Treatment Trends and Satisfaction with Repeated Use of Oral On-demand Sebetralstat for Hereditary Angioedema

Maeve O'Connor,[1,2] **Aleena Banerji,**[3,4] **Jonathan A. Bernstein,**[5] **William R. Lumry,**[6] **Michael E. Manning,**[7,8] **Marc A. Riedl,**[9] **Daniel F. Soteres,**[10] **Paolo Bajcic,**[11] **James Hao,**[11] **Michael D. Smith,**[11] **Paul K. Audhya,**[11] **Raffi Tachdjian**[12]

[1]Integrative Immunology Care, LLC, Charlotte, NC, USA; [2]Allergy, Asthma, & Immunology Research Institute, Charlotte, NC, USA; [3]Department of Medicine, Division of Rheumatology, Allergy, and Immunology, Massachusetts General Hospital, Boston, MA, USA; [4]Harvard Medical School, Boston, MA, USA; [5]University of Cincinnati College of Medicine and Bernstein Clinical Research Center, Cincinnati, OH, USA; [6]AARA Research Center, Dallas, TX, USA; [7]Allergy, Asthma and Immunology Associates, Ltd., Scottsdale, AZ, USA; [8]University of Arizona College of Medicine-Phoenix, Scottsdale, AZ, USA; [9]University of California, San Diego, La Jolla, CA, USA; [10]Asthma & Allergy Associates PC, Colorado Springs, CO, USA; [11]KalVista Pharmaceuticals, Framingham, MA, USA; [12]University of California, Los Angeles, School of Medicine, Los Angeles, CA, USA

Background

- International treatment guidelines recommend that all hereditary angioedema (HAE) attacks, regardless of severity or location, are considered for treatment with on-demand therapy[1,2]
- Sebetralstat, an oral plasma kallikrein inhibitor, has recently been approved for on-demand treatment of HAE attacks in patients ≥12 years old in the US, EU, Japan, and other countries[3-5]
- KONFIDENT-S (NCT05505916; EudraCT: 2021-001176-42) is an ongoing, 2-year, multicenter, open-label extension study investigating the tolerability, safety, and effectiveness of oral sebetralstat with no limit on the number of attacks that could be treated[6]
 - The trial design was patient centric, employing previously reported real-world elements[6] and allowing participants to determine whether to administer conventional injectable on-demand treatment or sebetralstat

Objective

- This analysis of the KONFIDENT-S trial examined trends and satisfaction associated with repeated on-demand treatment of HAE attacks with sebetralstat

Methods

- Eligible participants were adults and adolescents (≥12 years of age) with HAE-C1INH and at least 2 documented attacks within 3 months before enrollment or had completed the phase 3 KONFIDENT trial (**Figure 1**), as previously reported[6]
 - Participants receiving long-term prophylaxis (LTP) were required to be on a stable dose and regimen for ≥3 months immediately before and during the study
- Consistent with international HAE treatment guideline recommendations, participants were instructed to self-administer treatment (sebetralstat 600 mg) as early as possible after attack onset, regardless of severity or anatomical location
 - If warranted, an optional second administration of sebetralstat was permitted ≥3 hours after the first administration
- Conventional injectable on-demand treatment was allowed at any time, as determined by the participant, including as initial treatment for an attack

