Poster ID: 26

Clinical Outcomes of Sebetralstat On-demand Treatment of Hereditary Angioedema in Pediatric (2-11 Years) Patients from Interim Analysis of KONFIDENT-KID

H. James Wedner,[1] Adil Adatia,[2] Emel Aygören-Pürsün,[3] Mauro Cancian,[4] Aharon Kessel,[5] H. Henry Li,[6] Heloise Reumaux,[7] Erik Hansen,[8] Ya-Hsiu Chuang,[8] Matthew Iverson,[8] Michael D. Smith,[8] Paul K. Audhya,[8] Bob Geng[9]

[1]John T. Milliken Department of Medicine, Washington University School of Medicine, St. Louis, MO, USA; [2]University of Alberta, Edmonton, AB, Canada; [3]University Hospital Frankfurt, Goethe University Frankfurt, Frankfurt, Germany; [4]Azienda Ospedale Università di Padova, Padova, Italy; [5]Bnai Zion Medical Center, Haifa, Israel; [6]Institute for Asthma and Allergy, Wheaton, MD, USA; [7]Hôpital Jeanne de Flandre, Lille, France; [8]KalVista Pharmaceuticals, Framingham, MA, USA; [9]University of California – San Diego, La Jolla, CA, USA.

Background

- Approximately 40% of patients with hereditary angioedema (HAE) experience their first attack by age of 5, with potential age of onset as early as infancy[1,2]
- Earlier onset is associated with more severe course of disease and increased attack burden[1]
 - Children with HAE attacks experience significant anxiety, social isolation, and academic disruption[2,3] and there is a multifaceted psychosocial impact on their caregivers[3-5]
- On-demand options for children with HAE require intravenous (IV) infusion or subcutaneous (SC) injection and are associated with substantial burden, including anxiety and pain, leading to treatment avoidance, denial or delays[5,6]
 - HAE in children (<12 years old) is primarily managed with on-demand treatment
 - In the US, only IV plasma-derived C1 inhibitor (pdC1INH) is approved for children[7]
 - In the EU, additional approved treatment options in children aged 2 years and older include IV recombinant human C1 inhibitor (rhC1INH), IV pdC1INH, and SC icatibant[8-10]
- Sebetralstat, an oral plasma kallikrein inhibitor, has recently been approved for on-demand treatment of HAE attacks in patients ≥12 years old in the US, EU and other countries[11-13]

Objective

- This open-label, multicenter phase 3 KONFIDENT-KID trial was designed to evaluate the safety, pharmacokinetics (PK), and efficacy of sebetralstat orally disintegrating tablets (ODT) in children with hereditary angioedema with C1INH deficiency (HAE-C1INH) who are 2-11 years old[14]

Methods

Trial Design

- The trial design of the phase 3 KONFIDENT-KID trial (NCT06467084) is shown in **Figure 1**

