Time to End of Progression of Hereditary Angioedema Attacks Treated with Sebetralstat

Daniel F. Soteres MD,[1] William R. Lumry MD,[2] John Anderson MD,[3] Jonathan A. Bernstein MD,[4] Mauro Cancian MD,[5] Danny M. Cohn MD,[6] Henriette Farkas,[7] H. Henry Li MD,[8] James Hao PhD,[9] Michael D. Smith PharmD,[9] Paolo Bajcic MD,[9] Paul K. Audhya MD,[9] Markus Magerl MD[10,11]

[1]Allergy and Asthma Associates PC, Colorado Springs, CO, USA; [2]AARA Research Center, Dallas, TX, USA; [3]Clinical Research Center of Alabama, an affiliate of AllerVie Health, Birmingham, AL, USA; [4]University of Cincinnati College of Medicine and Bernstein Clinical Research Center, Cincinnati, OH, USA; [5]Departmental Allergy Division, Department of Systems Medicine, University of Padua, Padua, Italy; [6]Department of Vascular Medicine, Amsterdam Cardiovascular Sciences, Amsterdam University Medical Center, University of Amsterdam, Amsterdam, Netherlands; [7]Hungarian Angioedema Center of Reference and Excellence, Department of Internal Medicine and Hematology, Semmelweis University, Budapest, Hungary; [8]Institute for Asthma and Allergy, Wheaton, MD, USA; [9]KalVista Pharmaceuticals, Salisbury, United Kingdom, and Cambridge, MA USA; [10]Institute of Allergology, Charité-Universitätsmedizin Berlin, corporate member of Freie Universitätsmedizin Berlin and Humboldt-Universität zu Berlin, Germany; [10]Fraunhofer Institute for Translational Medicine and Pharmacology ITMP, Immunology and Allergology, Berlin, Germany

Background

- Global treatment guidelines recommend that people living with hereditary angioedema (HAE) treat attacks with on-demand medication as early as possible after recognizing the onset[1,2]
 - Early treatment is indicated to arrest progression of swelling, thereby reducing severity and morbidity, limit potential spread of the HAE attack from the initial site, shorten total attack duration, and reduce disruption in daily activities[1,2] (**Figure 1**)
- Currently, all approved on-demand therapies require injections and are associated with delays, which may lead to progression and therefore increased symptom burden[3-9]
- Sebetralstat is an oral plasma kallikrein inhibitor being evaluated for on-demand treatment of HAE attacks[10]
 - In pharmacologic studies, sebetralstat achieved rapid plasma exposure and near-complete inhibition of plasma kallikrein within 15 minutes in patients with HAE[9]

