Use of Sebetralstat as Short-term Prophylaxis for Patients With Hereditary Angioedema: Results From the KONFIDENT-S Study

Maeve O'Connor[1,2], **Avraham Beigelman**[3], **Mauro Cancian**[4], **Miloš Jeseňák**[5,6], **Anthony Jordan**[7], **Sorena Kiani-Alikhan**[8], **Tamar Kinaciyan**[9], **Evangelia Kompoti**[10], **Marcin Stobiecki**[11], **James Hao**[12], **Matthew Iverson**[12], **Andrea Zanichelli**[13,14]

[1]Allergy, Asthma & Immunology Research Institute, Charlotte, NC, USA; [2]Integrative Allergy & Immunology Care, LLC; [3]Kipper Institute of Allergy and Immunology, Schneider Children's Medical Center of Israel, Tel Aviv University, Tel Aviv, Israel; [4]Departmental Allergy Division, Department of Systems Medicine, University of Padua, Padua, Italy; [5]National Centre for Hereditary Angioedema - Department of Paediatrics and Adolescent Medicine and Department of Pulmonology and Phthisiology, Jessenius Faculty of Medicine in Bratislava, Comenius University in Bratislava, University Hospital, Martin, Slovakia; [6]Institute of Clinical Immunology and Medical Genetics, Jessenius Faculty of Medicine in Bratislava, Comenius University in Bratislava, University Hospital, Martin, Slovakia; [7]Department of Clinical Immunology and Allergy, Auckland City Hospital, Te Toka Tumai, Auckland, New Zealand; [8]Royal Free London NHS Foundation Trust, Department of Immunology, London, UK; [9]Department of Dermatology, ACARE and Hereditary Angioedema Center Vienna and Burgenland, Medical University of Vienna, Vienna, Austria; [10]Former Head of the Department of Allergology-Clinical Immunology, Laikon General Hospital of Athens, Athens, Greece; [11]Department of Clinical and Environmental Allergology, Jagiellonian University, Medical College, Krakow, Poland; [12]KalVista Pharmaceuticals, a company of the Chiesi Group, Framingham, MA, USA; [13]Dipartimento di Scienze Biomediche per la salute, University of Milan, Milan, Italy; [14]Operative Unit of Medicine, Angioedema Center, IRCCS Policlinico San Donato, San Donato Milanese, Milan, Italy

Key Takeaways

97.6% **Of Procedures Without HAE Attack**
when ≥2 600mg doses of oral sebetralstat were used for short-term prophylaxis (STP)

0 **Treatment-Related AEs**
with oral sebetralstat for STP (44 procedures across 16 patients)

These findings support the continued evaluation of sebetralstat as an alternative STP regimen

Background

- International hereditary angioedema (HAE) guidelines recommend STP with intravenous plasma-derived C1 inhibitor (IV pdC1INH) before medical or dental procedures to reduce the risk of perioperative angioedema attacks[1]
- IV pdC1INH products are approved for STP in the EU,[2] but no treatment options are indicated for STP in the US
- The use of STP is heterogenous and inconsistent,[3] likely due to the burden of IV administration, product availability, and procedure coordination[1,4]
- Sebetralstat has recently been approved in the US, Europe, Australia, Singapore, and Japan as the first oral on-demand treatment for patients with HAE[5-8]
- KONFIDENT-S, a 2-year open-label extension (NCT05505916), investigated the long-term effectiveness and safety of on-demand sebetralstat.[9] As part of this study, the potential of sebetralstat as an alternative STP regimen was also evaluated

Objective

- Evaluate the effectiveness of sebetralstat for STP of HAE attacks triggered by medical and dental procedures among KONFIDENT-S participants