Figure 1. KONFIDENT-S OLE Trial Design

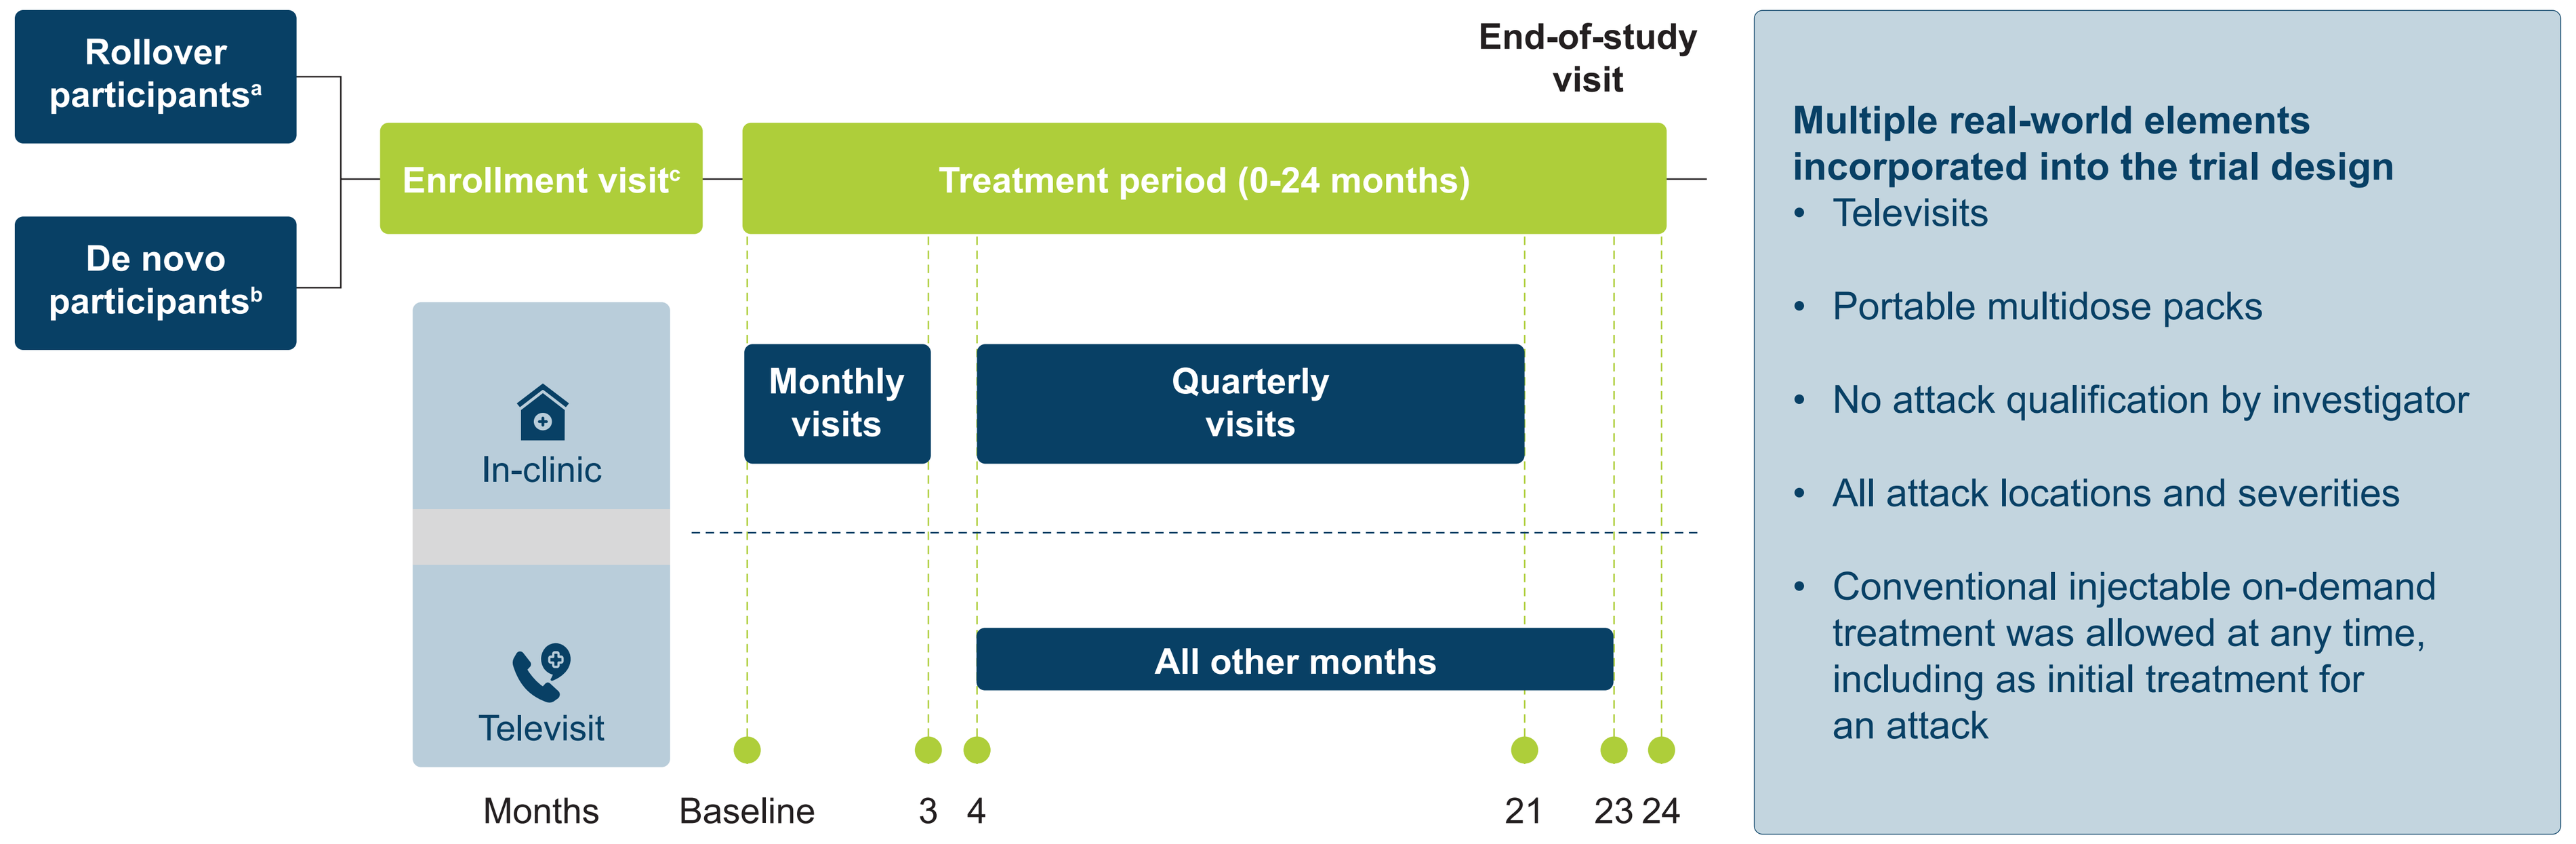

Rollover participants[a]
De novo participants[b]
Enrollment visit[c]
Treatment period (0-24 months)
End-of-study visit
In-clinic
Televisit
Monthly visits
Quarterly visits
All other months
Months
Baseline
3
4
21
23 24
Multiple real-world elements incorporated into the trial design
• Televisits
• Portable multidose packs
• No attack qualification by investigator
• All attack locations and severities
• Conventional injectable on-demand treatment was allowed at any time, including as initial treatment for an attack

[a]Completed the phase 3 KONFIDENT trial.
[b]All other participants, including those who participated in the phase 2 trial.
[c]For de novo participants, the enrollment visit is a screening visit.

- Treatment satisfaction for sebetralstat-treated attacks was assessed at 24 hours after administration of on-demand treatment for each attack using a 7-point scale (**Figure 2**)
 - No dissatisfaction was defined as either neutral (0) or satisfied (+1, +2, or +3)
- Trends were assessed using locally estimated scatterplot smoothing (LOESS) fit curves (smooth=0.5) in the first 30 attacks experienced by a participant
 - Changes in LOESS fit were analyzed by participant age, treatment regimen, and attack location

Figure 2. Treatment Satisfaction Scale (7-point Likert Scale)

Overall, how satisfied were you with sebetralstat therapy for this attack?