Figure 1. Impact of early versus late treatment on HAE attack burden

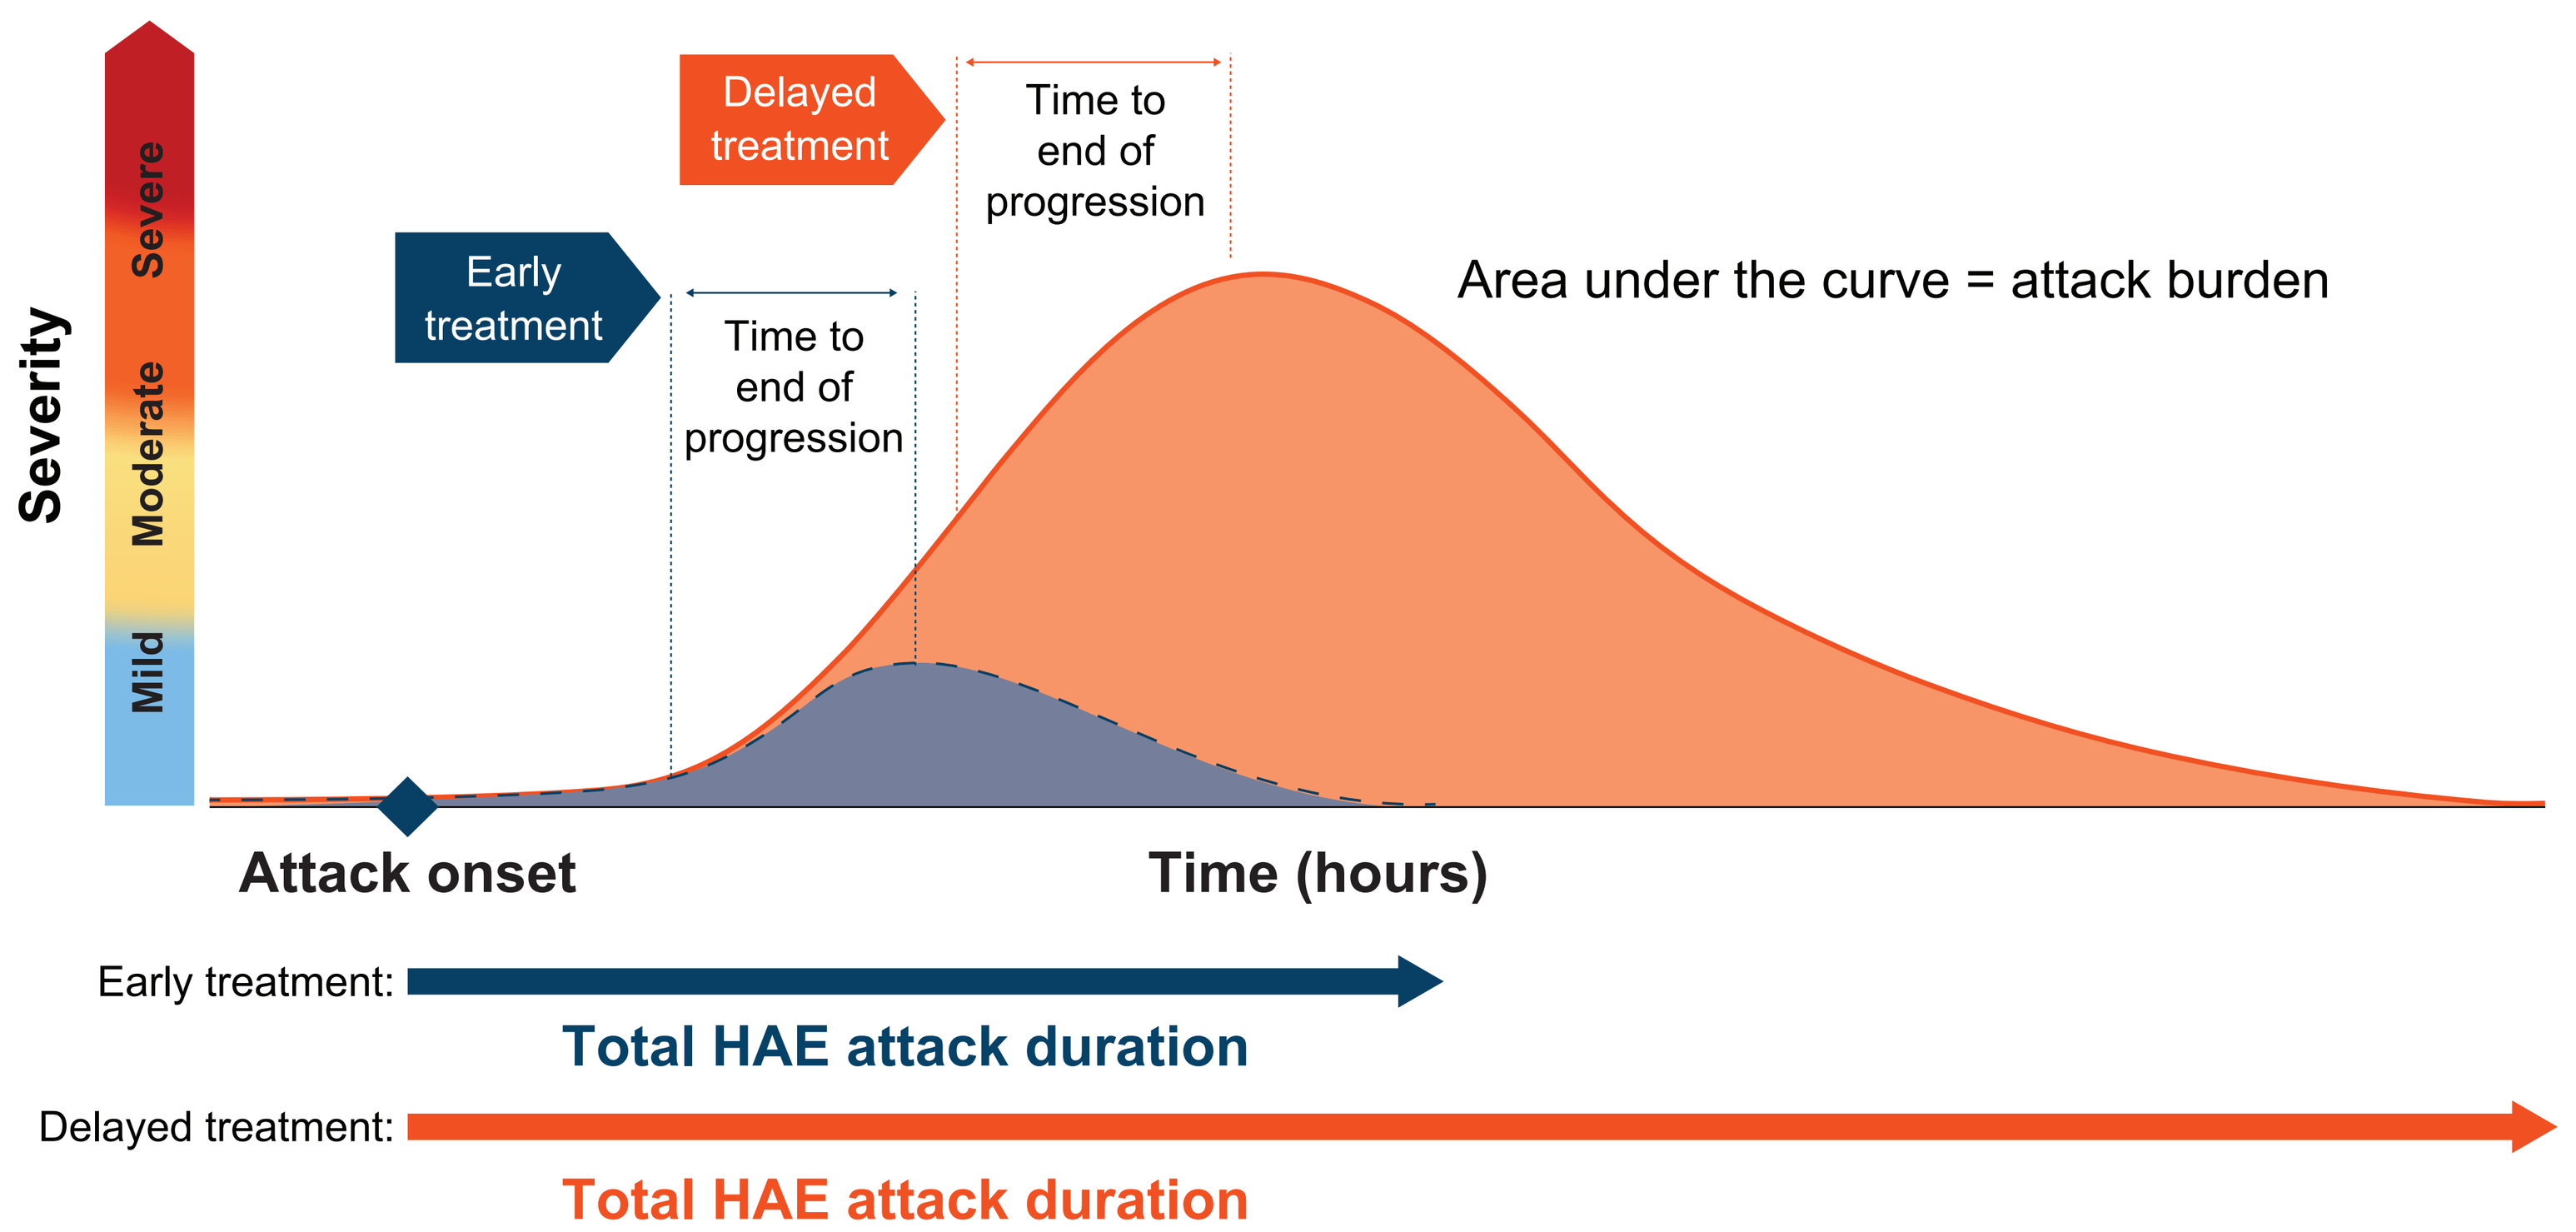

Delayed treatment
Time to end of progression
Early treatment
Time to end of progression
Area under the curve = attack burden
Severity
Severe
Moderate
Mild
Attack onset
Time (hours)
Early treatment:
Total HAE attack duration
Delayed treatment:
Total HAE attack duration

HAE, hereditary angioedema.

Objective

- This post hoc analysis examined the time to end of HAE attack progression following treatment with sebetralstat in the phase 3 KONFIDENT trial and KONFIDENT-S open-label extension (OLE) study

