figure 3**)
 - When attacks inducing moderate-to-extreme anxiety were assessed, reductions in anxiety were greater than in all attacks

Reduction in Anxiety

Figure 3. Changes in Anxiety Over Time

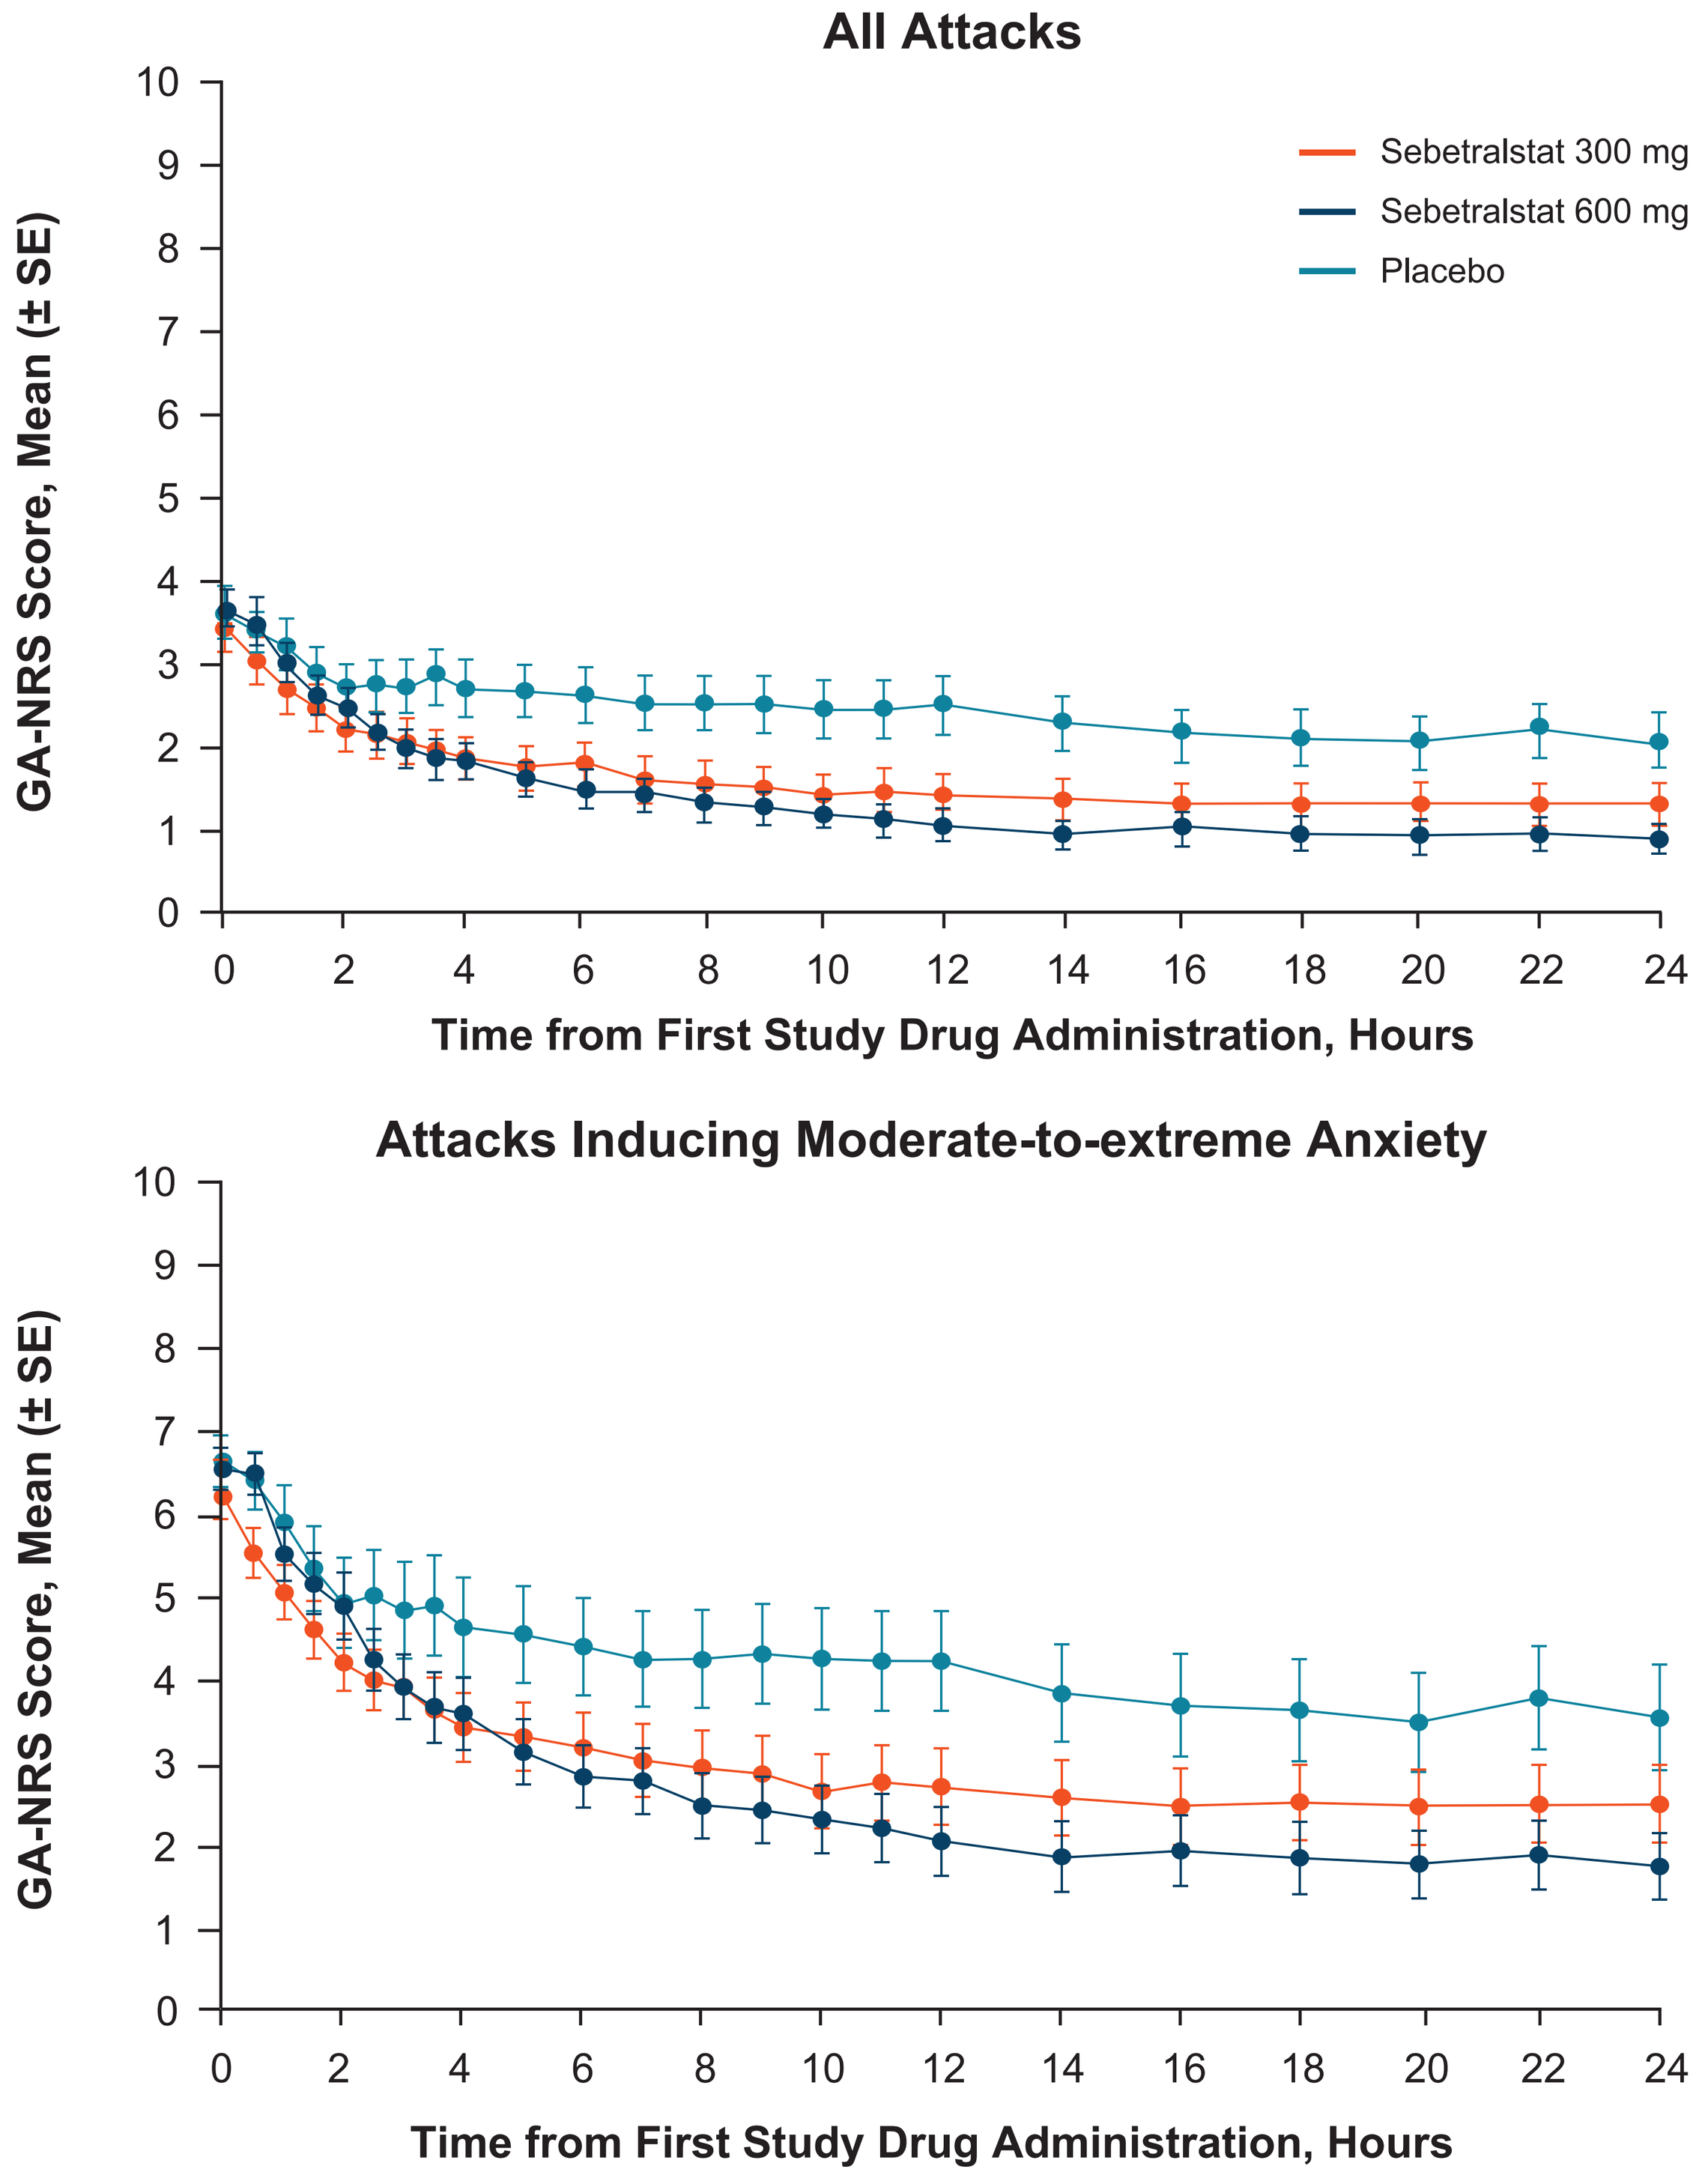
All Attacks
Sebetralstat 300 mg
Sebetralstat 600 mg
Placebo
GA-NRS Score, Mean (± SE)
Time from First Study Drug Administration, Hours
Attacks Inducing Moderate-to-extreme Anxiety
GA-NRS Score, Mean (± SE)
Time from First Study Drug Administration, Hours