Figure 1. KONFIDENT-KID Trial Design

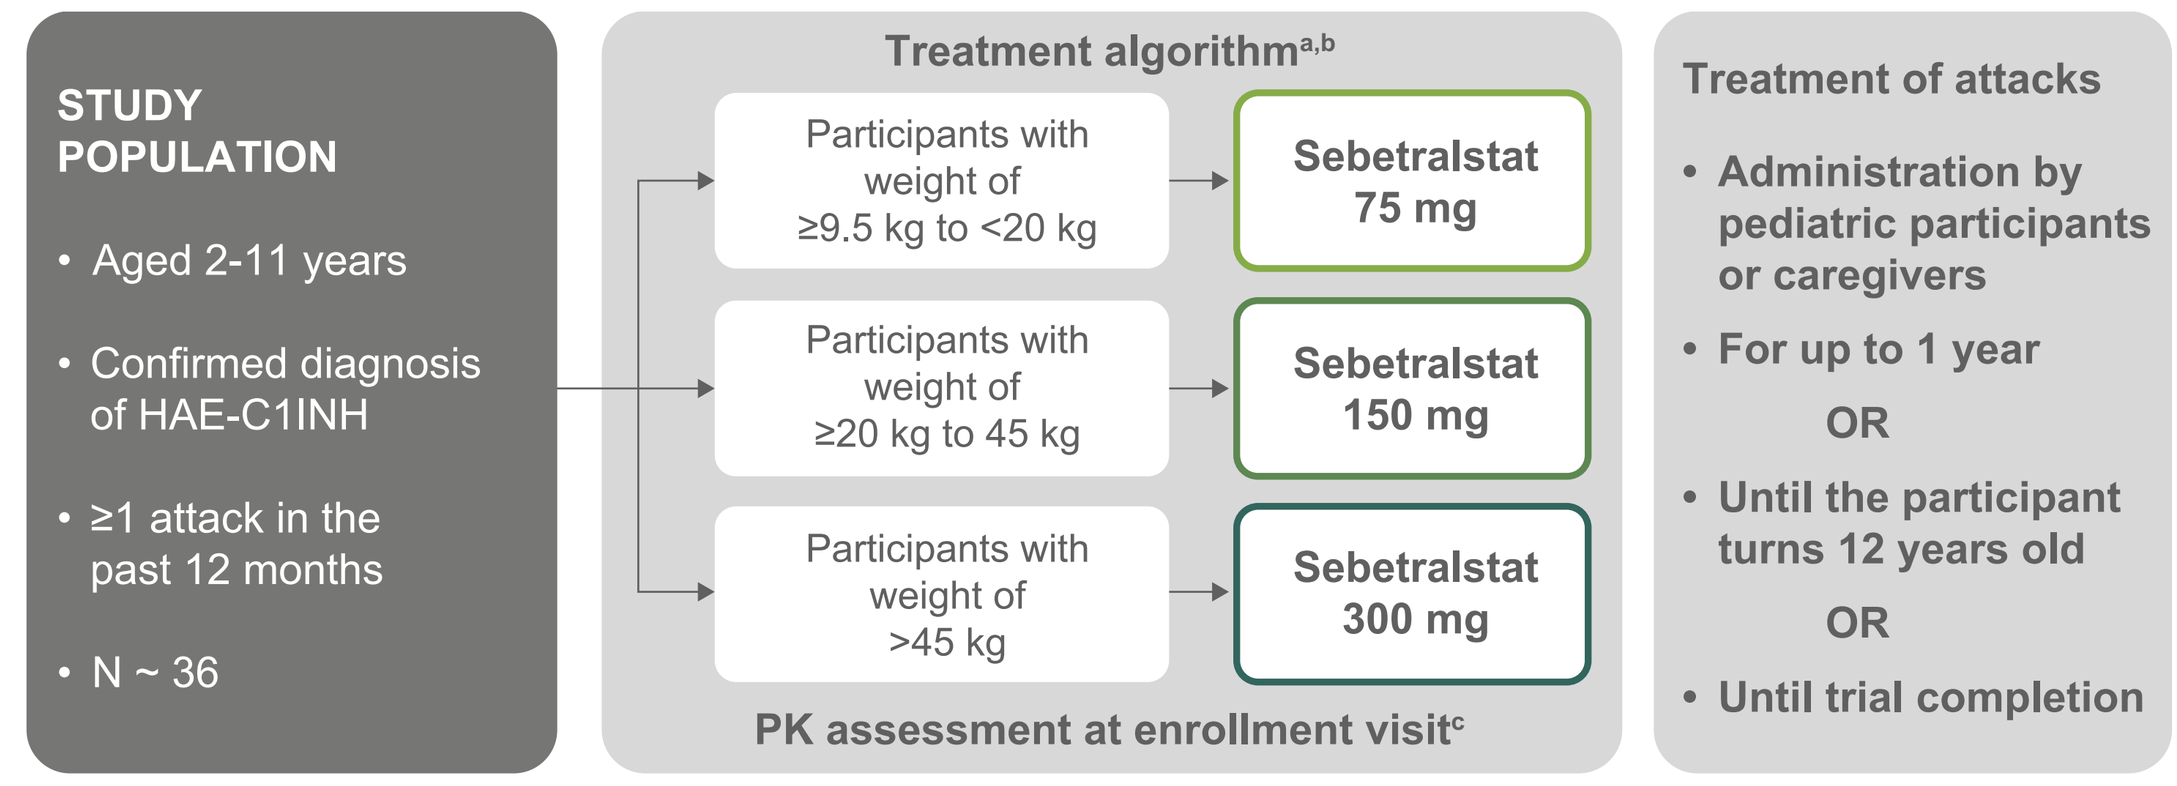

STUDY POPULATION
Aged 2-11 years
Confirmed diagnosis of HAE-C1INH
≥1 attack in the past 12 months
N ~ 36
Treatment algorithm[a,b]
Participants with weight of ≥9.5 kg to <20 kg
Sebetralstat 75 mg
Participants with weight of ≥20 kg to 45 kg
Sebetralstat 150 mg
Participants with weight of >45 kg
Sebetralstat 300 mg
PK assessment at enrollment visit[c]
Treatment of attacks
Administration by pediatric participants or caregivers
For up to 1 year
OR
Until the participant turns 12 years old
OR
Until trial completion

[a]All doses are equivalent to the 300-mg FCT dose in adults; provided as ODT.
[b]Trial was amended in the US to treat with the equivalent of sebetralstat 600 mg.
[c]PK samples were collected at 0.5, 2, and 4 hours. Simulated models were generated based on key parameters from each participant; 500 simulations were ran for each participant.
FCT, film-coated tablet; HAE-C1INH, hereditary angioedema with C1 inhibitor deficiency; ODT, orally disintegrating tablet; PK, pharmacokinetics.

Objectives and Endpoints

- The primary objective was to evaluate the safety of sebetralstat in this patient population
- Secondary objectives were to assess PK and efficacy
- Population pharmacokinetic (popPK) analyses included sebetralstat plasma concentrations from ten clinical trials
 - The final popPK model used to perform simulations was created from sex-specific age-matched body weights from NHANES 2017-2018
- Efficacy endpoints were assessed using Caregiver Global Impression of Change (CaGI-C) and Severity (CaGI-S)
 - Time to beginning of symptom relief was defined as CaGI-C ratings of at least 'A Little Better' for ≥2 time points in a row within 12 hours
 - Time to reduction in severity was defined as a decrease in CaGI-S rating from baseline for ≥2 time points in a row within 12 hours
 - Time to complete attack resolution was defined as a CaGI-S rating of 'None' within 24 hours

Results

Participant Demographics

- As of June 5, 2025, 36 pediatric participants were enrolled in KONFIDENT-KID (**Table 1**)

Table 1. Participant Demographics

	Participants in sebetralstat ODT dosing group[a]			
	75 mg n=3	150 mg n=27	300 mg n=6	All participants N=36
Age, mean (range), years	5.0 (4-5)	8.0 (6-11)	9.5 (8-10)	8.0 (4-11)
Sex, male, n (%)	1 (33.3)	14 (51.9)	3 (50.0)	18 (50.0)
Race, n (%)				
White	3 (100)	20 (74.1)	3 (50.0)	26 (72.2)
Other[b]	0	3 (11.1)	2 (33.3)	5 (13.9)
Not reported	0	4 (14.8)	1 (16.7)	5 (13.9)
Weight, mean (range), kg	19.4 (19.0-19.5)	28.0 (20.2-43.2)	50.6 (47.2-70.0)	28.8 (19.0-70.0)
HAE-C1INH-Type 1, n (%)	3 (100)	24 (88.9)	5 (83.3)	32 (88.9)

[a]No participants have received 600 mg sebetralstat.
[b]Includes Asian (n=2; 5.6%), Black or African American (n=1; 2.8%), American Indian/Native Alaskan (n=1; 2.8%), other (n=1; 2.8%).
Data cutoff date: June 5, 2025.
HAE-C1INH, hereditary angioedema with C1 inhibitor deficiency; ODT, orally disintegrating tablet.

