HSD42

Recent Trends in On-demand Treatment Prescriptions for Hereditary Angioedema: A US Claims Database Analysis

Raffi Tachdjian,[1] Sandra Christiansen,[2] Timothy J. Craig,[3] Daniel F. Soteres,[4] Maeve O'Connor,[5] Marc A. Riedl,[2] Aditya Sehgal,[6] Paul K. Audhya,[7] Alice Wang[7]

[1]University of California, Los Angeles, School of Medicine, Los Angeles, CA, USA; [2]University of California, San Diego, La Jolla, CA, USA; [3]Department of Medicine, Pediatrics, MFM and BioMedical Sciences Penn State University, Hershey, PA, USA; Vinmec International Hospital, Times City, Hanoi, Vietnam; and VinUniversity, Hanoi, Vietnam; [4]Asthma & Allergy Associates, PC and Research Center, Colorado Springs, CO, USA; [5]Allergy, Asthma, & Immunology Relief, Charlotte, NC, USA; [6]Pharmsight, Haryana, India; [7]KalVista Pharmaceuticals, Framingham, MA, USA

Background

- Hereditary angioedema (HAE), a rare, genetic disorder with an estimated global prevalence of 1 in 50,000, is characterized by unpredictable, potentially life-threatening acute attacks[1,2]
- HAE guidelines recommend that all attacks, irrespective of location or severity, be managed using on-demand treatment upon the earliest recognition of symptoms and that patients carry on-demand treatment with them at all times[1,2]
- Several on-demand treatment options are available in the United States (US), including those that are administered orally (sebetralstat [Ekterly]), via subcutaneous (SC) injection (ecallantide [Kalbitor]; icatibant [Firazyr]), and via intravenous (IV) infusion (plasma-derived C1 esterase inhibitor [C1INH; Berinert]; recombinant human C1INH [Ruconest])
- To decrease the frequency and burden of attacks, patients may also be prescribed long-term prophylaxis (LTP) treatment[1,2]
 - The following non-androgen LTP treatments are available in the US: oral berotralstat (Orladeyo), SC plasma-derived C1INH (Haegarda), SC lanadelumab (Takhzyro), and IV plasma-derived C1INH (Cinryze)
- Real-world data describing recent trends in on-demand treatment use in the US are limited. This analysis evaluated the utilization of on-demand treatment in patients with HAE in the US

Methods

- The IQVIA PharMetrics® Plus Closed Health Plan Claims Database was used to identify patients with ≥1 claim for HAE on-demand or LTP treatment during the study period (January 1, 2021–December 31, 2024)
- On-demand treatment utilization was estimated among patients with ≥1 claim for on-demand treatment
 - A standardized on-demand treatment dose was defined as 1 icatibant syringe, 2 recombinant human C1INH vials, 3 ecallantide vials, or 4 plasma-derived C1INH vials
 - The average number of on-demand treatment doses was estimated as the mean of the aggregates divided by all claims in each calendar year and extrapolated to the US population[3]
- The total quantity of non-androgen LTP treatment was estimated according to the number of syringes and vials dispensed during the study period among patients with ≥1 claim for non-androgen LTP treatment and was indexed to 100 in 2021 to account for different LTP formulations and dose strengths

Results

- During the 2021–2024 period, 3647 unique patients had ≥1 claim for on-demand or LTP treatment (mean [SD] age, 42 (18) years in 2024; 69%, female)
- The total number of on-demand treatment doses claimed per year was stable over time (**Figure 1**)
- Extrapolating to the overall US population, an average of 83,724 doses of on-demand treatment was estimated during the study period
- Mean number of on-demand treatment doses was largely unchanged (~14 per patient per year [PPPY]) during the study period