GA-NRS, Generalized Anxiety Numeric Rating Scale; SE, standard error.

- Least squares mean change from treatment administration was more pronounced in attacks that induced moderate-to-extreme anxiety than with all attacks (**Figure 4**)

Figure 4. LSM Change from Treatment Administration in GA-NRS by Treatment

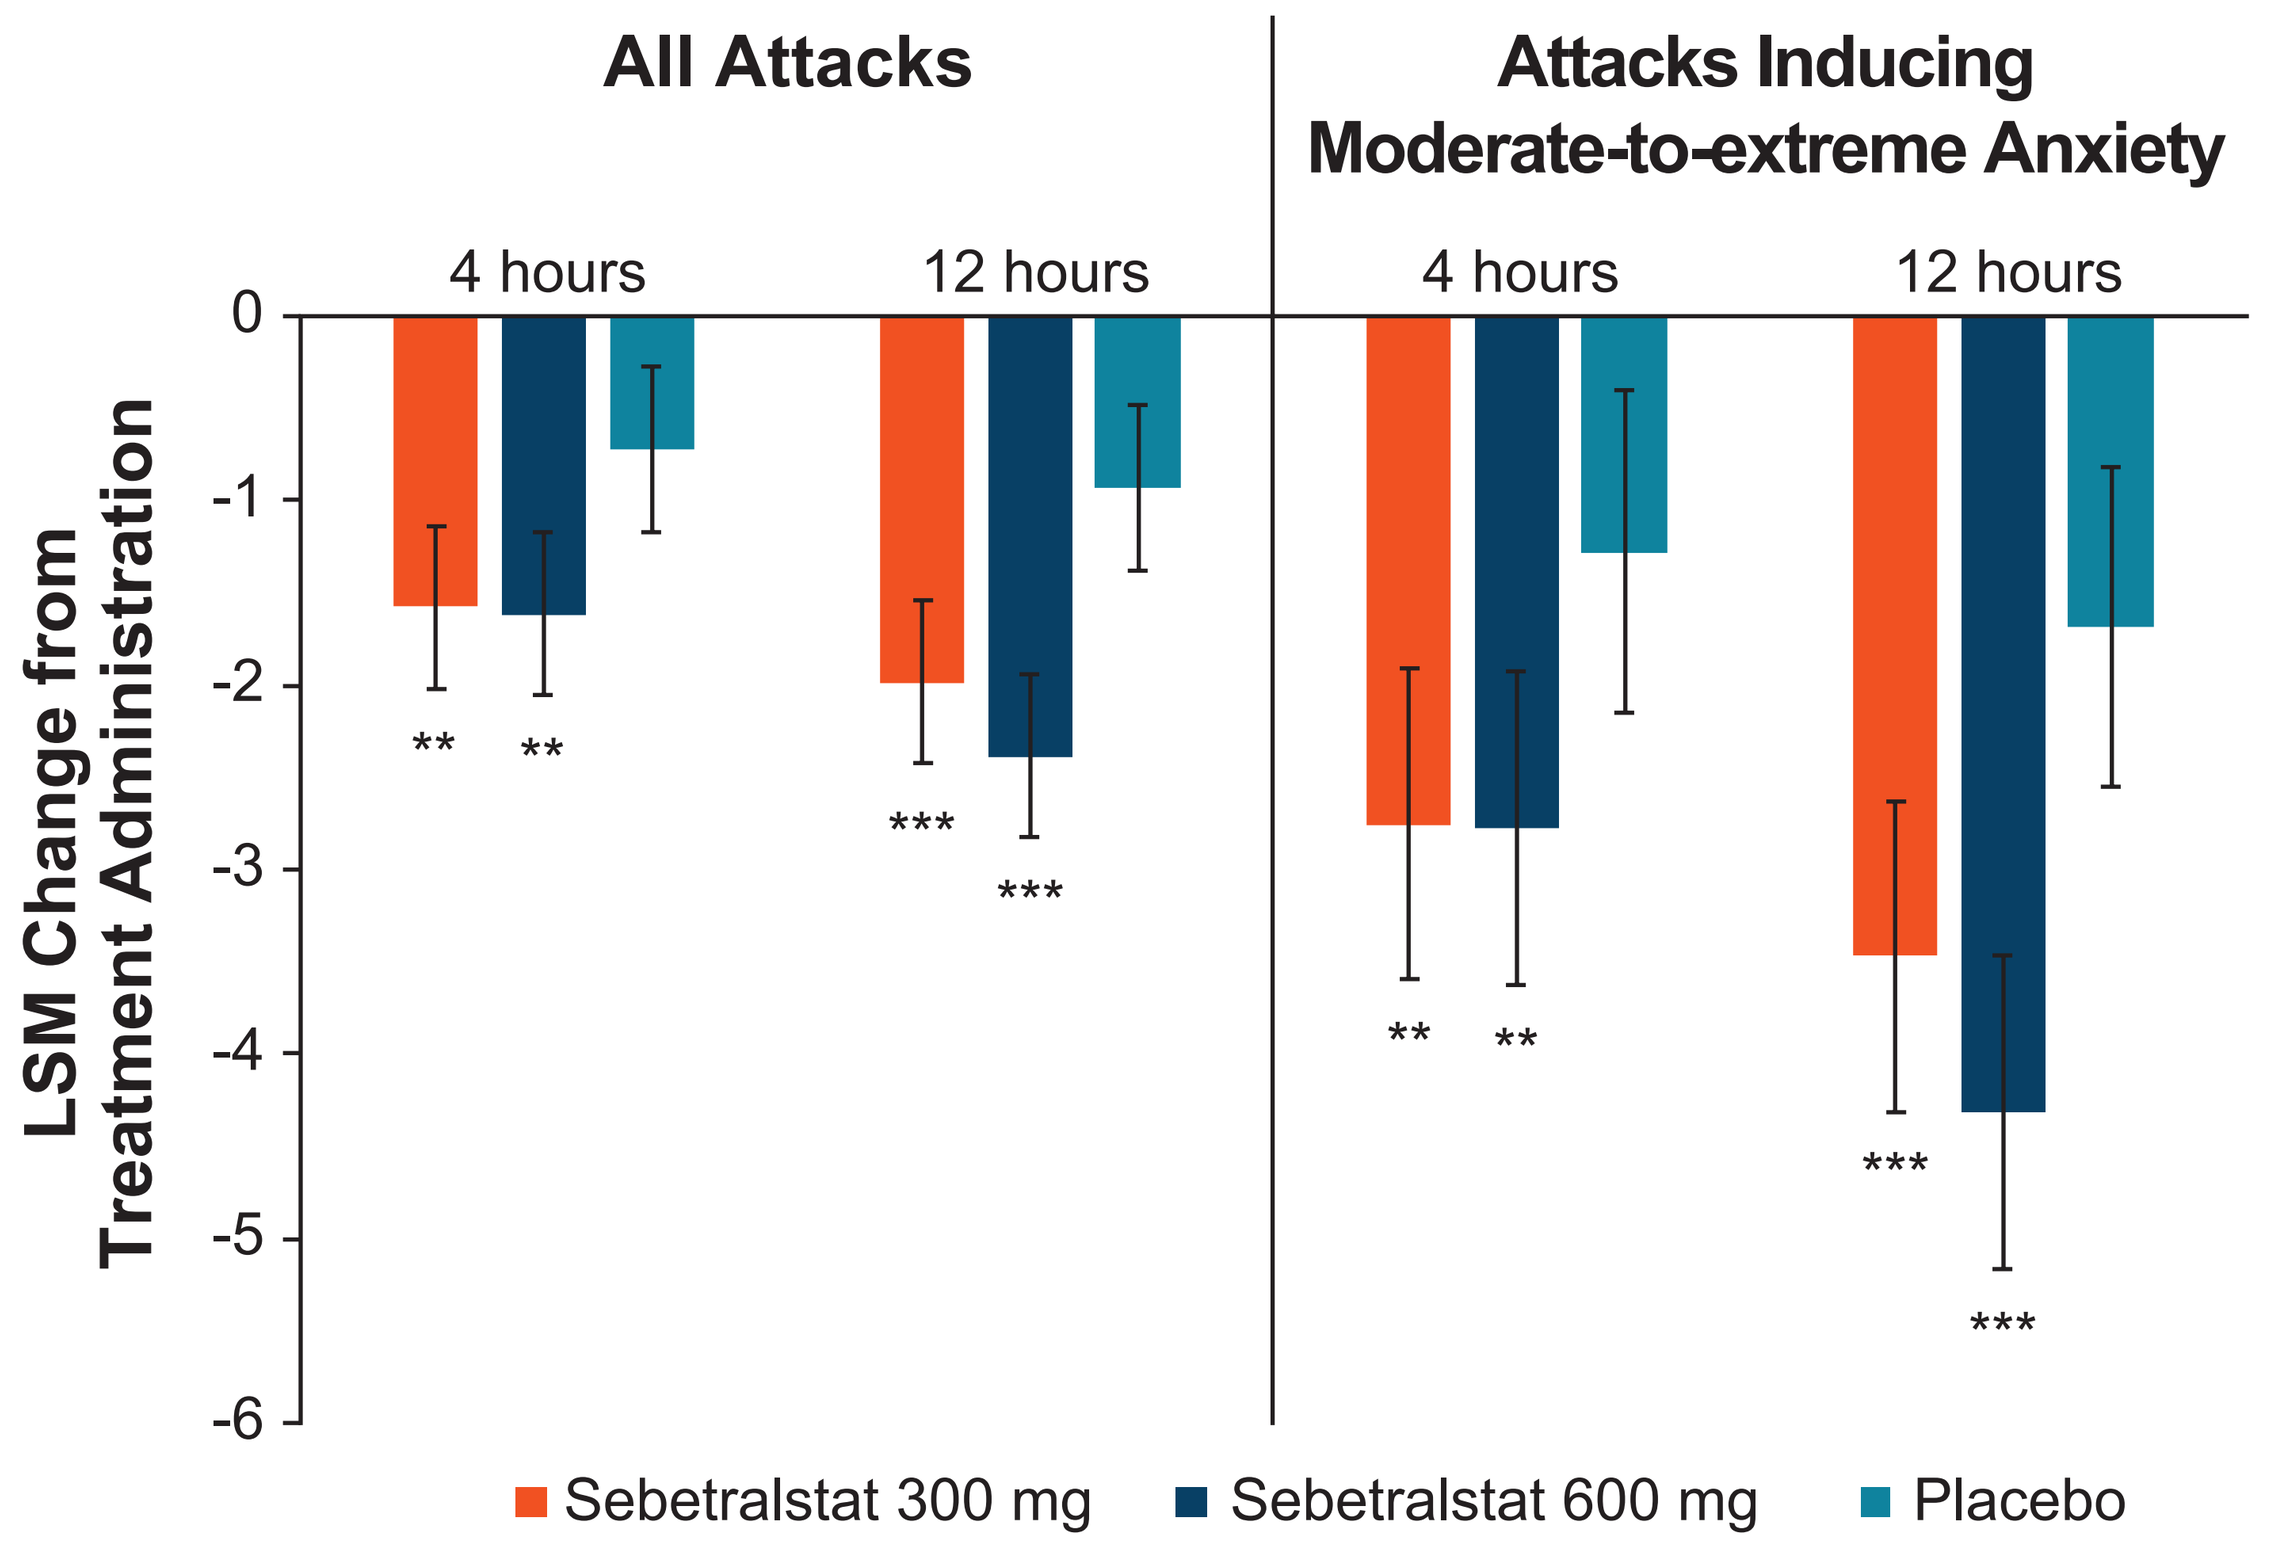
All Attacks
Attacks Inducing Moderate-to-extreme Anxiety
4 hours
12 hours
4 hours
12 hours
LSM Change from Treatment Administration
Sebetralstat 300 mg
Sebetralstat 600 mg
Placebo