Part 1: Pharmacokinetics

Figure 2. Population PK Predicted and Observed Pediatric Exposures

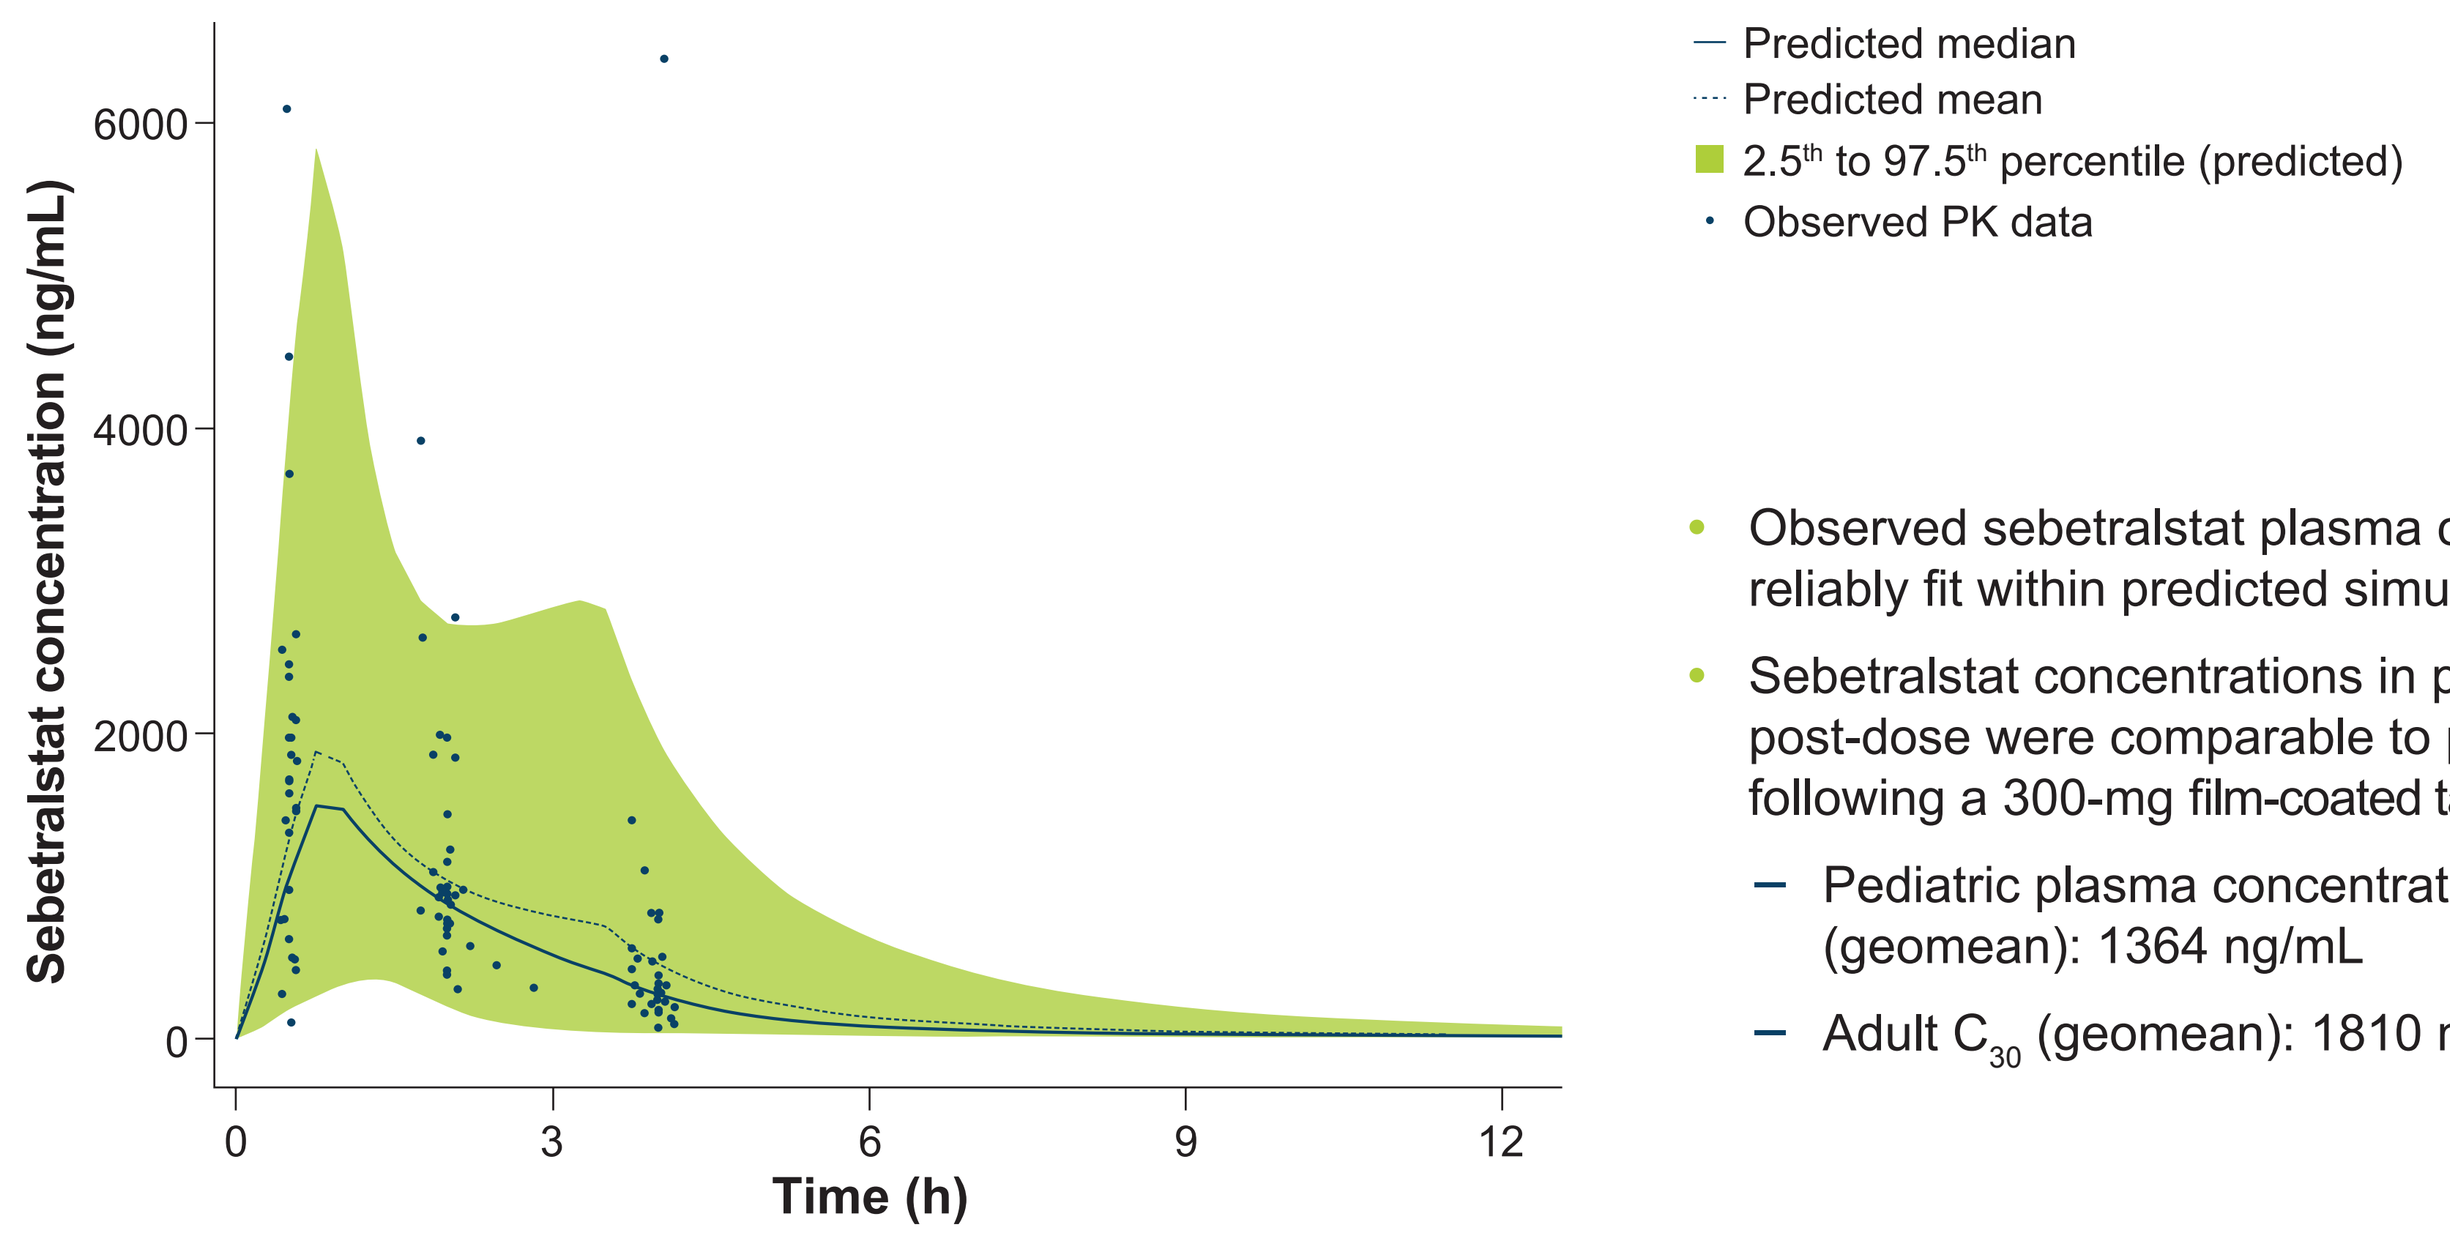

Sebetralstat concentration (ng/mL)
6000
4000
2000
0
0
3
6
9
12
Time (h)
Predicted median
Predicted mean
2.5th to 97.5th percentile (predicted)
Observed PK data

Data cutoff date: June 5, 2025.
PK, pharmacokinetic.