Figure 1. Annual on-demand treatment trends by total doses per year

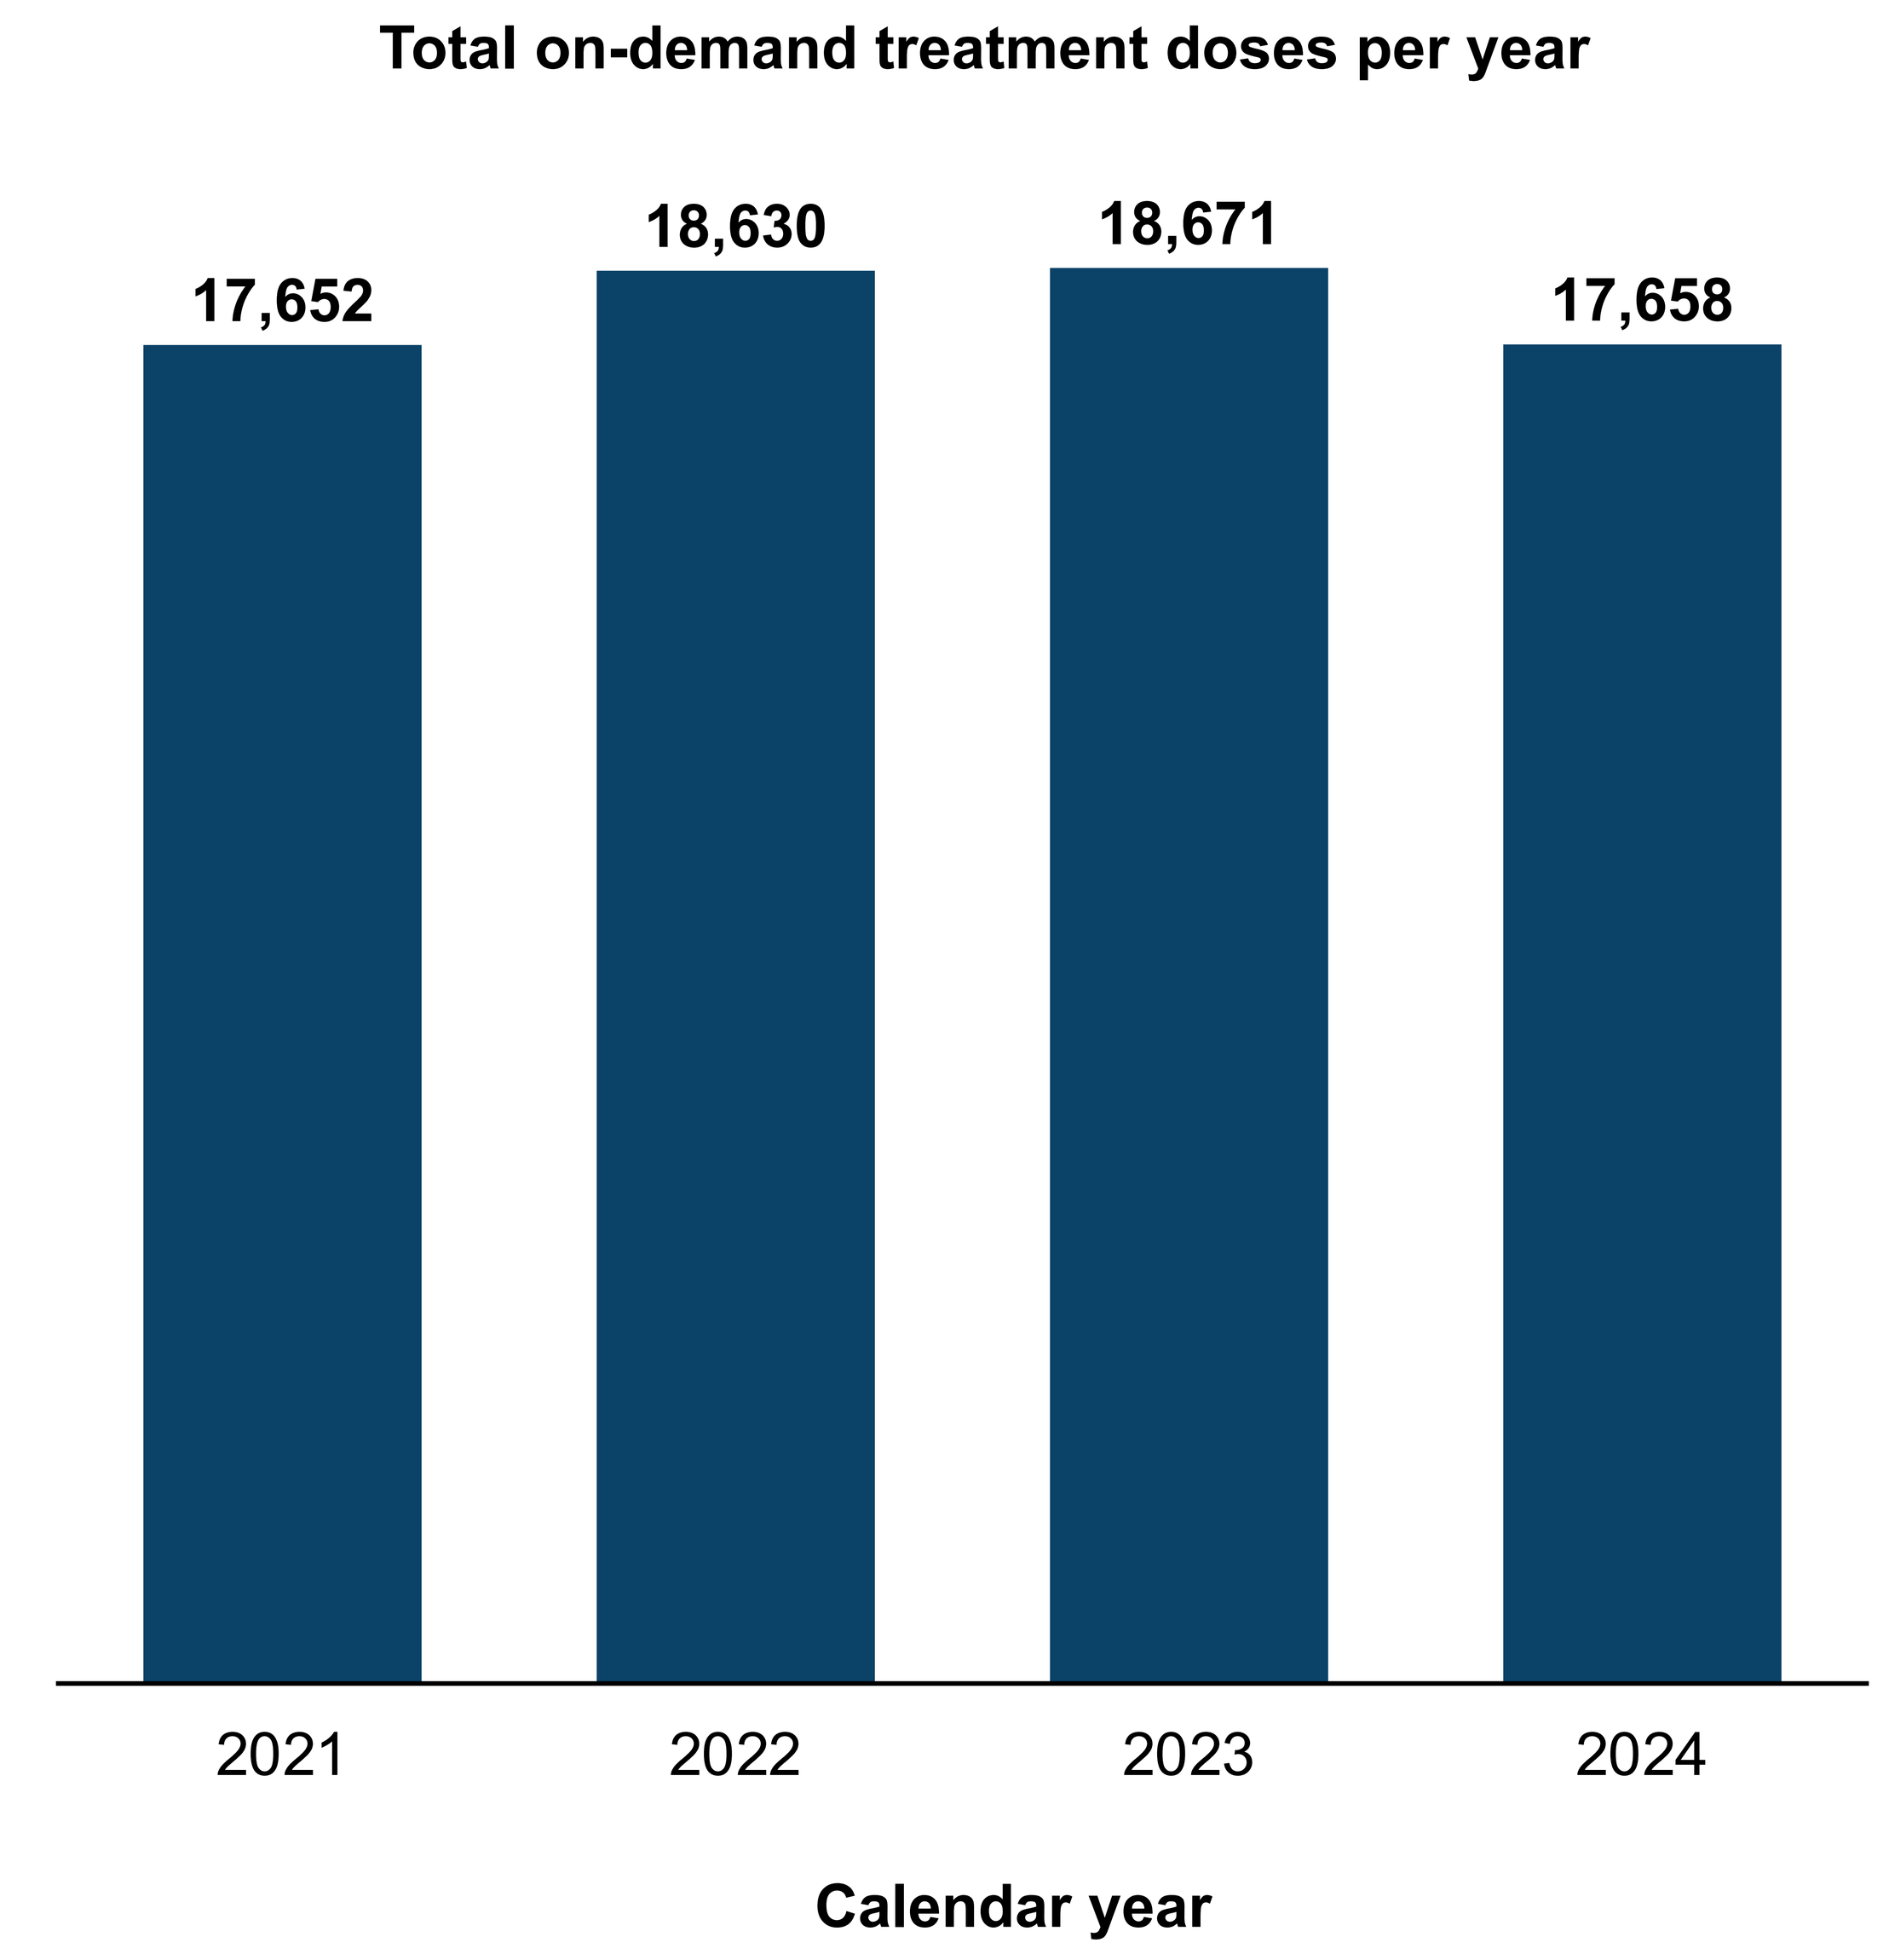
Total on-demand treatment doses per year
17,652
18,630
18,671
17,658
2021
2022
2023
2024
Calendar year