*P<0.05, **P<0.01, ***P<0.001.
GA-NRS, Generalized Anxiety Numeric Rating Scale; LSM, least squares mean.

Agreement of Reduced Anxiety with Trial Endpoints

- The median time to a ≥2-point reduction in anxiety in KONFIDENT for attacks treated with sebetralstat 300 mg or 600 mg was 2.3 hours (IQR: 0.8 to 10.1 and 1.3 to 5.5, respectively) versus >12 hours for placebo (**Figure 5**)

Figure 5. Time to Reduction in Anxiety

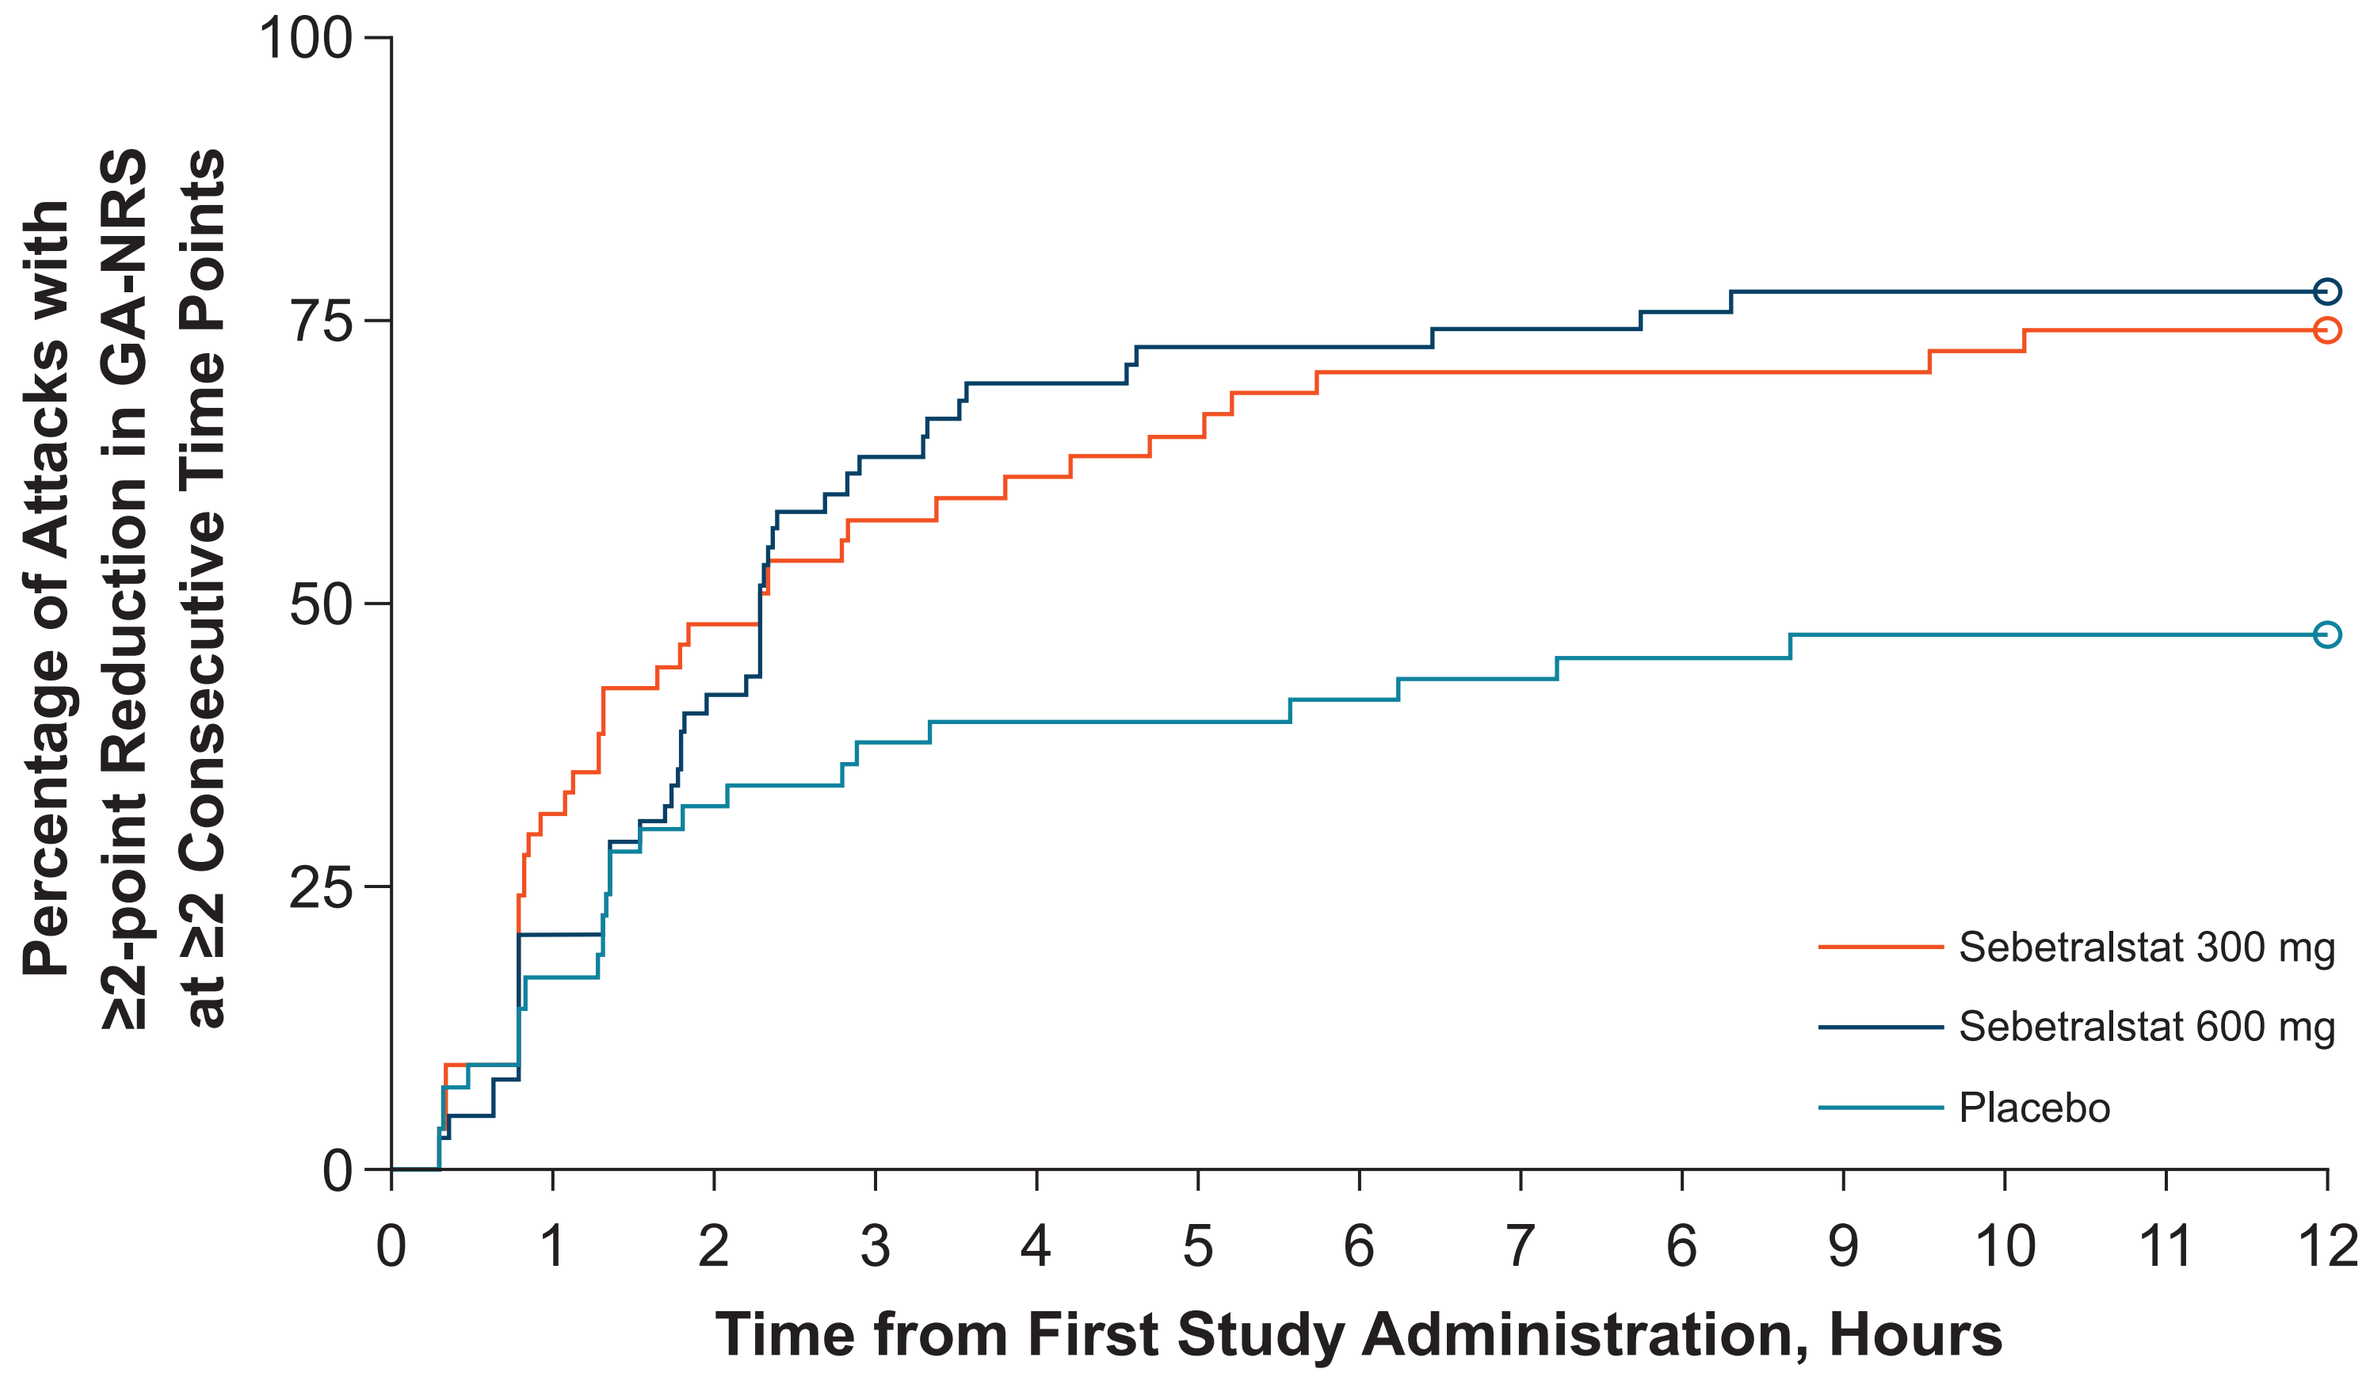
Percentage of Attacks with ≥2-point Reduction in GA-NRS at ≥2 Consecutive Time Points
Time from First Study Administration, Hours
Sebetralstat 300 mg
Sebetralstat 600 mg
Placebo