- Observed sebetralstat plasma concentrations from 35 patients reliably fit within predicted simulated exposures (**Figure 2**)
- Sebetralstat concentrations in pediatric participants 30 minutes post-dose were comparable to plasma concentrations in adults following a 300-mg film-coated tablet (FCT) dose
 - Pediatric plasma concentration 30 minutes after dosing (C_{30}) (geomean): 1364 ng/mL
 - Adult C_{30} (geomean): 1810 ng/mL[15]

Attack Characteristics

- Participants treated a mean of 0.8 (SD, 0.56) attacks/month with sebetralstat ODT
- Sixty-five attacks were treated with sebetralstat (**Table 2**) by 26 pediatric participants
 - The baseline severity of most (86.1%) attacks was mild or moderate (**Table 2**); 48% of attacks were mucosal (**Figure 3**)

Table 2. Baseline Attack Severity[a,b]

	Attacks in sebetralstat ODT dosing group			
	75 mg n=1	150 mg n=55	300 mg n=9	All attacks N=65
Severity, n (%)				
Mild[c]	1 (100)	16 (29.1)	4 (44.4)	21 (32.3)
Moderate	0	31 (56.4)	4 (44.4)	35 (53.8)
Severe	0	5 (9.1)	0	5 (7.7)
Very Severe	0	0	0	0

[a]Assessed by CaGI-S.
[b]Missing: 3 (5.5%) attacks in 150-mg group and 1 (11.1%) attack in 300-mg group.
[c]Includes attacks with baseline CaGI-S rating of 'None': 1 (1.8%) attack in 150-mg group and 2 (22.2%) attacks in 300-mg group.
Data cutoff date: June 5, 2025.
CaGI-S, Caregiver Global Impression of Severity; ODT, orally disintegrating tablet.

Figure 3. Primary Attack Location[a]