- The average shares of icatibant/Firazyr, recombinant human C1INH, plasma-derived C1INH, and ecallantide during the study period were 72%, 16%, 8%, and 3%, respectively; the share of each agent per calendar year was consistent (**Figure 2**)

Figure 2. Annual share of specific on-demand treatments

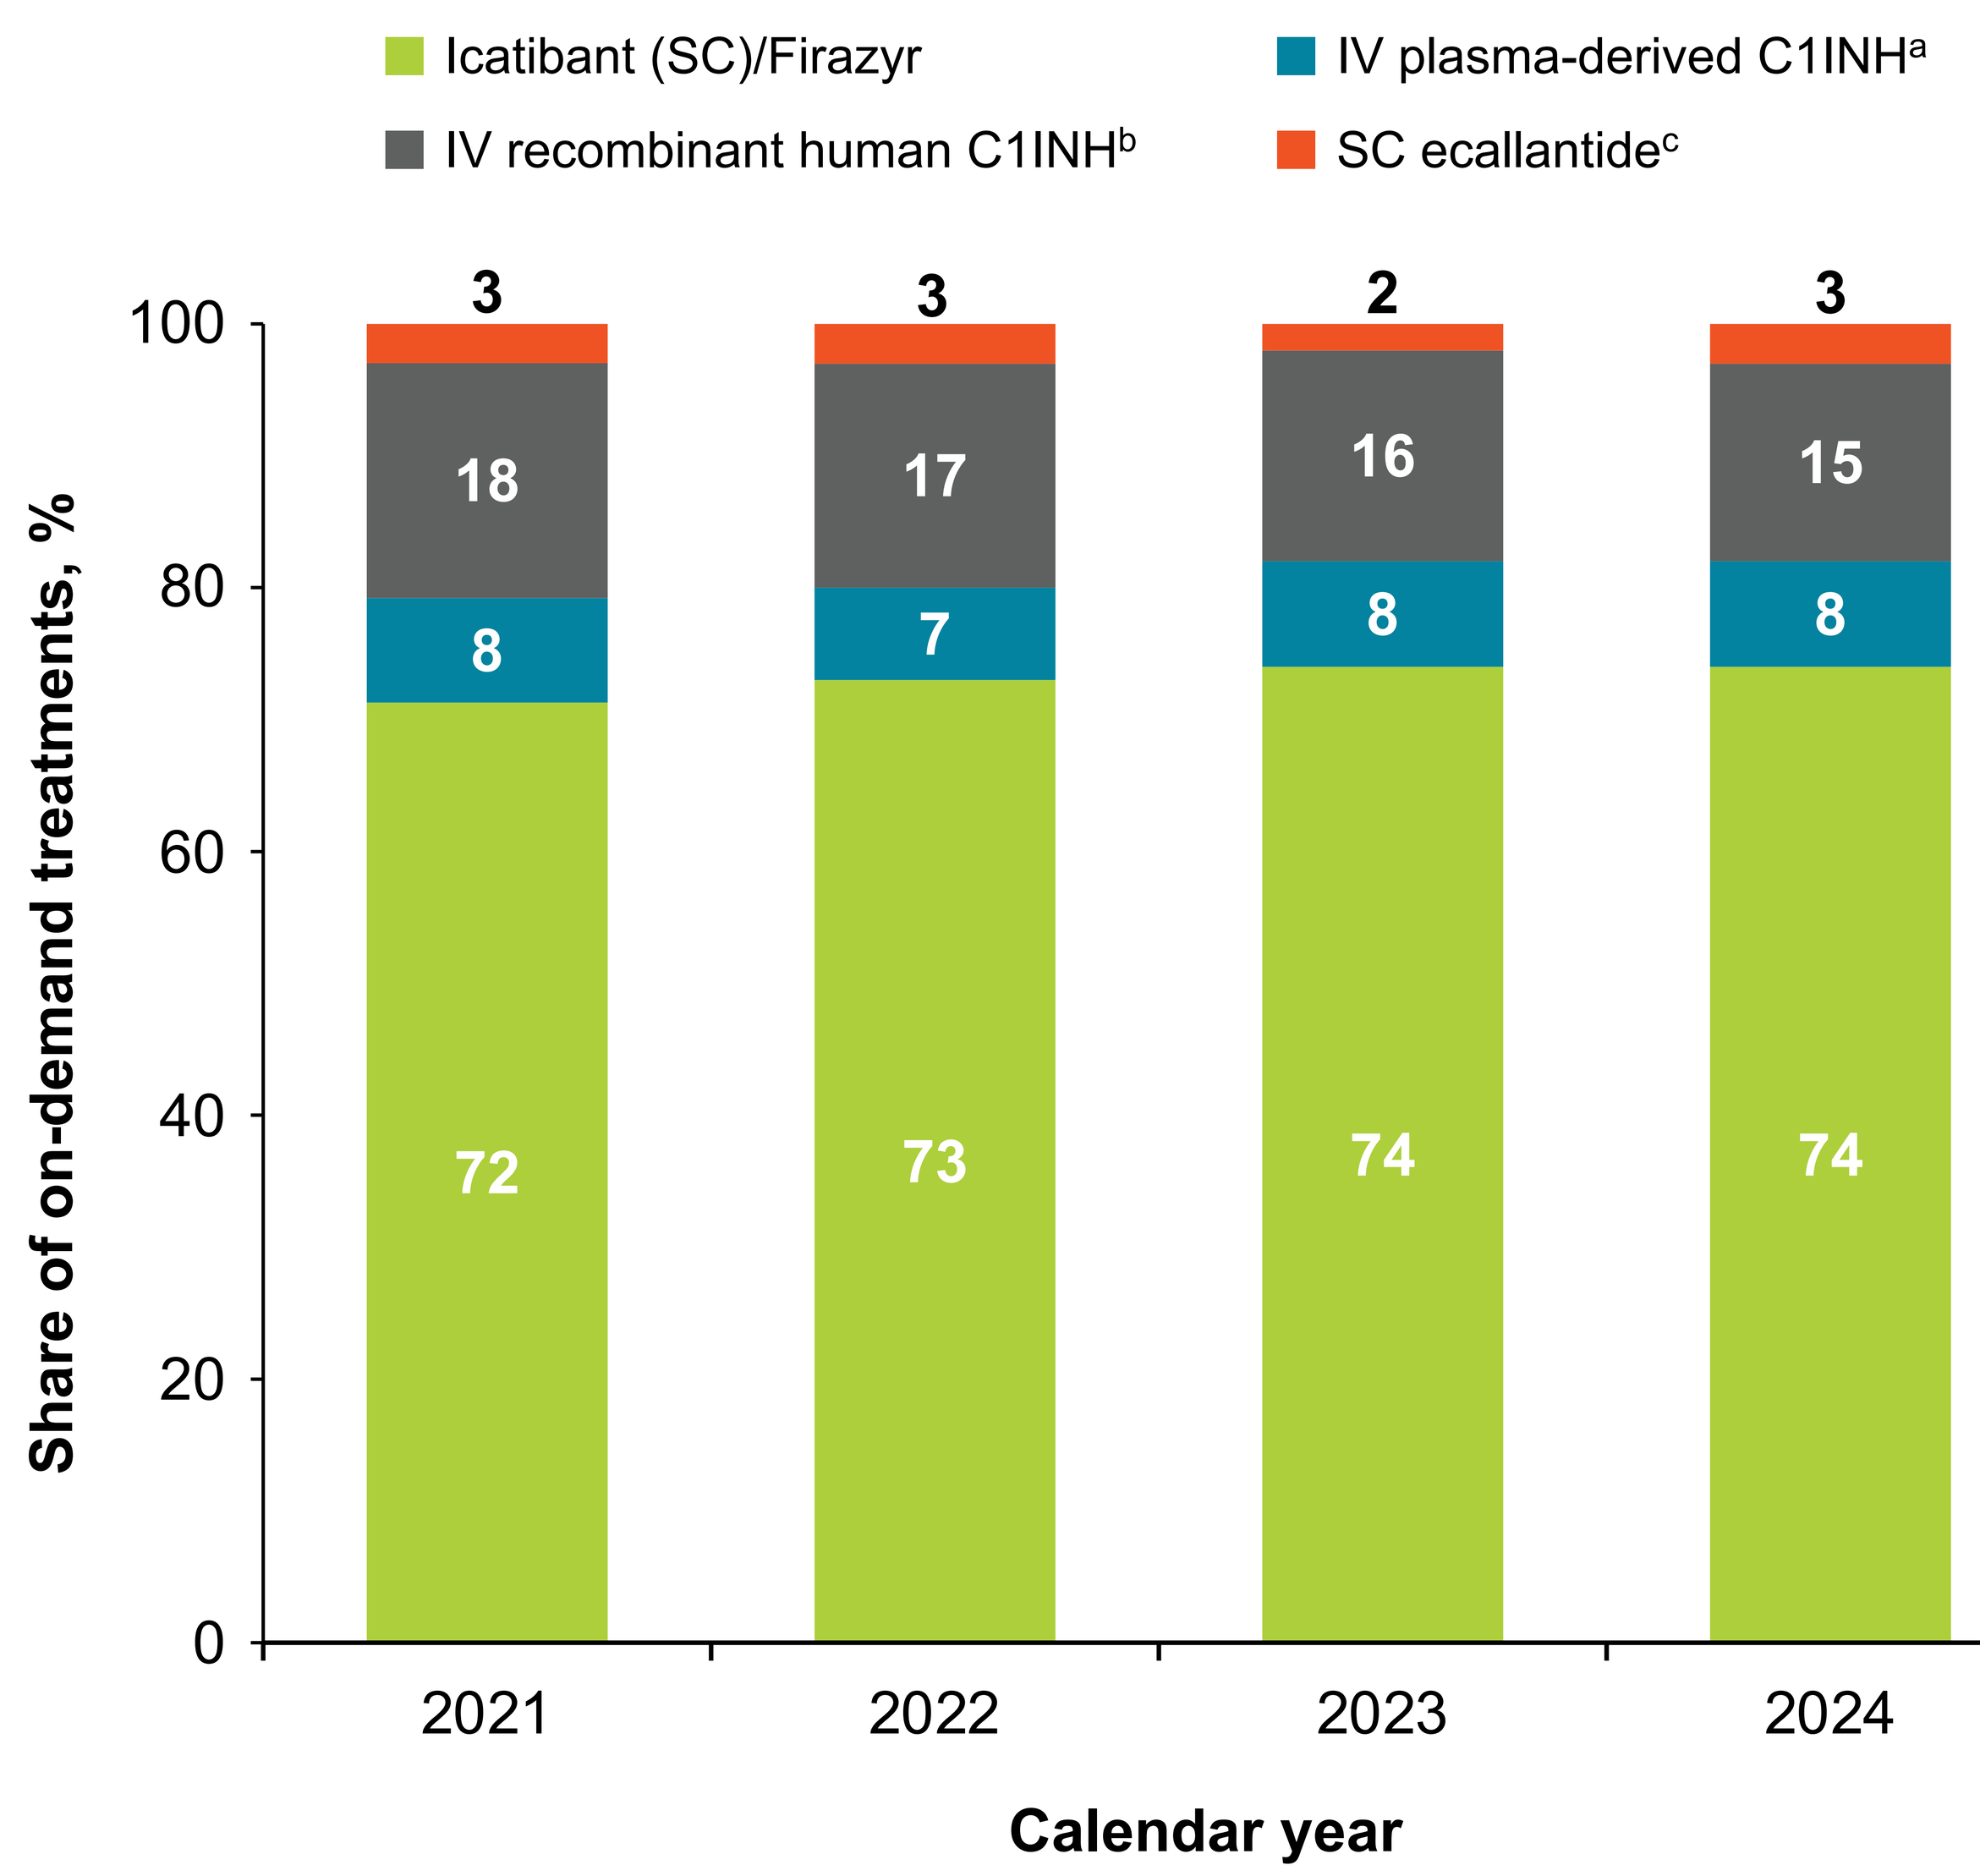
Icatibant (SC)/Firazyr
IV plasma-derived C1INH[a]
IV recombinant human C1INH[b]
SC ecallantide[c]
Share of on-demand treatments, %
0
20
40
60
80
100
3
18
8
72
3
17
7
73
2
16
8
74
3
15
8
74
2021
2022
2023
2024
Calendar year