-3	-2	-1	0	+1	+2	+3
Extremely Dissatisfied	Very Dissatisfied	Dissatisfied	Neutral	Satisfied	Very Satisfied	Extremely Satisfied

Results

- As of July 9, 2025, 2464 attacks were treated with sebetralstat 600 mg film-coated tablets by 136 participants (**Table 1**)
 - The median number of attacks treated by participants was 12 (IQR, 6-25); 24 participants (17.6%) treated ≥30 attacks

Table 1. Participant Demographics

	All participants (N=136)
Age, median (IQR), years	35.5 (21.0-49.0)
≥18 years, n (%)	113 (83.1)
Sex, female, n (%)	89 (65.4)
Race, n (%)[a]	
White	101 (74.3)
Asian	17 (12.5)
Other or Multiple	11 (8.1)
BMI, median (IQR), kg/m^2	25.4 (22.3-30.7)
HAE-C1INH-Type 1, n (%)	127 (93.4)
Current treatment regimen. n (%)	
On-demand only	100 (73.5)
On-demand + LTP	36 (26.5)

Data cutoff: July 9, 2025.
BMI, body mass index; HAE-C1INH, hereditary angioedema due to C1 inhibitor deficiency; IQR interquartile range; LTP, long-term prophylaxis.
[a]7 participants (5.1%) did not report race.

- Across the first 30 attacks, a median of 19.3% of attacks were treated with a second administration of sebetralstat within 12 hours
 - The proportion of attacks treated with a second administration within 12 hours followed a downward trend based on LOESS fit from attack 1 to attack 30 (**Figure 3**)
 - The largest downward trends were observed in adult participants and mucosal attacks (LOESS fit % change of -16.1 and -13.0, respectively)

Figure 3. Second Administration of Sebetralstat by Sebetralstat-treated HAE Attack Number