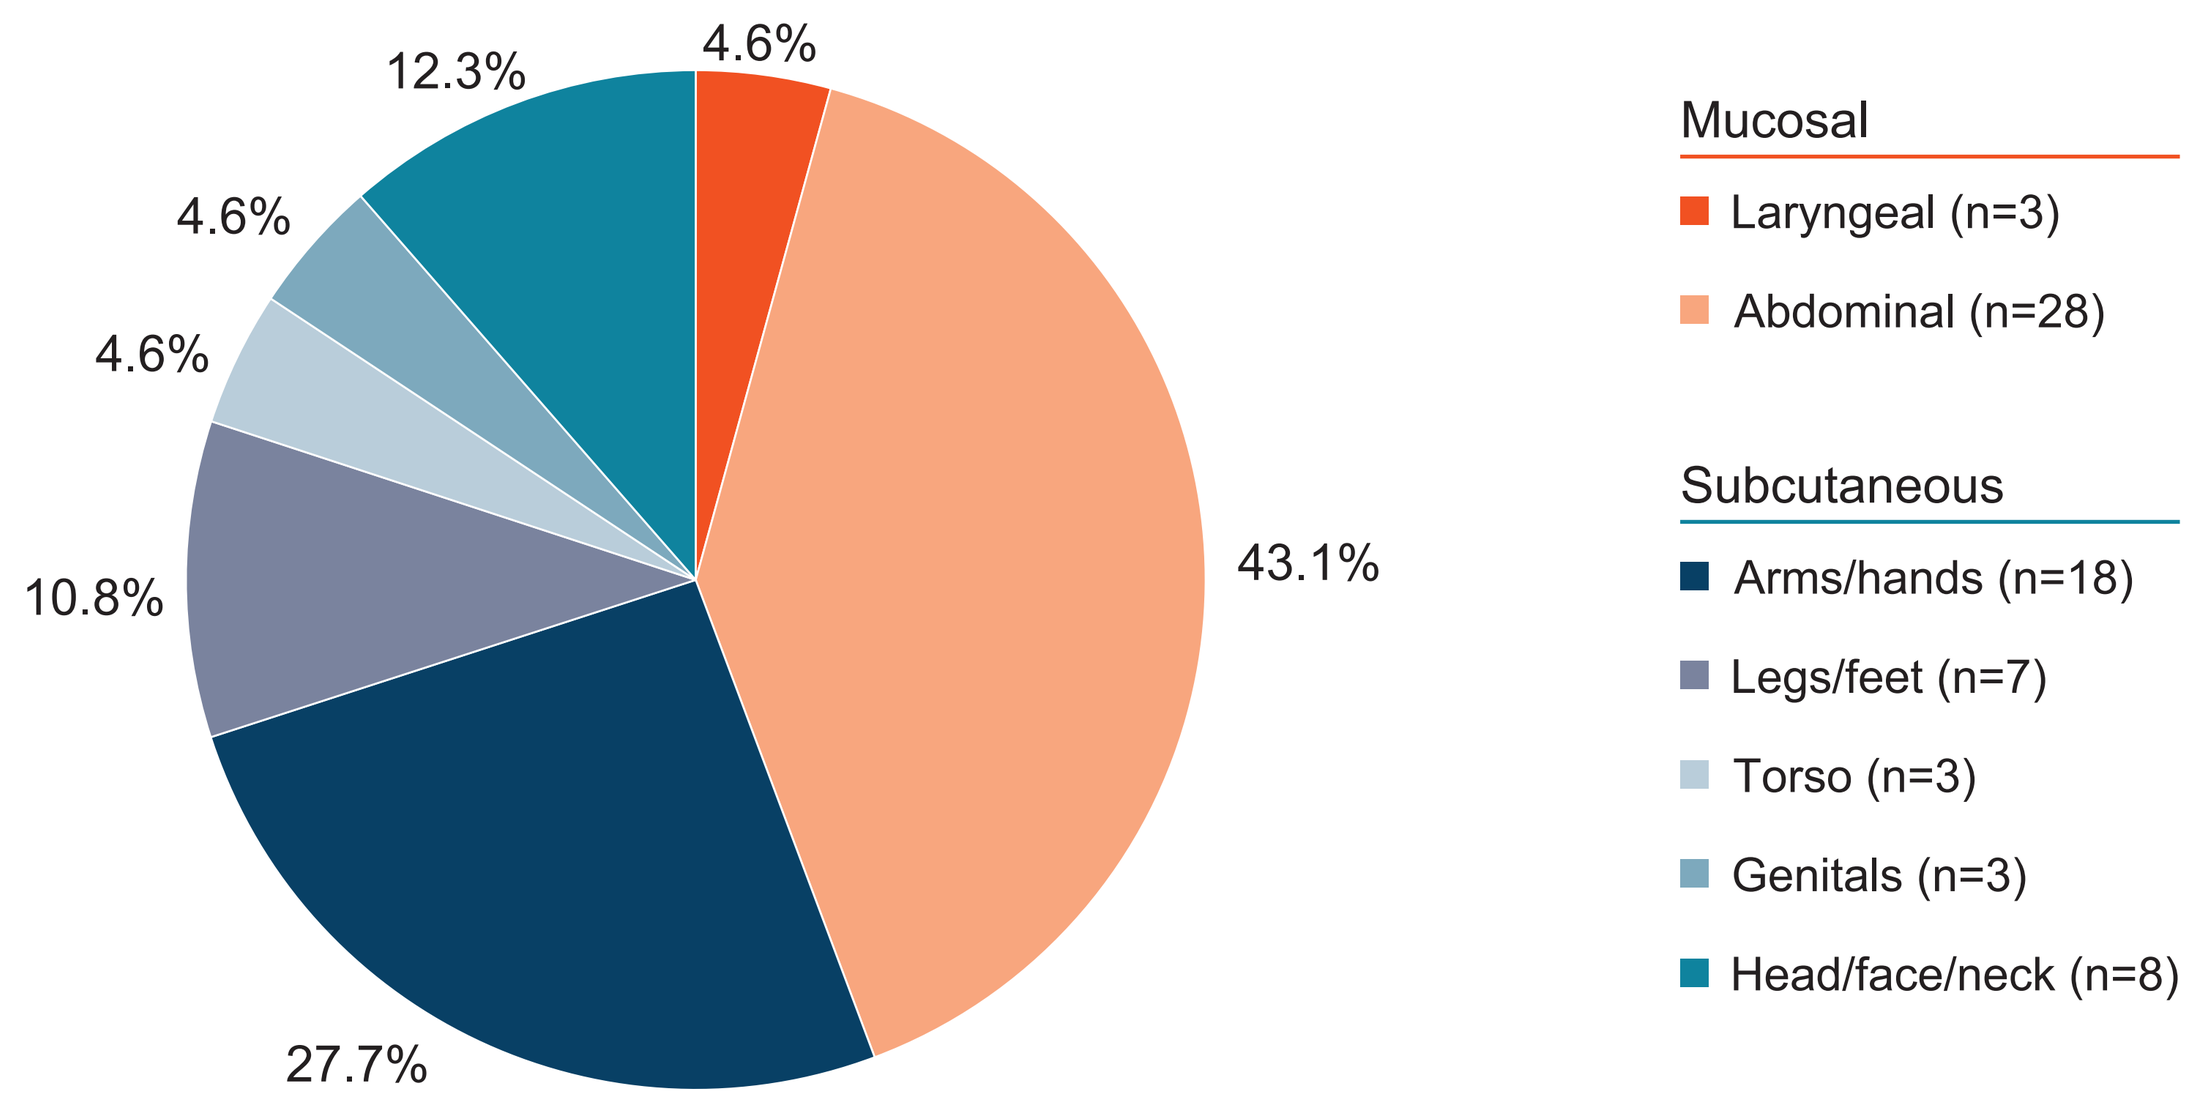

4.6%
43.1%
27.7%
10.8%
4.6%
4.6%
12.3%
Mucosal
Laryngeal (n=3)
Abdominal (n=28)
Subcutaneous
Arms/hands (n=18)
Legs/feet (n=7)
Torso (n=3)
Genitals (n=3)
Head/face/neck (n=8)

[a]Missing: 2 (3.6%) attacks in 150-mg group and 1 (11.1%) attack in 300-mg group.
Data cutoff date: June 5, 2025.

Time to Treatment

- Across all attacks, the median (IQR) time to treatment was 30 minutes (5-135) (**Figure 4**)
 - 61.3% of attacks were treated in <1 hour of onset

Figure 4. Time from Attack Onset to Sebetralstat ODT Administration[a,b]