GA-NRS, Generalized Anxiety Numeric Rating Scale.

- The time to reduction in anxiety showed strong agreement with the primary and key secondary endpoints in KONFIDENT based on Cohen's kappa analysis
 - Primary endpoint: time to beginning of symptom relief within 12 hours (*P*<0.0001)
 - Key secondary endpoints: time to reduction in severity within 12 hours (*P*<0.0001); time to complete attack resolution within 24 hours (*P*<0.0001)

Discussion

- **In the phase 3 KONFIDENT trial, anxiety was rated as moderate to extreme at the time of treatment in >40% of attacks**
 - **In the absence of injectables, anxiety was correlated with baseline attack severity**
- **Compared with placebo, on-demand treatment with sebetralstat reduced anxiety, especially in attacks rated as inducing moderate-to-extreme anxiety**
- **The median time to reduction in anxiety was 2.3 hours with sebetralstat and >12 hours with placebo**
- **The time to reduction in anxiety showed strong agreement with the primary and key secondary endpoints in KONFIDENT**

References

1. Lumry WR et al. *Allergy*. 2021;76(4):1188-1198.
2. Hews-Girard J et al. *Allergy Asthma Clin Immunol*. 2021;17(1):61.
3. US Food and Drug Administration. The voice of the patient. Report Date: May 2018. https://www.fda.gov/media/113509/download. Accessed October 27, 2022.
4. Christiansen S et al. *Ann Allergy Asthma Immunol.* 2024;134(5):570-579.
5. Riedl M et al. *New Engl J Med*. 2024;391(1):32-43.

Acknowledgements

Medical writing support was provided under the guidance of the authors by Richard W. Davis IV, PhD, and Jennifer McKinney, PhD, of ApotheCom, San Francisco, CA, USA, and was funded by KalVista Pharmaceuticals.

Please visit the KalVista virtual medical booth after this presentation to view this poster.

Presented at the US Hereditary Angioedema Association (HAEA) National Summit; July 10-13, 2025; Baltimore, MD, USA

Copyright © 2025 KalVista Pharmaceuticals, Inc. All rights reserved.