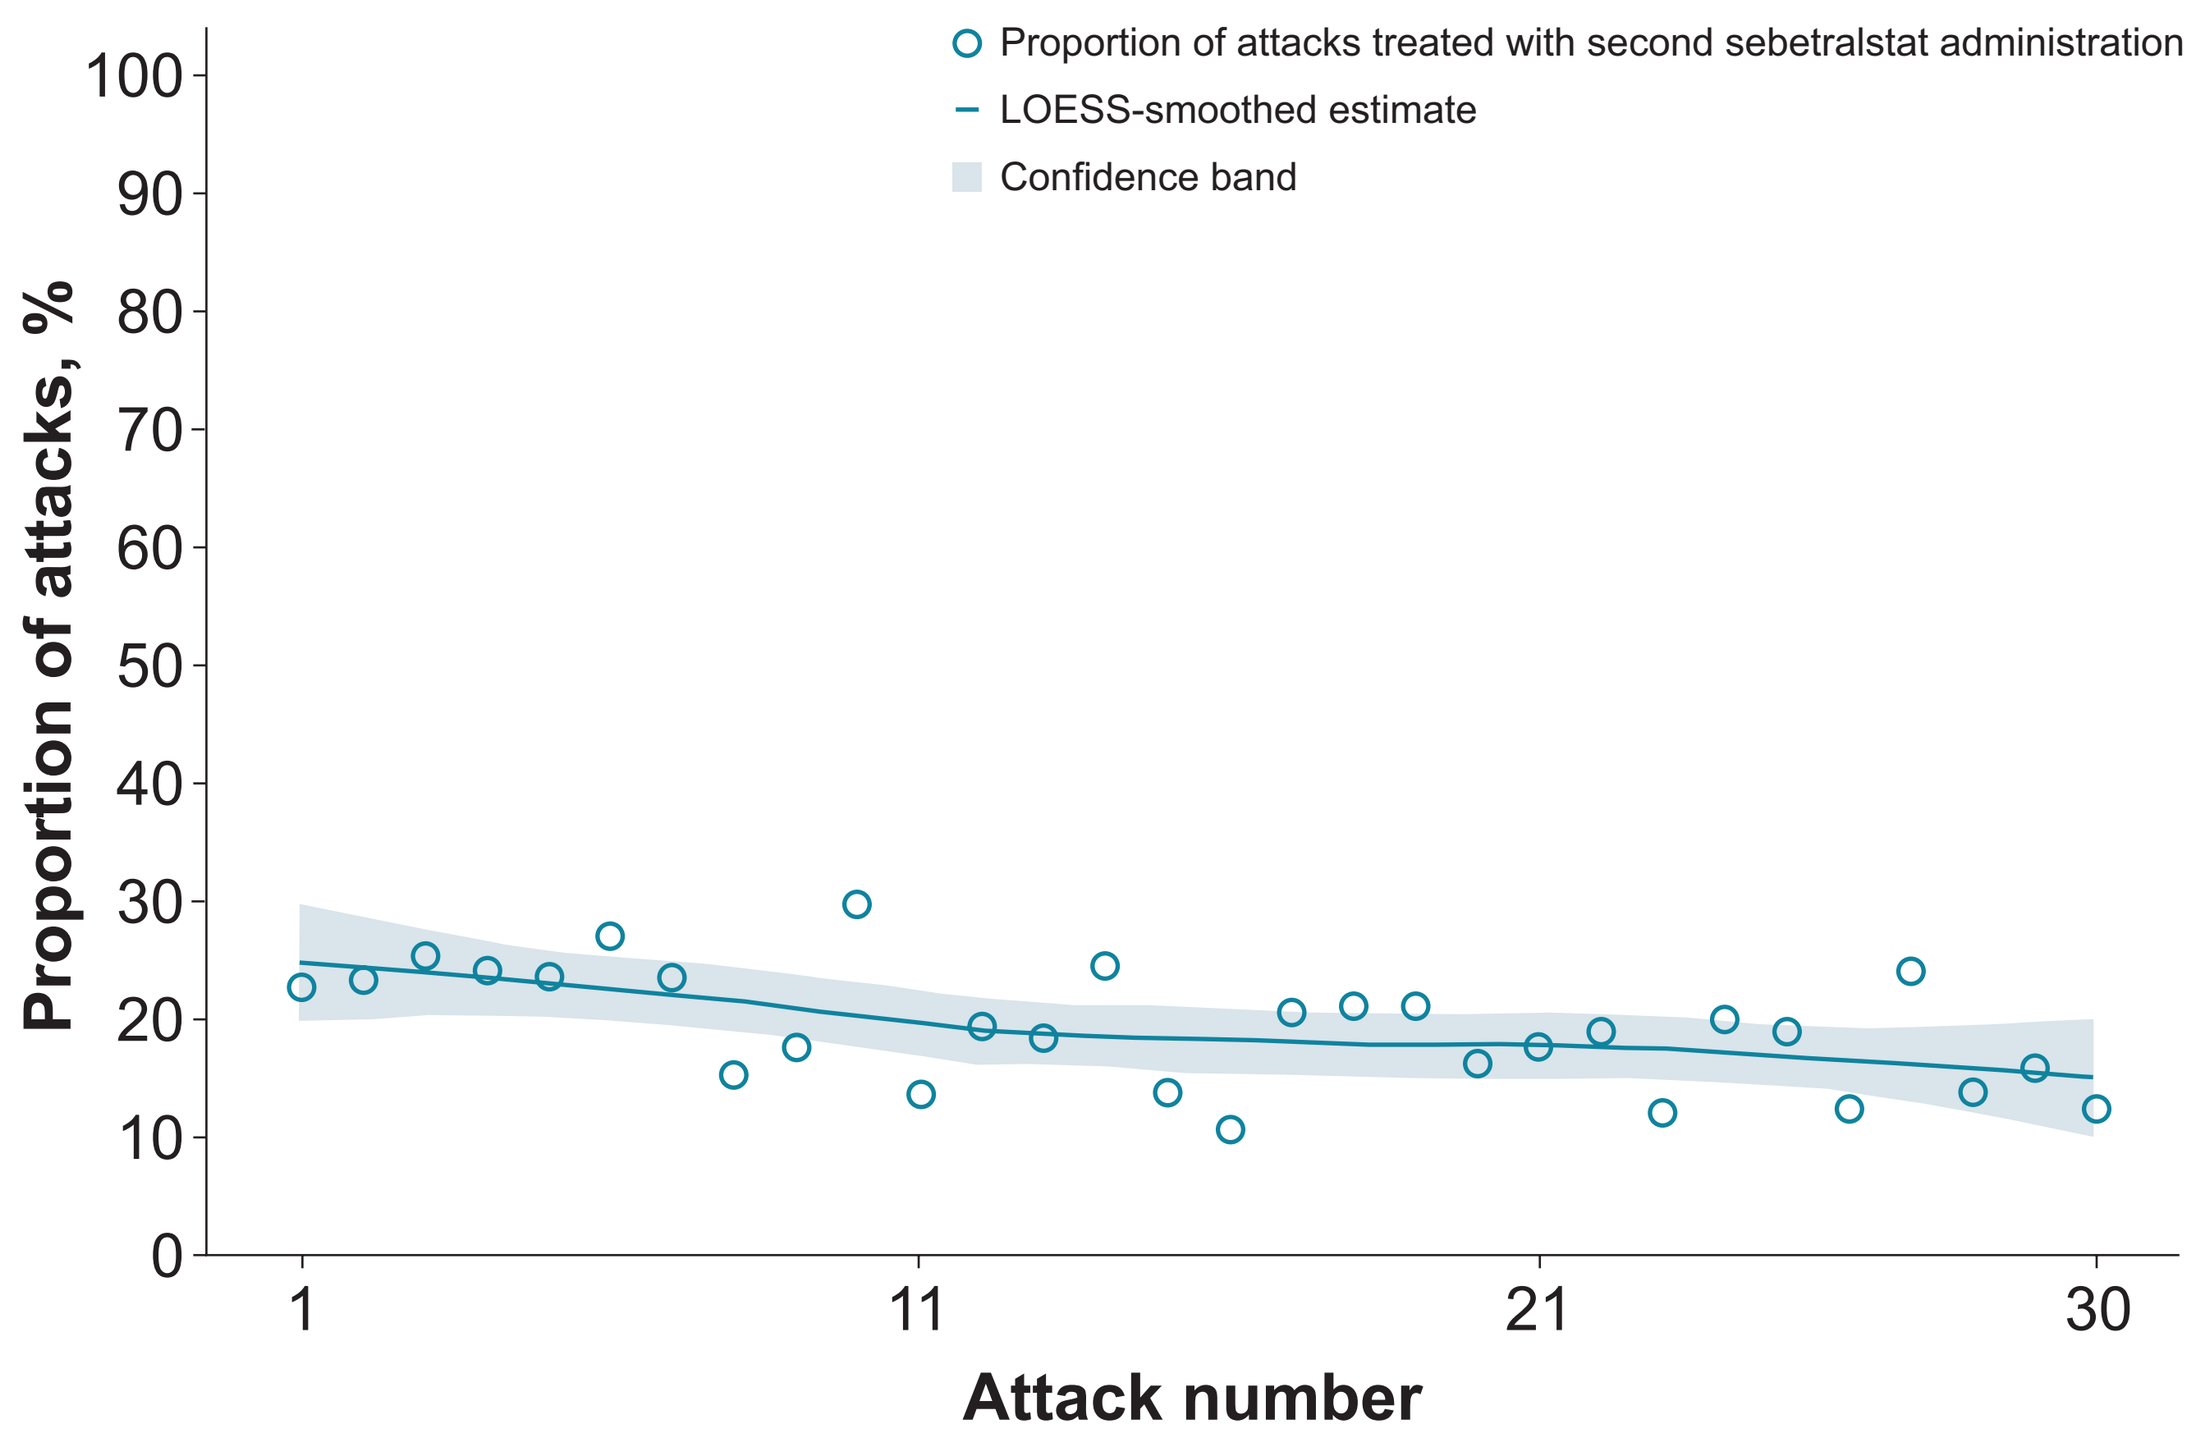

Proportion of attacks treated with second sebetralstat administration
LOESS-smoothed estimate
Confidence band
Proportion of attacks, %
100
90
80
70
60
50
40
30
20
10
0
1
11
21
30
Attack number

Data cutoff: July 9, 2025.
LOESS, locally estimated scatterplot smoothing.