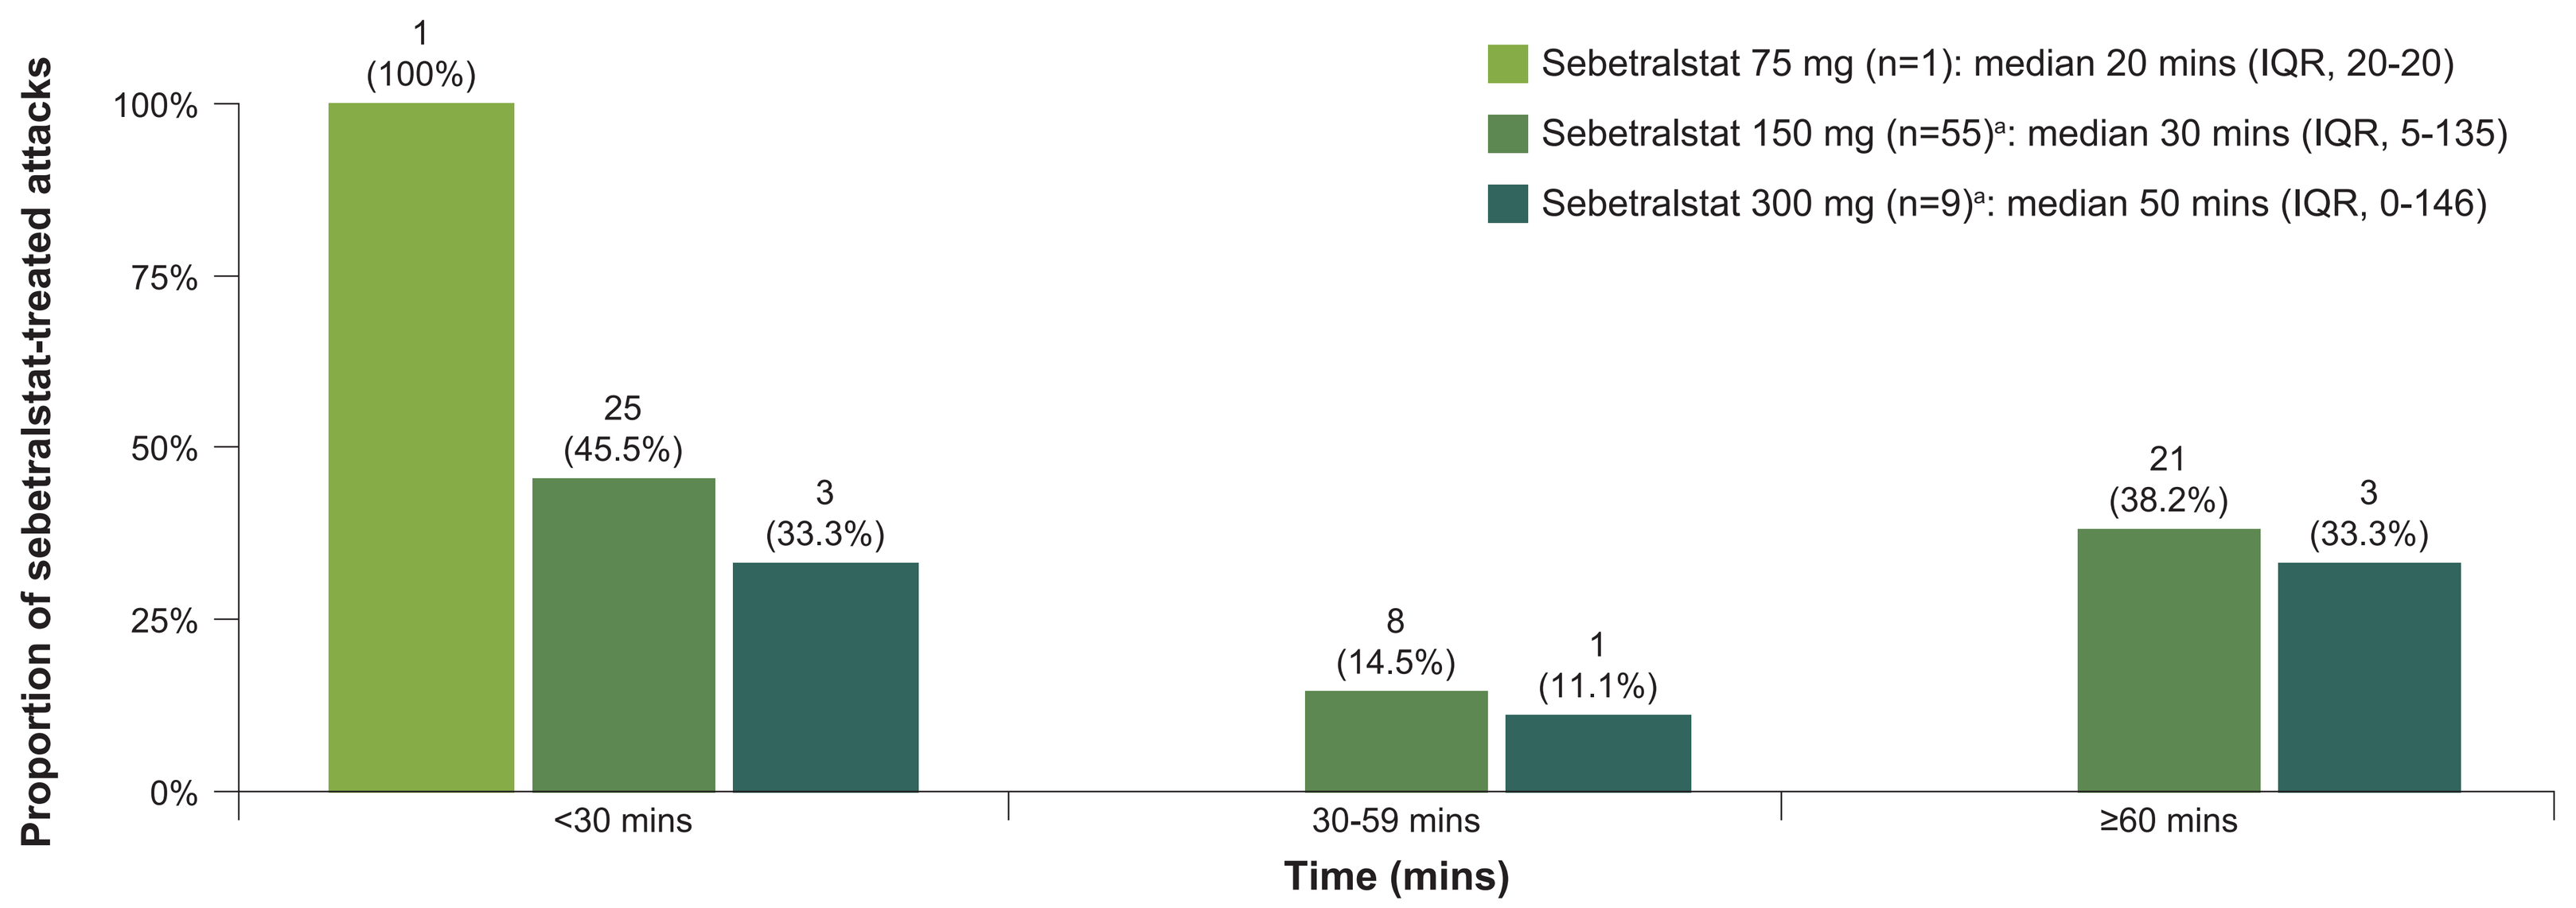

Proportion of sebetralstat-treated attacks
100%
75%
50%
25%
0%
1 (100%)
25 (45.5%)
3 (33.3%)
8 (14.5%)
1 (11.1%)
21 (38.2%)
3 (33.3%)
<30 mins
30-59 mins
≥60 mins
Time (mins)
Sebetralstat 75 mg (n=1): median 20 mins (IQR, 20-20)
Sebetralstat 150 mg (n=55)a: median 30 mins (IQR, 5-135)
Sebetralstat 300 mg (n=9)a: median 50 mins (IQR, 0-146)