Methods

Study Design

- KONFIDENT (NCT05259917) was a phase 3, double-blind, randomized, placebo-controlled, 3-way crossover trial[10]
 - Adults and adolescents (≥12 years old) with HAE due to C1-inhibitor deficiency (HAE-C1INH) and ≥2 documented HAE attacks within 3 months were randomly assigned to 1 of 6 treatment sequences in which 3 eligible HAE attacks were treated with 1 to 2 doses of sebetralstat 300 mg, sebetralstat 600 mg, or placebo
- KONFIDENT-S is an ongoing multicenter OLE study (NCT05505916)
 - Adults and adolescents (≥12 years old) with HAE-C1INH and ≥2 documented HAE attacks within 3 months or who completed the phase 3 KONFIDENT trial self-administered sebetralstat 600 mg (2 × 300-mg tablets) for each HAE attack as early as possible after onset
- In both KONFIDENT and KONFIDENT-S, Patient Global Impression of Severity (PGI-S) ratings from "None" to "Very Severe" were recorded at the time of treatment, every 0.5 hours during the first 4 hours after taking the trial agent, and every hour from 5 to 12 hours

Effectiveness Endpoint

- End of progression was analyzed post hoc as the time at which the worst HAE attack severity was recorded within 4 hours on the PGI-S scale for attacks treated with sebetralstat
 - HAE attacks treated with conventional therapy were censored at the time of use
 - HAE attacks with no post baseline assessments were excluded (except for attacks treated with conventional therapy)

Results

Participants and HAE Attacks

- Overall, data from 110 participants from the KONFIDENT trial and 134 participants from the KONFIDENT-S study (**Table 1**) who treated 172 and 1591 HAE attacks with sebetralstat (**Table 2**), respectively, were analyzed post hoc

Table 1. Demographics and disease characteristics

	KONFIDENT[10] N=110	KONFIDENT-S N=134
Age, mean (range), years	39.5 (25.0 to 49.0)	35.0 (21.0 to 49.0)
Sex, female, n (%)	66 (60.0)	87 (64.9)
Race, n (%)		
White	92 (83.6)	99 (73.9)
Asian	10 (9.1)	17 (12.7)
Black or African American	1 (0.9)	0
Other or not reported[a]	7 (6.4)	18 (13.4)
BMI, mean (range), kg/m^2	26.2 (22.8 to 31.7)	25.4 (22.3 to 30.6)
HAE-C1INH-Type1, n (%)	101 (91.8)	125 (93.3)
Treatment regimen, LTP + on-demand, n (%)	24 (21.8)	35 (26.1)

BMI, body-mass index; HAE-C1INH, hereditary angioedema due to C1 inhibitor deficiency; LTP, long-term prophylaxis; N/n, number of participants.
[a]Includes 1 participant in KONFIDENT-S who identified as "Multiple."
Data cutoff date was January 31, 2024 for KONFIDENT and September 14, 2024 for KONFIDENT-S.

Table 2. Sebetralstat-treated HAE attack characteristics

	KONFIDENT Sebetralstat 300 mg n=84[a]	KONFIDENT Sebetralstat 600 mg n=88[a]	KONFIDENT-S Sebetralstat 600 mg n=1591[a]
HAE attack location, n (%)[b]			
Subcutaneous	48 (57.1)	49 (55.7)	930 (58.5)
Mucosal	35 (41.7)	39 (44.3)	650 (40.9)
Abdominal	32 (38.1)	35 (39.8)	603 (37.9)
Laryngeal	1 (1.2)	1 (1.1)	30 (1.9)
Baseline PGI-S rating, n (%)[c,d]			
Mild	36 (42.9)	40 (45.5)	594 (37.3)
Moderate	33 (39.3)	30 (34.1)	668 (42.0)
Severe	12 (14.3)	16 (18.2)	262 (16.5)
Very severe	2 (2.4)	2 (2.3)	58 (3.6)
Time to treatment, median (IQR), min	36.0 (7.0-131.0)	38.5 (4.5-141.0)	14.5 (1.0-76.0)

IQR, interquartile range; n, number of attacks; PGI-S, Patient Global Impression of Severity.
[a]Number of HAE attacks with a baseline PGI-S score KONFIDENT: sebetralstat 300 mg n=83; sebetralstat 600 mg n=88. KONFIDENT-S: sebetralstat 600 mg n=1582.
[b]Baseline attack location was missing for 1 attack (1.2%) treated with sebetralstat 300 mg in KONFIDENT and 11 attacks (0.7%) treated with sebetralstat 600 mg in KONFIDENT-S.
[c]PGI-S rating of "None" included in "Mild": KONFIDENT sebetralstat 300 mg, 0; KONFIDENT sebetralstat 600 mg, 0; KONFIDENT-S sebetralstat 600 mg, 23 (1.4%).
[d]PGI-S missing: KONFIDENT sebetralstat 300 mg, 1 (1.2%); KONFIDENT sebetralstat 600 mg, 0; KONFIDENT-S sebetralstat 600 mg, 9 (0.6%).
Data cutoff date was January 31, 2024 for KONFIDENT and September 14, 2024 for KONFIDENT-S.