- Across the first 30 attacks, a median of 5.1% of attacks were treated with a conventional injectable on-demand treatment within 12 hours
 - The proportion of attacks treated with conventional injectable on-demand treatment within 12 hours followed a downward trend based on LOESS fit from attack 1 to attack 30 (**Figure 4**)
 - The largest downward trends were observed in mucosal attacks and in participants receiving LTP[a] (LOESS fit % change of -9.4 and -9.0, respectively)

Figure 4. Conventional Injectable On-demand Treatment Use by Sebetralstat-treated HAE Attack Number

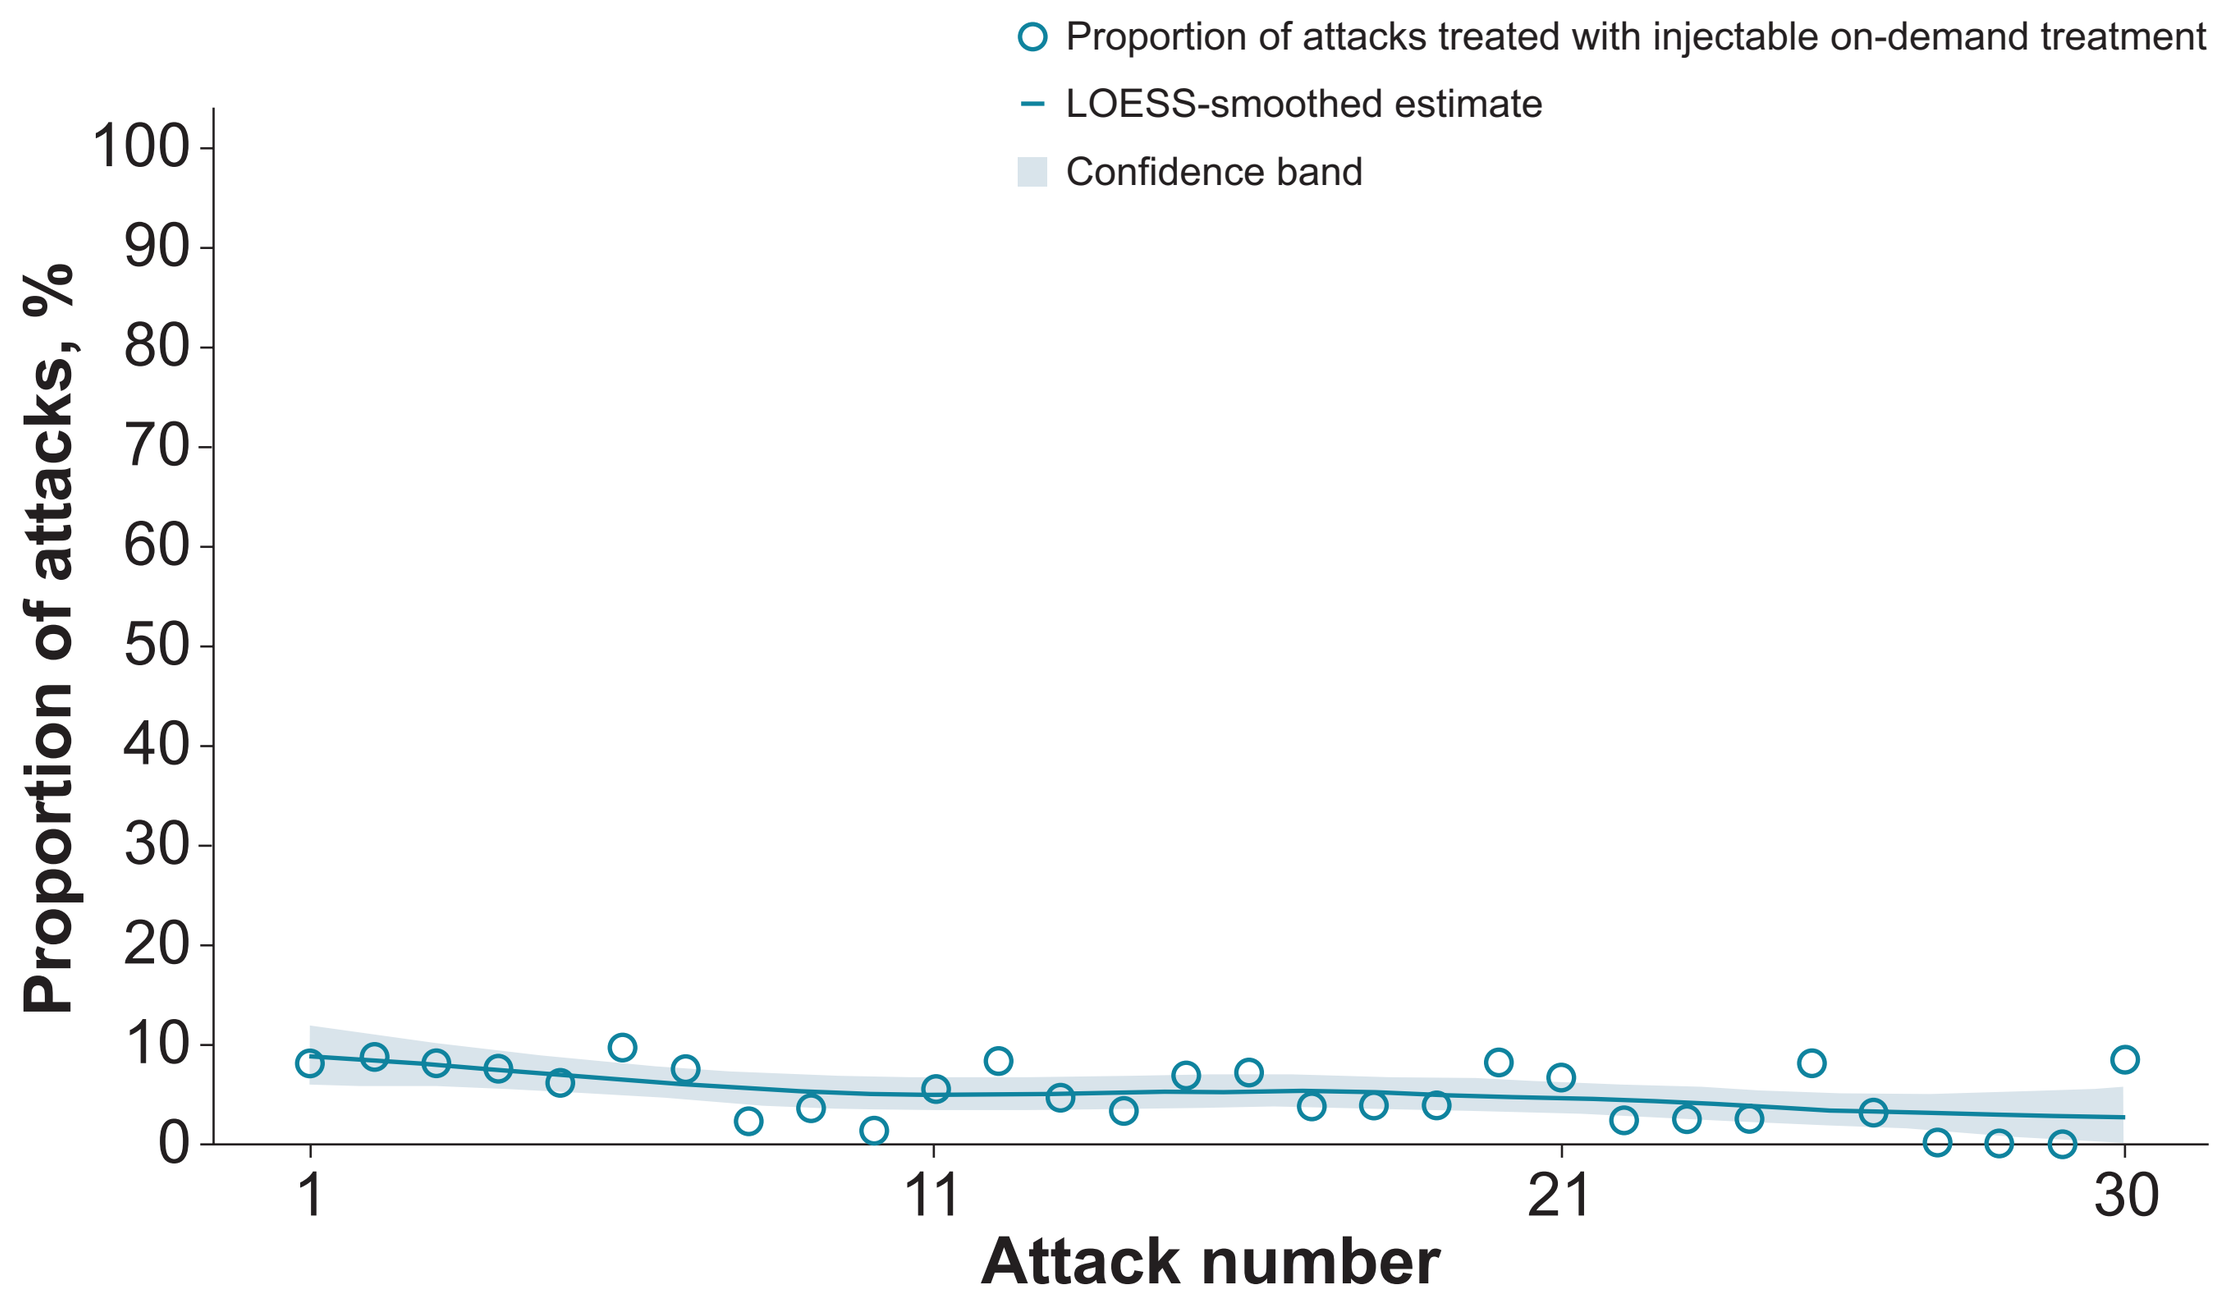

Proportion of attacks treated with injectable on-demand treatment
LOESS-smoothed estimate
Confidence band
Proportion of attacks, %
100
90
80
70
60
50
40
30
20
10
0
1
11
21
30
Attack number

Data cutoff: July 9, 2025.
LOESS, locally estimated scatterplot smoothing.