[a]Data missing for 1 (1.8%) attack in the 150-mg group and 2 (22.2%) attacks in the 300-mg group.
[b]Caregiver or child could administer treatment.
Data cutoff date: June 5, 2025.
IQR, interquartile range; ODT, orally disintegrating tablet.

Part 2: Interim Efficacy

Table 3. Efficacy of Sebetralstat in Pediatric Participants

	Attacks in sebetralstat ODT dosing group		
	75 mg n=1	150 mg n=55	300 mg n=9
Time to beginning of symptom relief within 12 hours, median (IQR), hours	**NE**	**1.50** (0.50 to 4.00)	**NE** (0.50 to >12)
Time to reduction in attack severity within 12 hours, median (IQR), hours	**NE**	**4.00** (1.50 to >12)	**NE** (1.00 to >12)
Time to complete attack resolution within 24 hours, median (IQR), hours	**NE**	**12.00** (8.00 to 24.00)	**18.00** (4.00 to >24)

Data cutoff date: June 5, 2025.
NE, not estimable; IQR, interquartile range.

- Overall, 78.9% attacks achieved beginning of symptom relief within 12 hours (includes all attacks with post-baseline values)
- Conventional medication was utilized within 12 hours for 3.1% of attacks

Safety and Tolerability

- Fifteen adverse events occurred in 9 participants; none were serious or considered treatment-related (**Table 4**)
- No reports of difficulty swallowing sebetralstat ODT

Table 4. Safety and Tolerability

	All participants[a] N=26
Any TEAE, n (%)	9 (34.6)
Treatment-related	**0**
Serious TEAE, n (%)	0
Treatment-related	**0**
Severe TEAE, n (%)	0
Treatment-related	**0**
Any TEAE leading to discontinuation, n (%)	0
Any TEAE leading to death, n (%)	0

[a]Participants who treated at least 1 HAE attack with sebetralstat.
Data cutoff date: June 5, 2025.
HAE, hereditary angioedema; TEAE, treatment-emergent adverse event.

Conclusions

- **Children with HAE aged 2-11 years have a high unmet need for non-parenteral treatment options**
- **Observed sebetralstat plasma concentrations reliably fit within predicted exposures**
 - **Sebetralstat concentrations in children 30 minutes post-dose were comparable to plasma concentrations in adults following a 300-mg dose**
- **A high number of attacks were treated with sebetralstat in this study**
 - **Participants treated a mean of 0.8 attacks/month with sebetralstat**
- **Sebetralstat enabled early treatment (median: 30 minutes) by participants or their caregivers**
 - **Attacks occurred in all locations with 32% still mild at time of treatment**
- **Sebetralstat was generally safe, well tolerated, and demonstrated rapid symptom relief, reduction in severity and complete attack resolution**
- **Sebetralstat ODT has the potential to address high unmet need for children (2-11 years) with HAE and their caregivers**

References

1. Tachdjian R et al. *Clin Pediatr (Phila)*. 2023;62(9):973-980.
2. Pagnier A et al. *Pediatr Allergy Immunol*. 2024;35(12):e14268.
3. Piotrowicz-Wójcik K et al. *Children (Basel)*. 2024;11(2):237.
4. Lo SH et al. *PharmacoEconomics Open*. 2022;6(2):231-239.
5. Martinez-Saguer I et al. *Ann Allergy Asthma Immunol*. 2023;131(5):S36-S37.
6. Tachdjian R et al. *J Allergy Clin Immunol*. 2023;2(suppl):Ab133.
7. Berinert (C1 esterase inhibitor [human]). PI. CSL Behring; 2022.
8. Ruconest 2100 Units powder for solution for injection. SmPC. Pharming Group N. V. The Netherlands; 2015.
9. Cinryze 500 IU powder and solvent for solution for injection. SmPC. Takeda Manufacturing AG; 2022.
10. Firazyr (icatibant). SmPC (EU). Takeda Pharmaceuticals; 2025.
11. Ekterly (sebetralstat). PI. KalVista Pharmaceuticals; 2025.
12. Ekterly (sebetralstat). SmPC (UK). KalVista Pharmaceuticals; 2025.
13. Ekterly (sebetralstat). SmPC (EU). KalVista Pharmaceuticals; 2025.
14. ClinicalTrials.gov NCT06467084. Accessed October 9, 2025. https://clinicaltrials.gov/study/NCT06467084.
15. Data on file. KalVista Pharmaceuticals, Inc.

Acknowledgments

We thank the people living with HAE and their families; the HAEA, HAEi, and member organizations; and the investigator teams who are contributing to the international KONFIDENT-KID trial. Medical writing assistance was provided under the direction of the authors by Richard W. Davis IV, PhD, of ApotheCom (San Francisco, CA) and was supported by KalVista. Pharmaceuticals.

Please visit the KalVista virtual medical booth after the presentation to view this poster.

Presented at the 63rd Western Society of Allergy, Asthma, & Immunology Annual Scientific Meeting; February 1-5, 2026; Wailea, Hawaii.

Copyright © 2026 KalVista Pharmaceuticals, Inc. All rights reserved.