[a]Berinert. [b]Ruconest. [c]Kalbitor.
C1INH, C1 esterase inhibitor; IV, intravenous; SC, subcutaneous.

- The percentage of patients with ≥1 claim for any LTP treatment increased from 64% in 2021 to 68% in 2024
- Between 2021 and 2024, the use of oral LTP treatment increased by 134%, whereas the use of IV and SC LTP treatments decreased by 71% and 14%, respectively (**Figure 3**)

Figure 3. Indexed LTP treatment volume over time by administration route

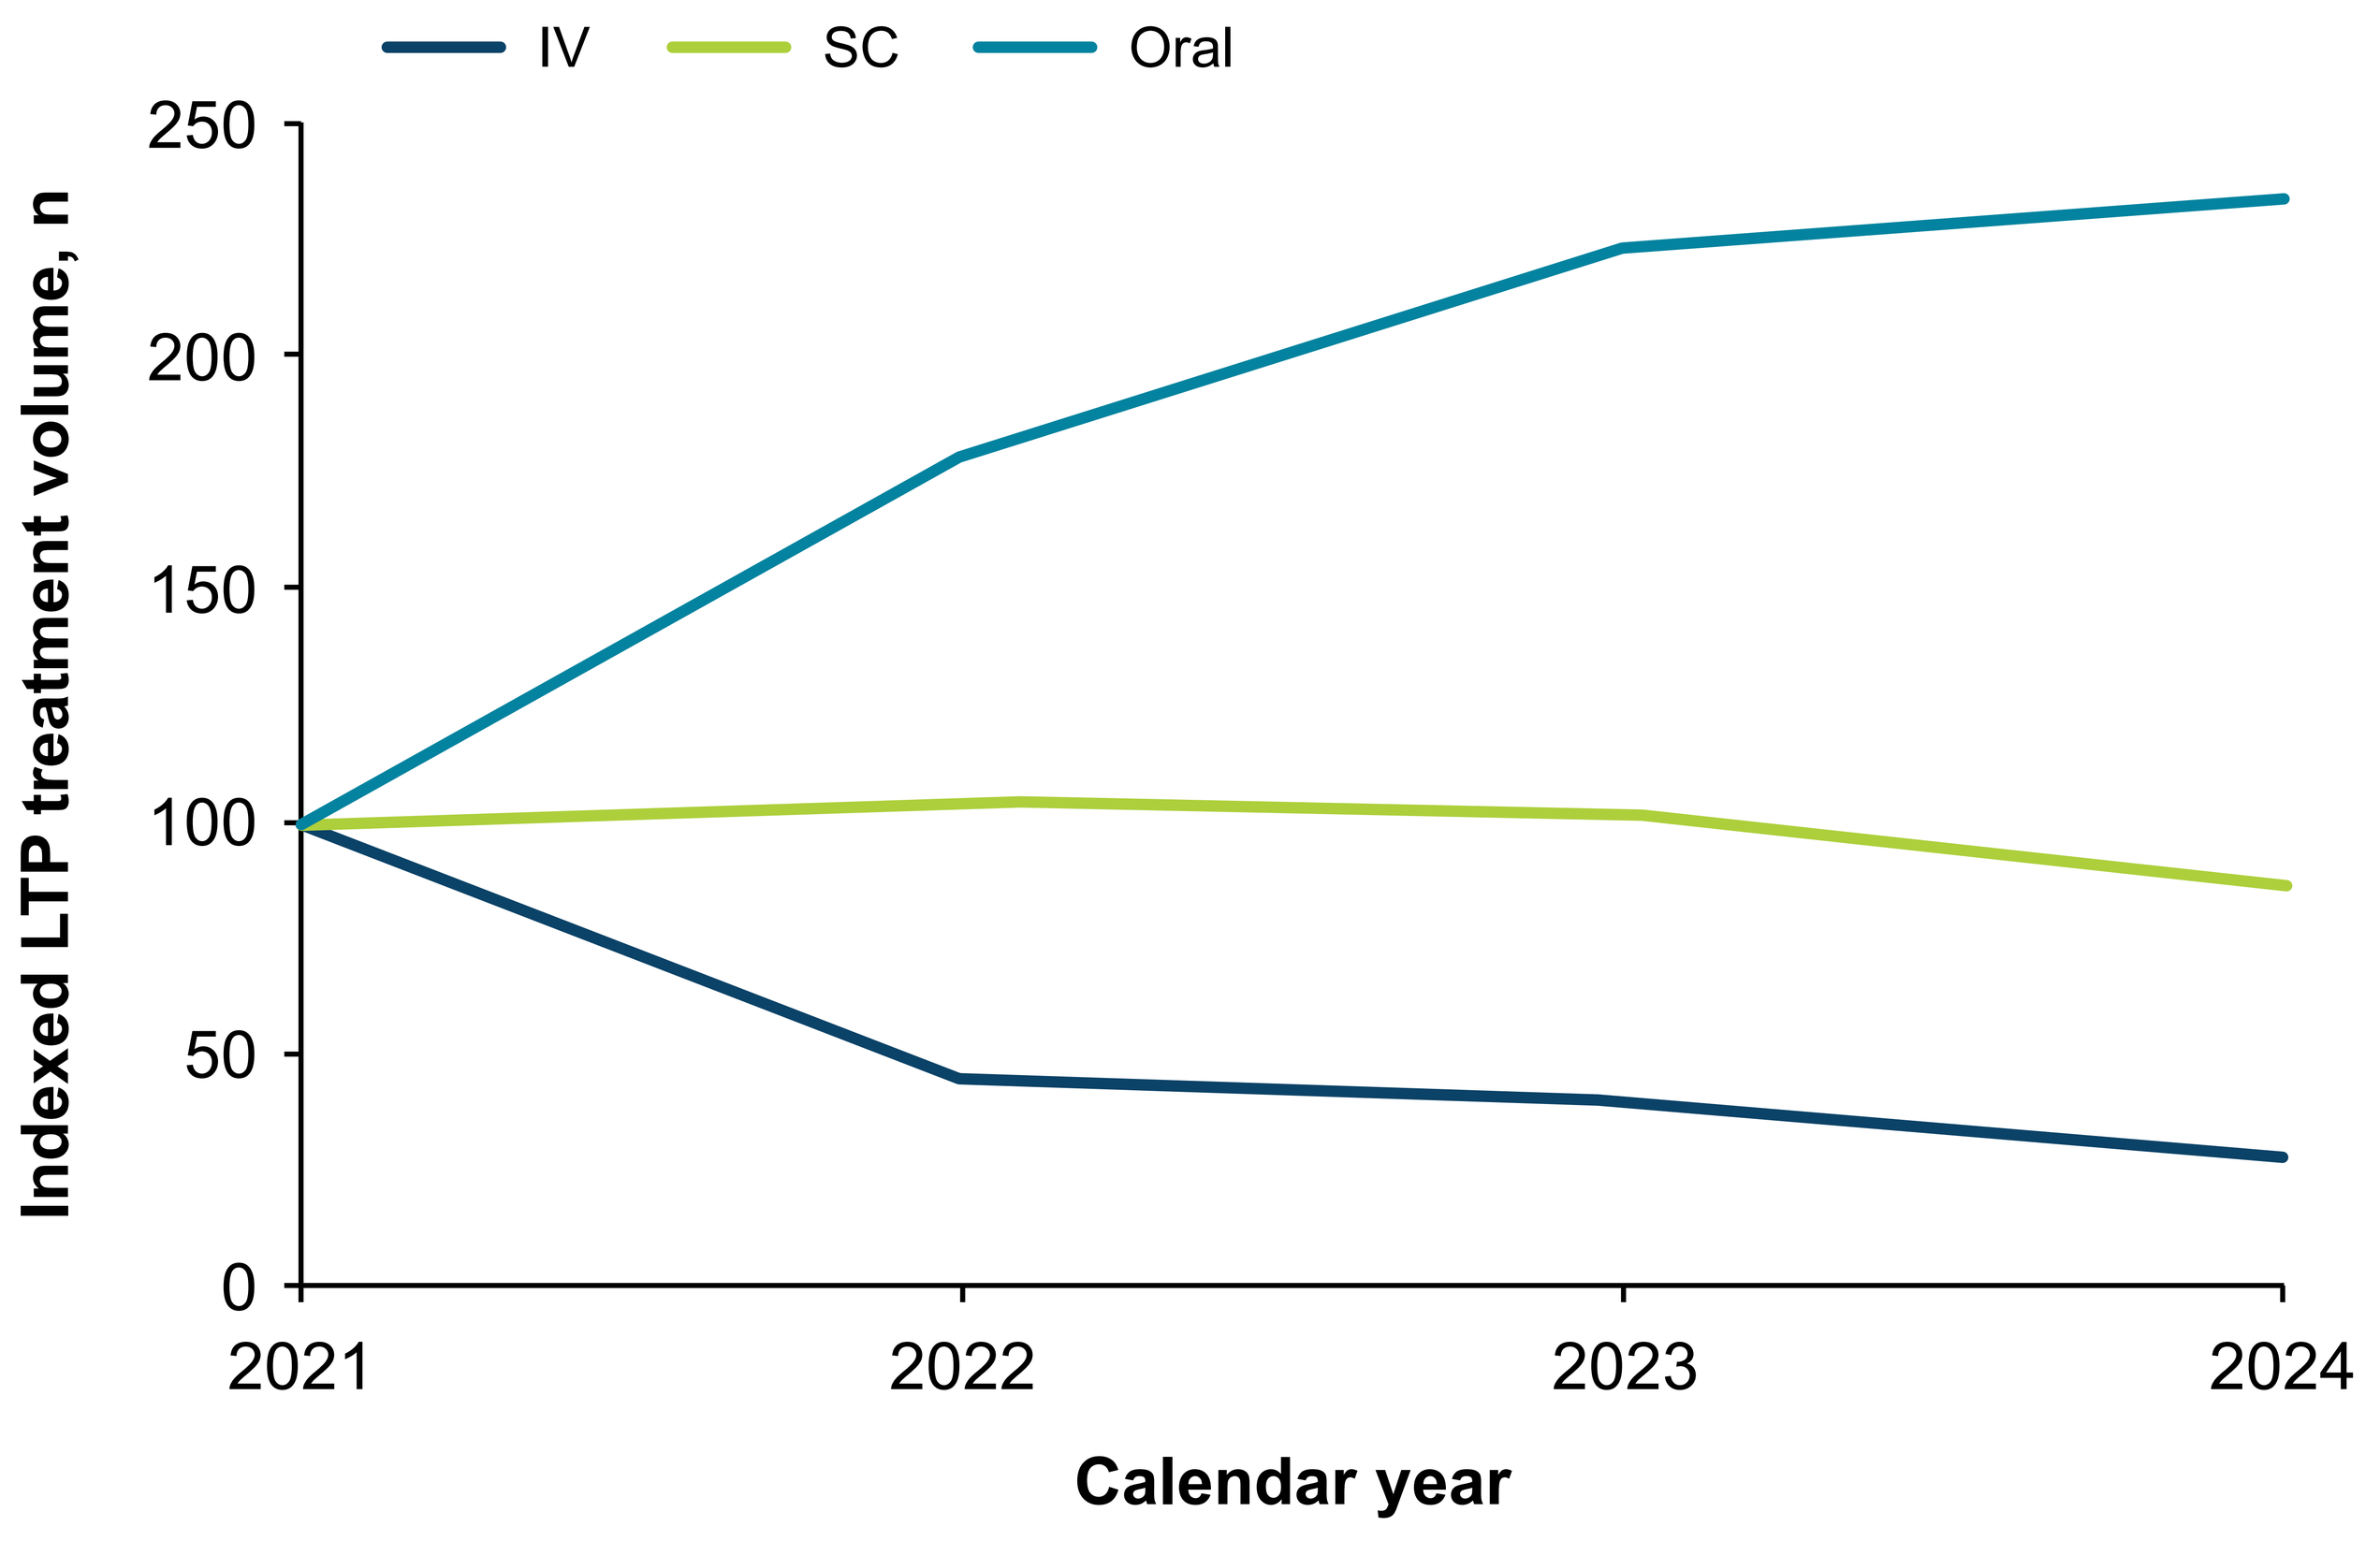
IV
SC
Oral
Indexed LTP treatment volume, n
0
50
100
150
200
250
2021
2022
2023
2024
Calendar year