Effectiveness

- The median time to end of progression was 19.2 minutes (IQR, 16.8 to 46.2) with sebetralstat 300 mg and 19.8 minutes (IQR, 16.8 to 97.2) with sebetralstat 600 mg in the KONFIDENT trial; in the KONFIDENT-S study, the median time was 19.8 minutes (IQR, 16.2 to 42.6) with sebetralstat 600 mg (**Figure 2**)
- In total, 69 (82.1%) HAE attacks treated with sebetralstat 300 mg, 79 (89.8%) treated with sebetralstat 600 mg in KONFIDENT, and 1437 (90.3%) treated with sebetralstat 600 mg in KONFIDENT-S reached end of progression within 4 hours

Figure 2. Time to end of progression

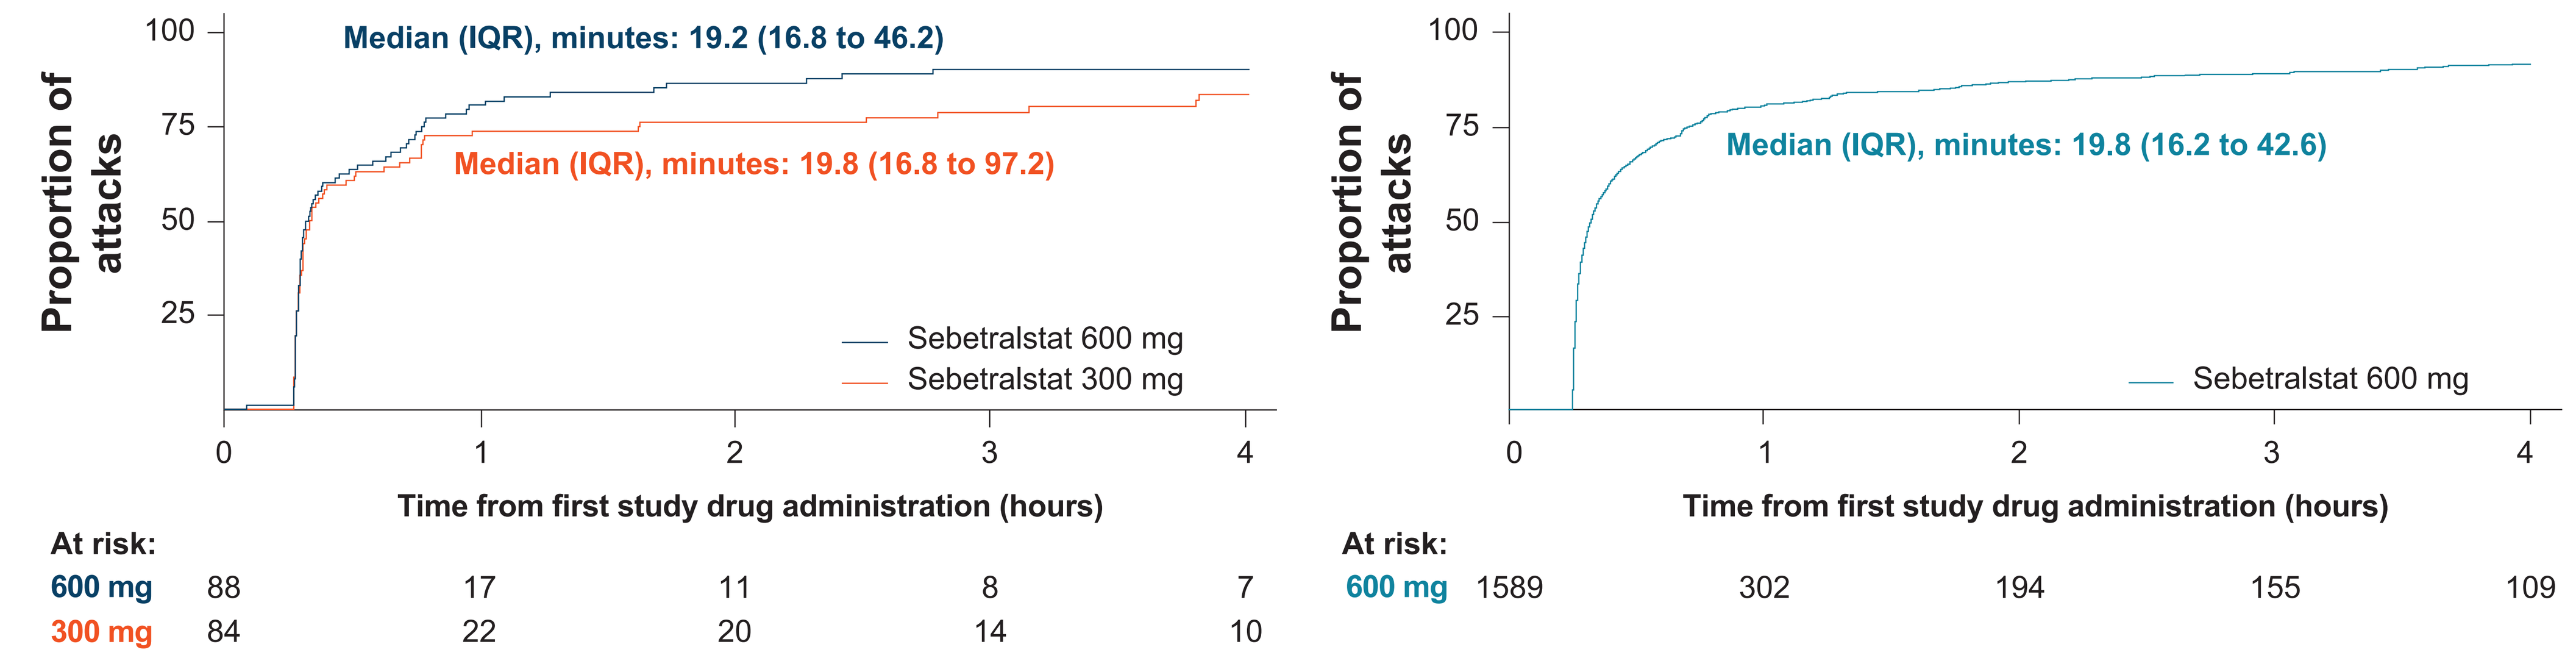

Median (IQR), minutes: 19.2 (16.8 to 46.2)
Median (IQR), minutes: 19.8 (16.8 to 97.2)
Proportion of attacks
100
75
50
25
0
1
2
3
4
Sebetralstat 600 mg
Sebetralstat 300 mg
Time from first study drug administration (hours)
At risk:
600 mg 88 17 11 8 7
300 mg 84 22 20 14 10
Median (IQR), minutes: 19.8 (16.2 to 42.6)
Sebetralstat 600 mg
At risk:
600 mg 1589 302 194 155 109

IQR, interquartile range; PGI-S, Patient Global Impression of Severity.
Time to end of progression was defined as the time at which the worst HAE attack severity was recorded using the 5-point PGI-S scale within 4 hours. Data cutoff date was January 31, 2024 for KONFIDENT and September 14, 2024 for KONFIDENT-S.

Figure 3. Time course of sebetralstat-treated HAE attacks (illustrative)