Methods

- The KONFIDENT-S trial population included patients ≥12 years of age with HAE-C1INH[9]
- Patients scheduled to undergo a medical or dental procedure were instructed to self-administer 3 doses of sebetralstat 600 mg as STP (**Figure 1**)
 - Participants using long-term prophylaxis (LTP) were excluded from the STP evaluation
 - The dosing interval was based on pharmacokinetic data with the goal of providing 24 hours of coverage
- Treatment effectiveness was assessed by the number of HAE attacks within 24 hours of procedure start
- Safety was assessed using adverse event (AE) reporting

Figure 1. Sebetralstat dosing schedule for STP

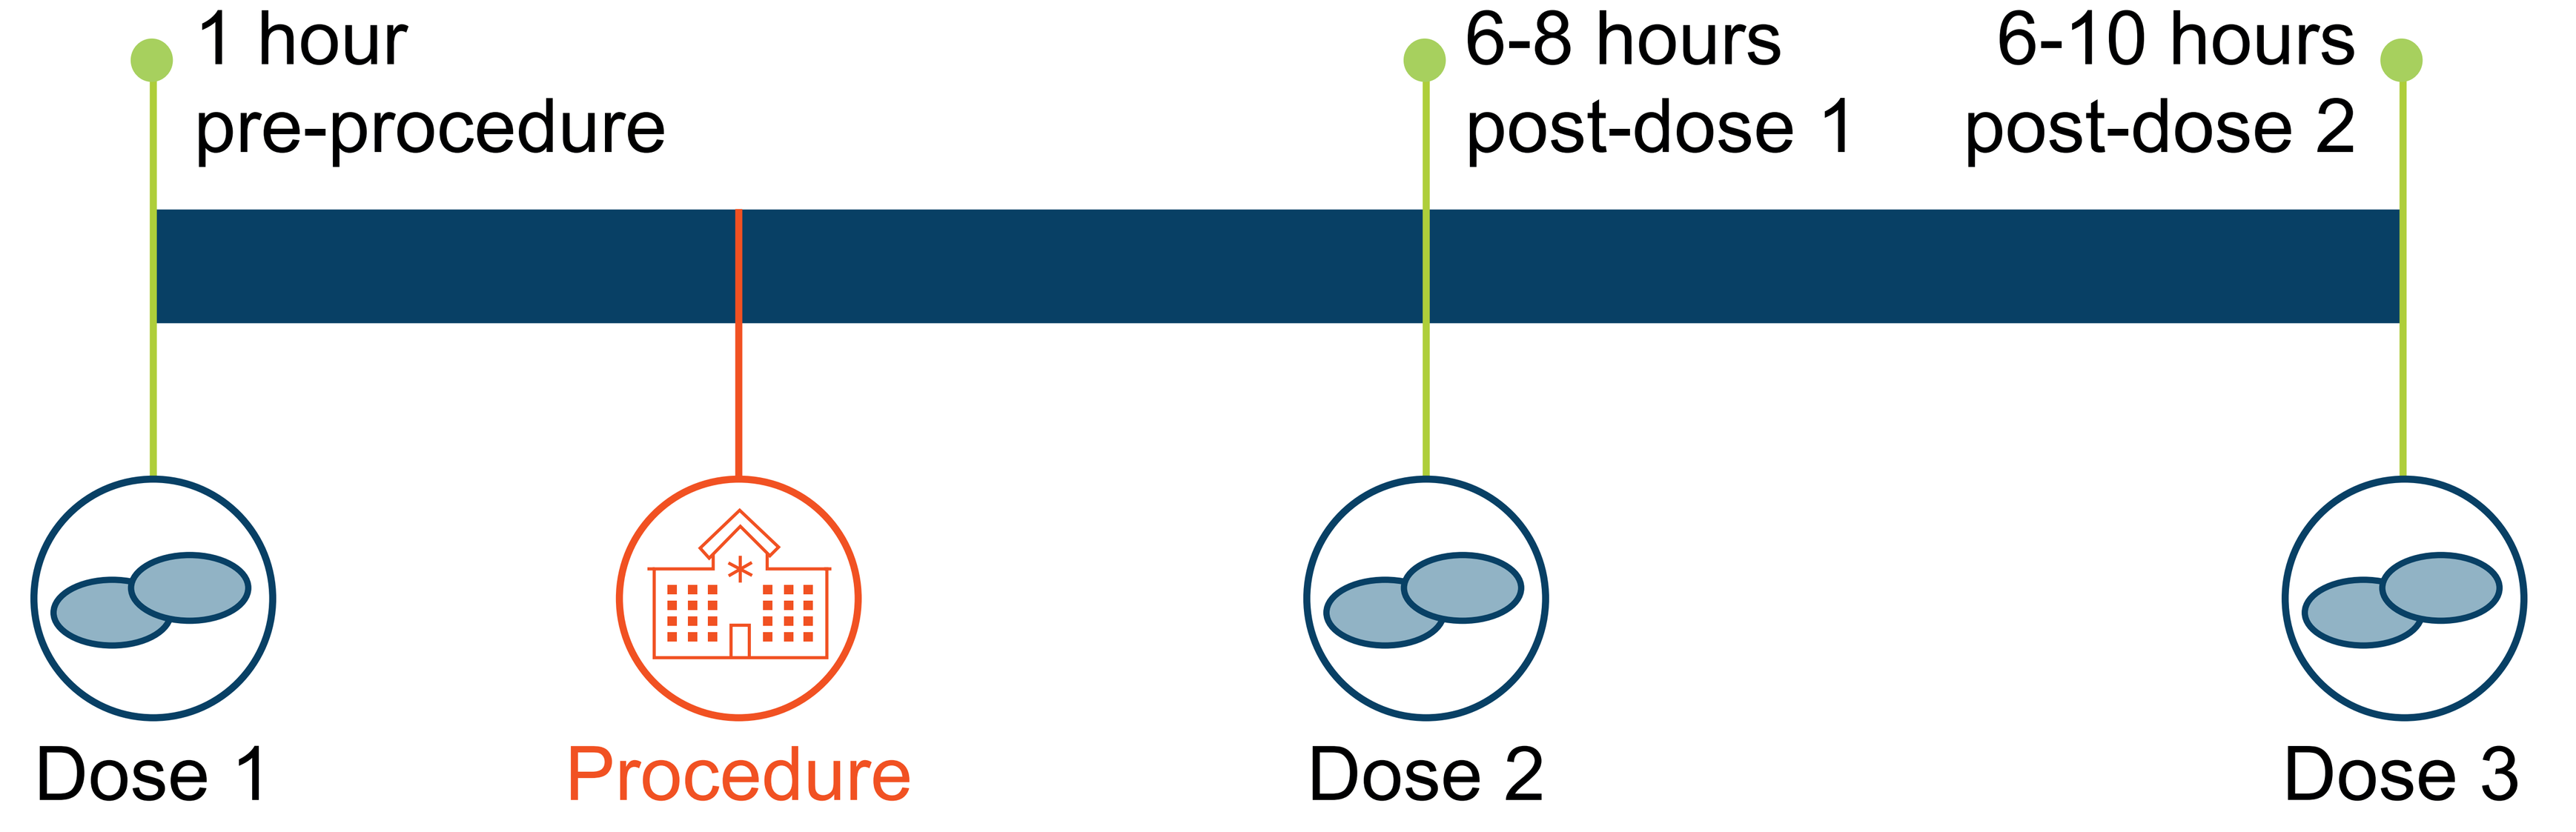
1 hour
pre-procedure
6-8 hours
post-dose 1
6-10 hours
post-dose 2
Dose 1
Procedure
Dose 2
Dose 3

Results

Participant Demographics and Characteristics

- At data cutoff (Sept 30, 2025), 16 of 136 patients (11.8%) in KONFIDENT-S had used sebetralstat as STP for medical or dental procedures (**Table 1**)