- Participants were satisfied with sebetralstat for 83.1% of their first 30 attacks, with no discernible longitudinal trend in satisfaction levels based on LOESS fit (**Figure 5**)
 - Of the subgroups assessed, positive trends in satisfaction were noted among adolescents,[a] mucosal attacks, and participants receiving LTP[a] (LOESS fit % change: 14.8, 13.1, and 10.6, respectively)
- No dissatisfaction was reported for 91.1% of sebetralstat-treated attacks (6.3% neutral, 19.1% satisfied, 38.4% very satisfied, 27.2% extremely satisfied)

Figure 5. Treatment Satisfaction by Sebetralstat-treated Attack Number

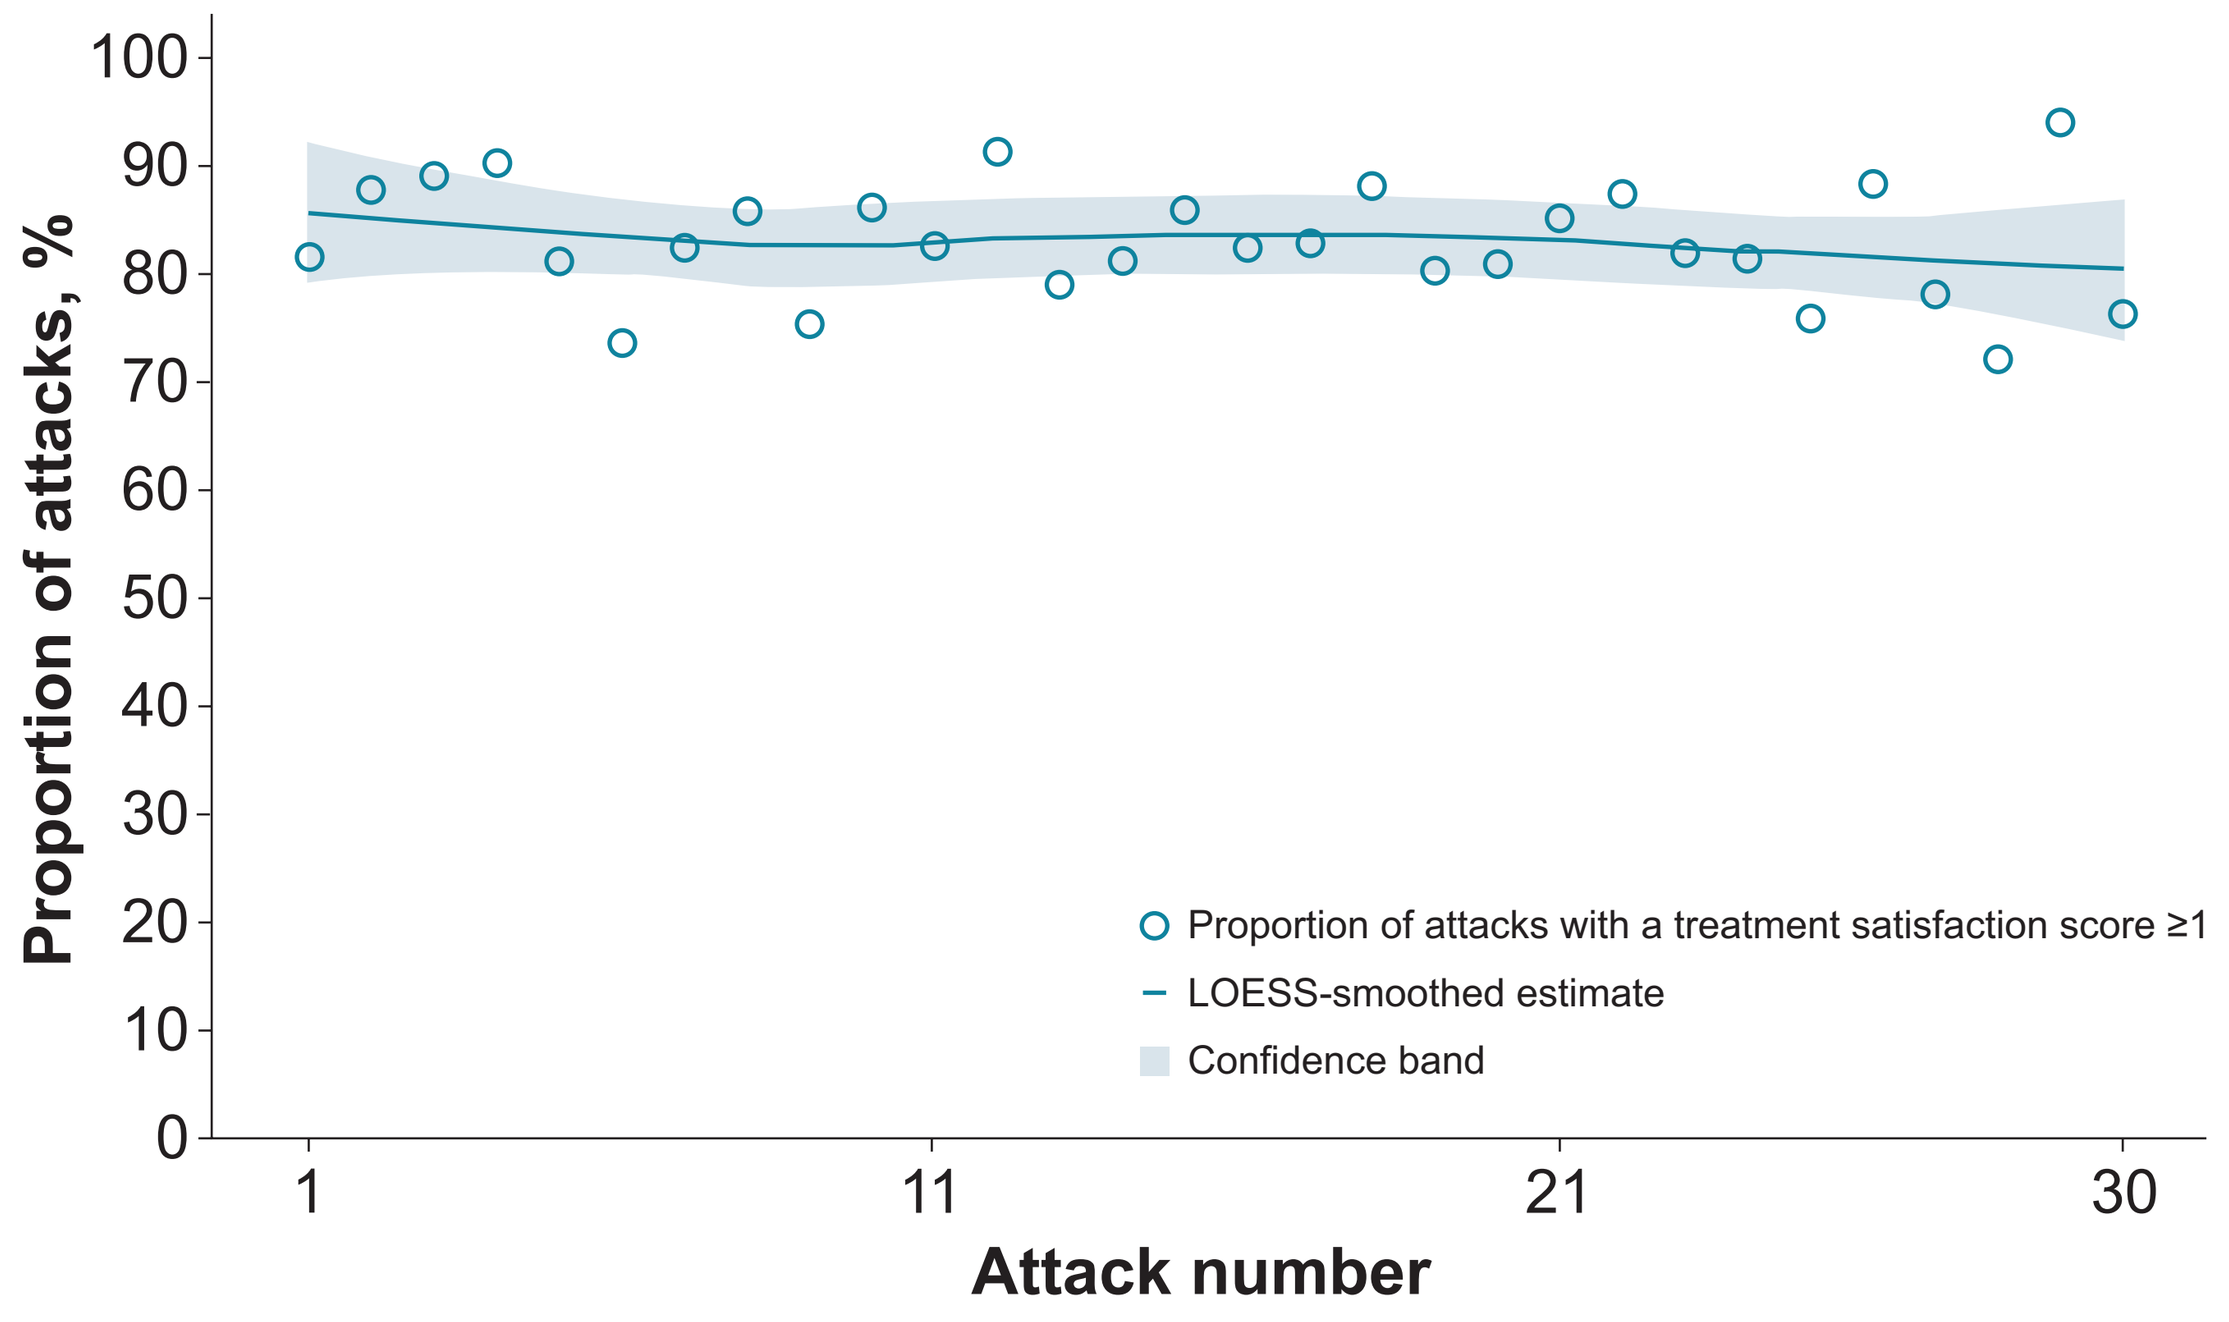

Proportion of attacks with a treatment satisfaction score ≥1
LOESS-smoothed estimate
Confidence band
Proportion of attacks, %
100
90
80
70
60
50
40
30
20
10
0
1
11
21
30
Attack number

Data cutoff: July 9, 2025.
LOESS, locally estimated scatterplot smoothing.
[a]Subgroups were small and results should be interpreted with caution

Conclusions

- **Across repeated treatments of HAE attacks with sebetralstat, the median rates of second administration and conventional injectable treatment were 19.3% and 5.1%, respectively, with downward trends observed from attack 1 to attack 30**
 - **The reduction in second administrations was most pronounced in adults and those experiencing mucosal attacks**
 - **The reduction in conventional injectable treatment was most pronounced in participants receiving LTP and those experiencing mucosal attacks**