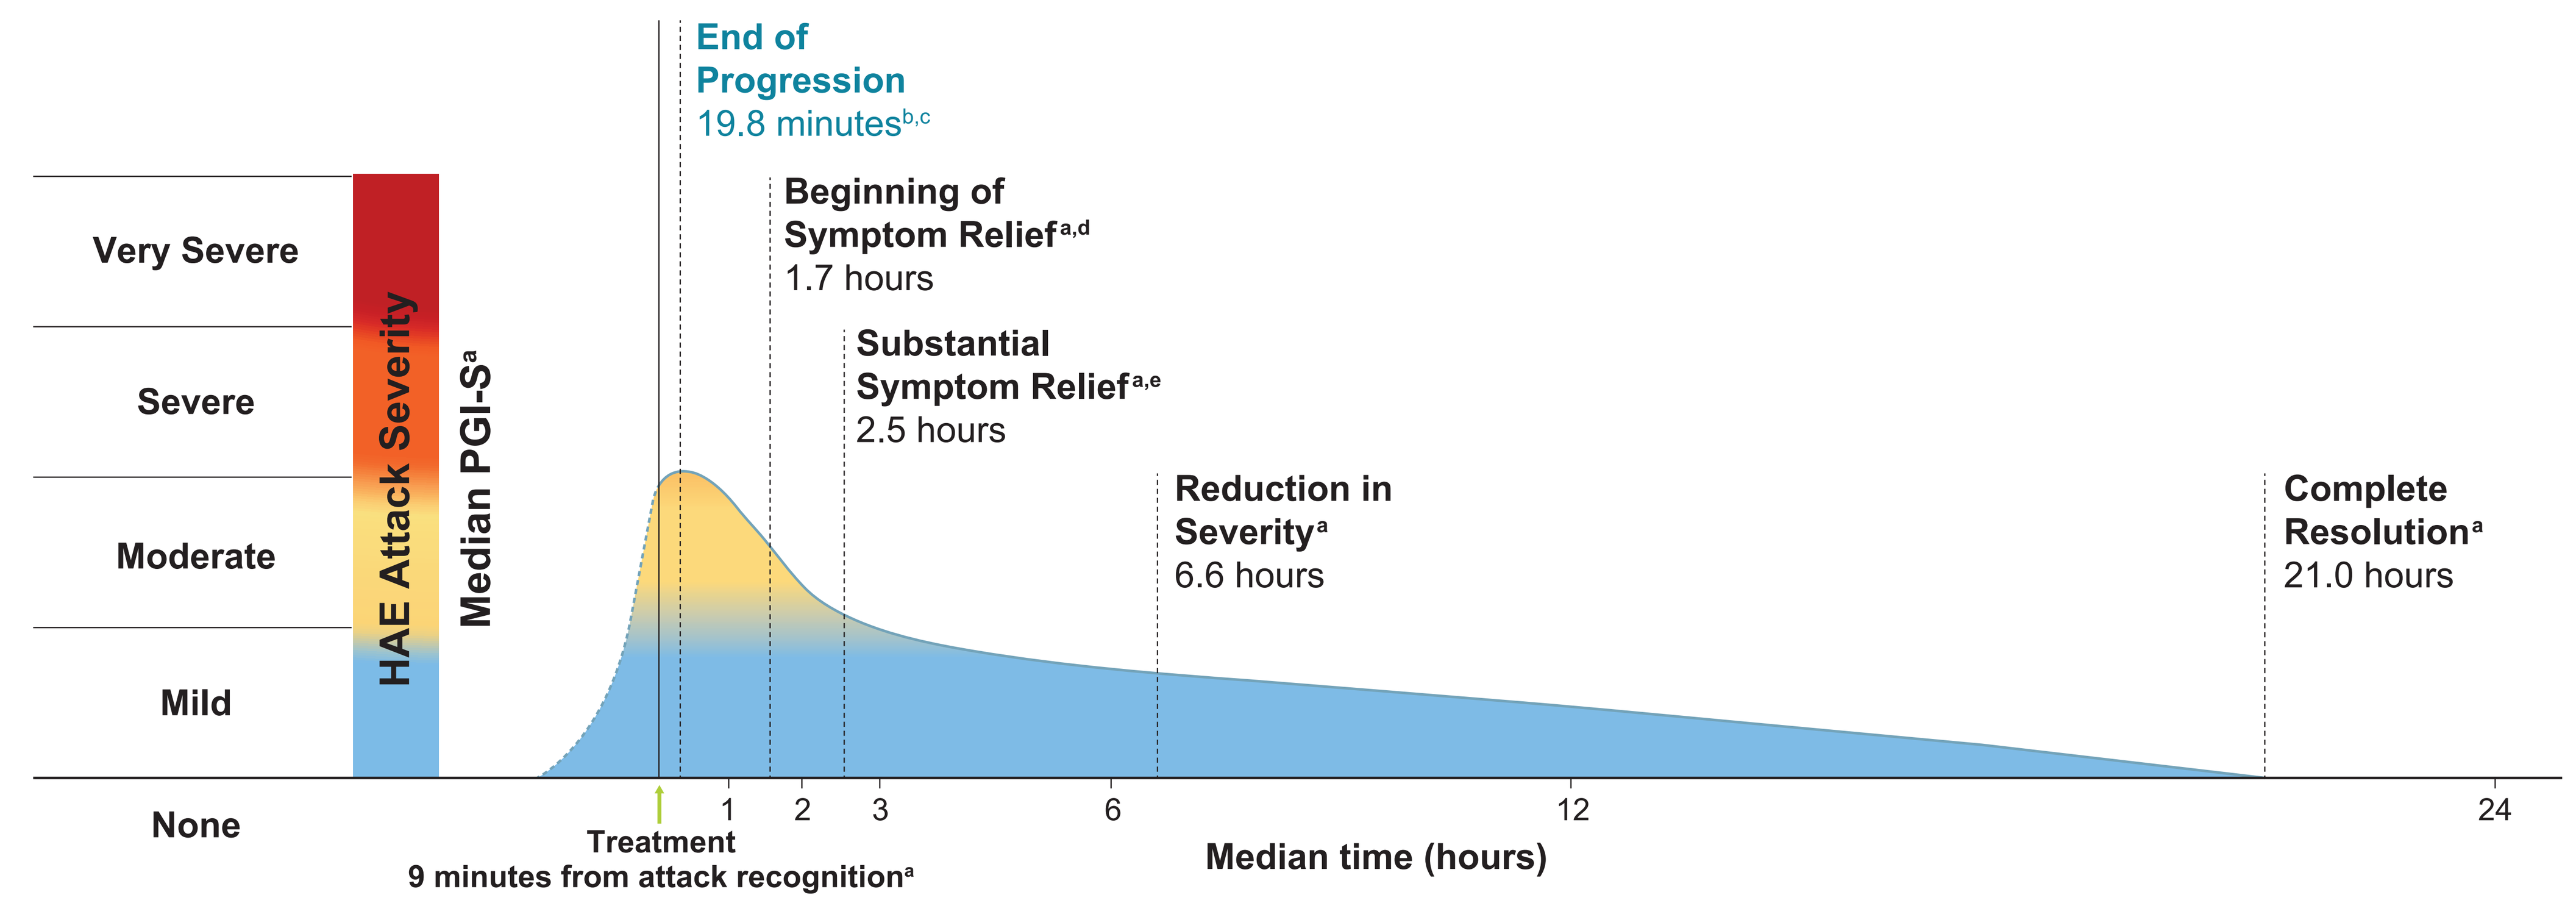

End of Progression
19.8 minutes[b,c]
Beginning of Symptom Relief[a,d]
1.7 hours
Substantial Symptom Relief[a,e]
2.5 hours
Reduction in Severity[a]
6.6 hours
Complete Resolution[a]
21.0 hours
Very Severe
Severe
Moderate
Mild
None
HAE Attack Severity
Median PGI-S[a]
1
2
3
6
12
24
Treatment
9 minutes from attack recognition[a]
Median time (hours)

HAE, hereditary angioedema; OLE, open-label extension; PGI-C, Patient Global Impression of Change; PGI-S, Patient Global Impression of Severity.
[a]Based on data cutoff date of January 31, 2024 (640 attacks treated with sebetralstat 600 mg).[12]
[b]Based on data cutoff date of September 14, 2024 (1706 attacks).
[c]Time to end of progression was defined as the time at which the worst HAE attack severity was recorded on the PGI-S scale within 4 hours.
[d]Defined as a PGI-C rating of at least "A Little Better" for at least 2 consecutive time points (with missing data entries in between) within 12 hours.
[e]Defined as time to substantial reduction of symptom burden (PGI-S of "Mild" for 2 consecutive time points) within 24 hours.
Graph generated based on median time to endpoints as observed in the KONFIDENT-S OLE study. Area under the curve represents the HAE attack burden.

Conclusions

- **Results of this post hoc analysis suggested that sebetralstat ended the progression of the HAE attack early, with a median of 19.8 minutes in KONFIDENT-S**
 - **These results are consistent with those observed with sebetralstat 300 mg and sebetralstat 600 mg in the phase 3 KONFIDENT trial**