The total quantity of LTP treatment dispensed was indexed to 100 in 2021 to account for different LTP formulations and dose strengths.
IV, intravenous; LTP, long-term prophylaxis; SC, subcutaneous.

Limitations

- Information on HAE type (ie, type 1, type 2, or HAE-nC1NH) was not available and cannot be attributed to the observed treatment utilization patterns
- Claims data may not represent individuals who are uninsured, enrolled in clinical trials, or participating in patient assistance programs
- Claims data may not reflect actual HAE attack occurrence or treatment use

Conclusions

- **Extrapolating to the overall US population, an average of 83,724 doses of on-demand treatment were estimated during the study period**
- **The mean number of on-demand treatment doses PPPY was 14, and the share of doses during the study period was stable**
- **Overall LTP treatment utilization increased slightly, with significant growth in the use of orally administered LTP treatment offsetting decreases in the use of injectable LTP treatment**
- **Despite growth in LTP treatment in recent years, the overall on-demand treatment quantity dispensed during the study period remained stable, highlighting the continued clinical importance of on-demand treatment in managing HAE**

References

1. Busse PJ, et al. *J Allergy Clin Immunol Pract*. 2021;9(1):132–150.e3.
2. Maurer M, et al. *Allergy*. 2022;77(7):1961–1990.
3. Castaldo AJ, et al. *Ann Allergy Asthma Immunol.* 2025;135(3):303–310.

Disclosures

RT has received grants, consulting fees, honoraria, and/or served on advisory boards and/or data safety monitoring boards for AstraZeneca, Astria Therapeutics, BioCryst, CSL Behring, GSK, Ionis Pharmaceuticals, KalVista Pharmaceuticals, Pharming, Pharvaris, Regeneron, Sanofi, and Takeda. **SC** is a member of the Medical Advisory Board for the US Hereditary Angioedema Association (HAEA) and has participated in advisory boards for BioCryst, CSL Behring, and KalVista Pharmaceuticals. **TJC** served as a speaker/researcher for Astria, CSL Behring, KalVista Pharmaceuticals, and Takeda; a researcher for ADARx Pharmaceuticals, Argo, BioMarin, Intellia Therapeutics, Ionis, and Pharvaris; speaker for Grifols; consultant for ADARx Pharmaceuticals, Astria, BioCryst, CSL Behring, Intellia Therapeutics, KalVista Pharmaceuticals, Pharvaris, and Takeda; Director of the Angioedema Centers of Reference and Excellence (ACARE) International Hereditary Angioedema Center; and member of the Medical Advisory Board for the US HAEA. **DFS** has received grants, consulting fees, and/or honoraria from BioCryst, BioMarin, CSL Behring, KalVista Pharmaceuticals, Pharming, Pharvaris, and Takeda. **MO** has received grants, royalties or licenses, consulting fees, honoraria, payment for expert testimony, meeting/travel support, and/or served on advisory boards and/or data safety monitoring boards for ADMA Biologics, ARS Pharma, AstraZeneca, BioCryst, Blueprint Medicines, Cogent Biosciences, Cycle Pharma, Grifols, GSK, KalVista Pharmaceuticals, Novartis, Pharming, Pharvaris, Regeneron, Sanofi, and Takeda. **MAR** has served as a speaker and/or advisor and/or has received research funding from Astria, BioCryst, BioMarin, Celldex, CSL Behring, Cycle Pharma, Grifols, Intellia Therapeutics, Ionis, KalVista Pharmaceuticals, Pfizer, Pharming, Pharvaris, Sanofi/Regeneron, and Takeda. **AS** received consulting fees from KalVista. **PKA** and **AW** are salaried employees of KalVista Pharmaceuticals.

Acknowledgments

Medical writing and editorial support for the development of this poster, under the direction of the authors, were provided by Tiffany DeSimone, PhD, and Kathleen A. Blake, PhD, of Ashfield MedComms, an Inizio company, and were funded by KalVista Pharmaceuticals, Inc.

Please scan this QR code to view the poster after the presentation

KalVista Pharmaceuticals

Copyright © 2026 KalVista Pharmaceuticals, Inc. All rights reserved.

Presented at the International Society for Pharmacoeconomics and Outcomes Research (ISPOR) 2026 Annual Meeting; May 17–20, 2026; Philadelphia, PA, USA.