- **Patient satisfaction with sebetralstat treatment remained consistently high throughout the study alongside lessening use of second administrations or conventional injectable treatment**

References

1. Maurer M et al. *Allergy*. 2022;77(7):1061-1990.
2. Busse PJ et al. *J Allergy Clin Immunol Pract*. 2021;9(1):132-150.e3.
3. Ekterly (sebetralstat). PI. KalVista Pharmaceuticals; 2025.
4. Ekterly (sebetralstat). SmPC (EU). KalVista Pharmaceuticals; 2025.
5. KalVista Pharmaceuticals Announces Approval of EKTERLY® (sebetralstat) in Japan, First and Only Oral On-demand Treatment for Hereditary Angioedema. Business Wire. Accessed January 14, 2026. https://www.businesswire.com/news/home/20251222393408/en
6. Farkas H et al. *J Allergy Clin Immunol Pract*. 2025;13(11):3094-3103.

Acknowledgments

We thank the people living with HAE and their families; the HAEA, HAEi, and member organizations; and the investigator teams who are contributing to the international KONFIDENT-S trial. Medical writing assistance was provided under the direction of the authors by Bridget Healy, MBChB, MPH, of ApotheCom, San Francisco, CA, USA, and was funded by KalVista Pharmaceuticals.

Please visit the KalVista virtual medical booth after the presentation to view this poster.

Presented at the Eastern Allergy Conference 2026; May 28-31, 2026; Palm Beach, Florida

Copyright © 2026 KalVista Pharmaceuticals, Inc. All rights reserved.