- **The time to end of HAE attack progression closely followed the expected time to near-complete inhibition of plasma kallikrein (15 minutes) based on prior pharmacodynamic studies[11]**
- **Oral sebetralstat allowed for rapid treatment and halted HAE attack progression early in patients with HAE**

References

1. Busse PJ et al. *J Allergy Clin Immunol Pract*. 2021;9(1):132-150.
2. Maurer M et al. *Allergy*. 2022;77(7):1061-1990.
3. Maurer M et al. *PLoS One*. 2013;8(2):e53773.
4. Mendivil J et al. *Orphanet J Rare Dis*. 2021;16(1):94.
5. Gower RG et al. *Allergy Asthma Clin Immunol*. 2021;17(1):100.
6. Beyaz S et al. *Allergy Asthma Proc*. 2022;43(2):148-154.
7. Maurer M et al. *Allergy Asthma Proc*. 2014;35(5):377-381.
8. Betschel SD et al. *Allergy Asthma Clin Immunol*. 2024;20:43.
9. Christiansen S et al. *Ann Allergy Asthma Immunol*. 2024;S1081-1206(24)01732-0.
10. Riedl M et al. *N Engl J Med*. 2024;391:32-43.
11. Aygören-Pürsün E et al. *Lancet*. 2023;401(10375):458-469.
12. Farkas H et al. Presented at: EAACI Annual Meeting; 31 May-3 June, 2024; Valencia, Spain.

Acknowledgments

Medical writing assistance was provided under the direction of the authors by Richard W. Davis IV, PhD, of ApotheCom (San Francisco, CA) and was supported by KalVista Pharmaceuticals. KalVista thanks the people living with HAE and their families, their advocates, and the investigator teams who supported KONFIDENT and KONFIDENT-S.

Please visit the KalVista virtual medical booth after the presentation to view this poster.

Presented at the Eastern Allergy Conference 2025; May 29-June 1, 2025; Palm Beach, FL, USA

Copyright © 2025 KalVista Pharmaceuticals, Inc. All rights reserved.