Table 1. Demographics and clinical characteristics of the STP population (N=16)

Age, mean (range), years	34.6 (16–58)
Age-group, n (%)	
<18 years	2 (12.5)
≥18 years	14 (87.5)
Females, n (%)	10 (62.5)
Race, n (%)	
White	11 (68.8)
Asian	5 (31.3)
Type 1 HAE-C1INH, n (%)	16 (100)
Family history of HAE-C1INH, n (%)	12 (75)
Years of diagnosis at screening, mean (range)	13.0 (0–34)

HAE-C1INH, hereditary angioedema with deficiency of functional C1-esterase inhibitor.

Figure 2. Sebetralstat STP use by procedure (N=44)[a]

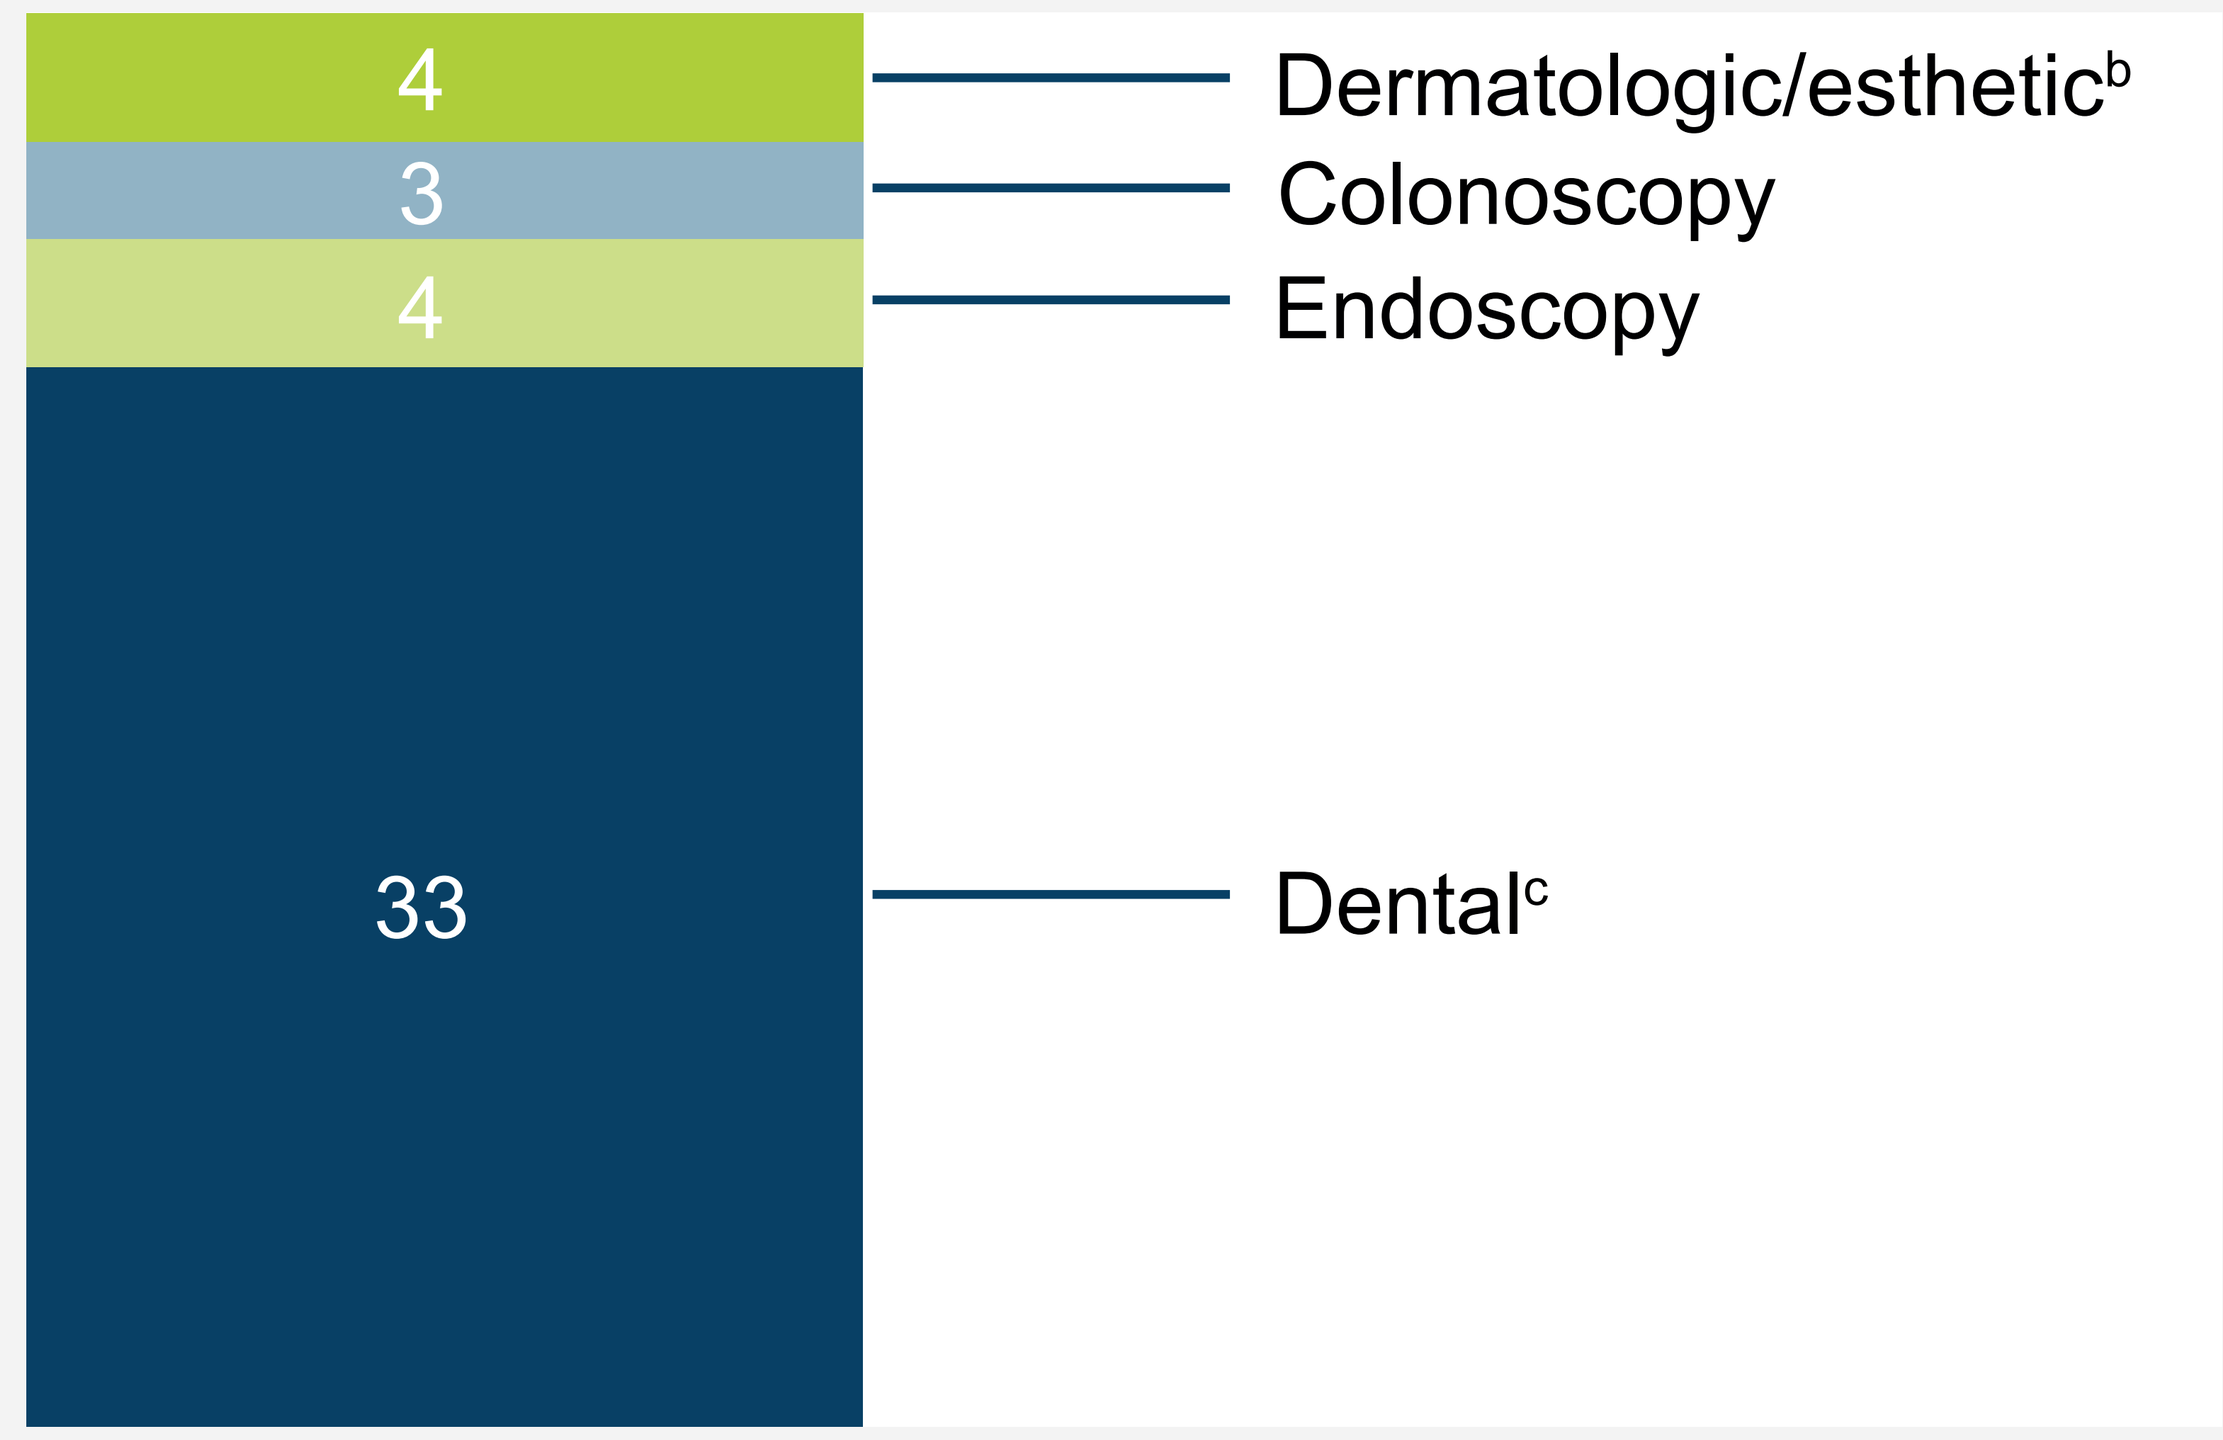
4
Dermatologic/esthetic[b]
3
Colonoscopy
4
Endoscopy
33
Dental[c]

[a]In one case, STP was used to cover 2 procedures (colonoscopy and gastroscopy).
[b]Dermal filler injection, lipoma excision, papilloma excision, and eyebrow microblading.
[c]Dental procedures (e.g., filling, root canal, extraction): 27; Dental care (other): 6.

Figure 3. Sebetralstat STP doses taken

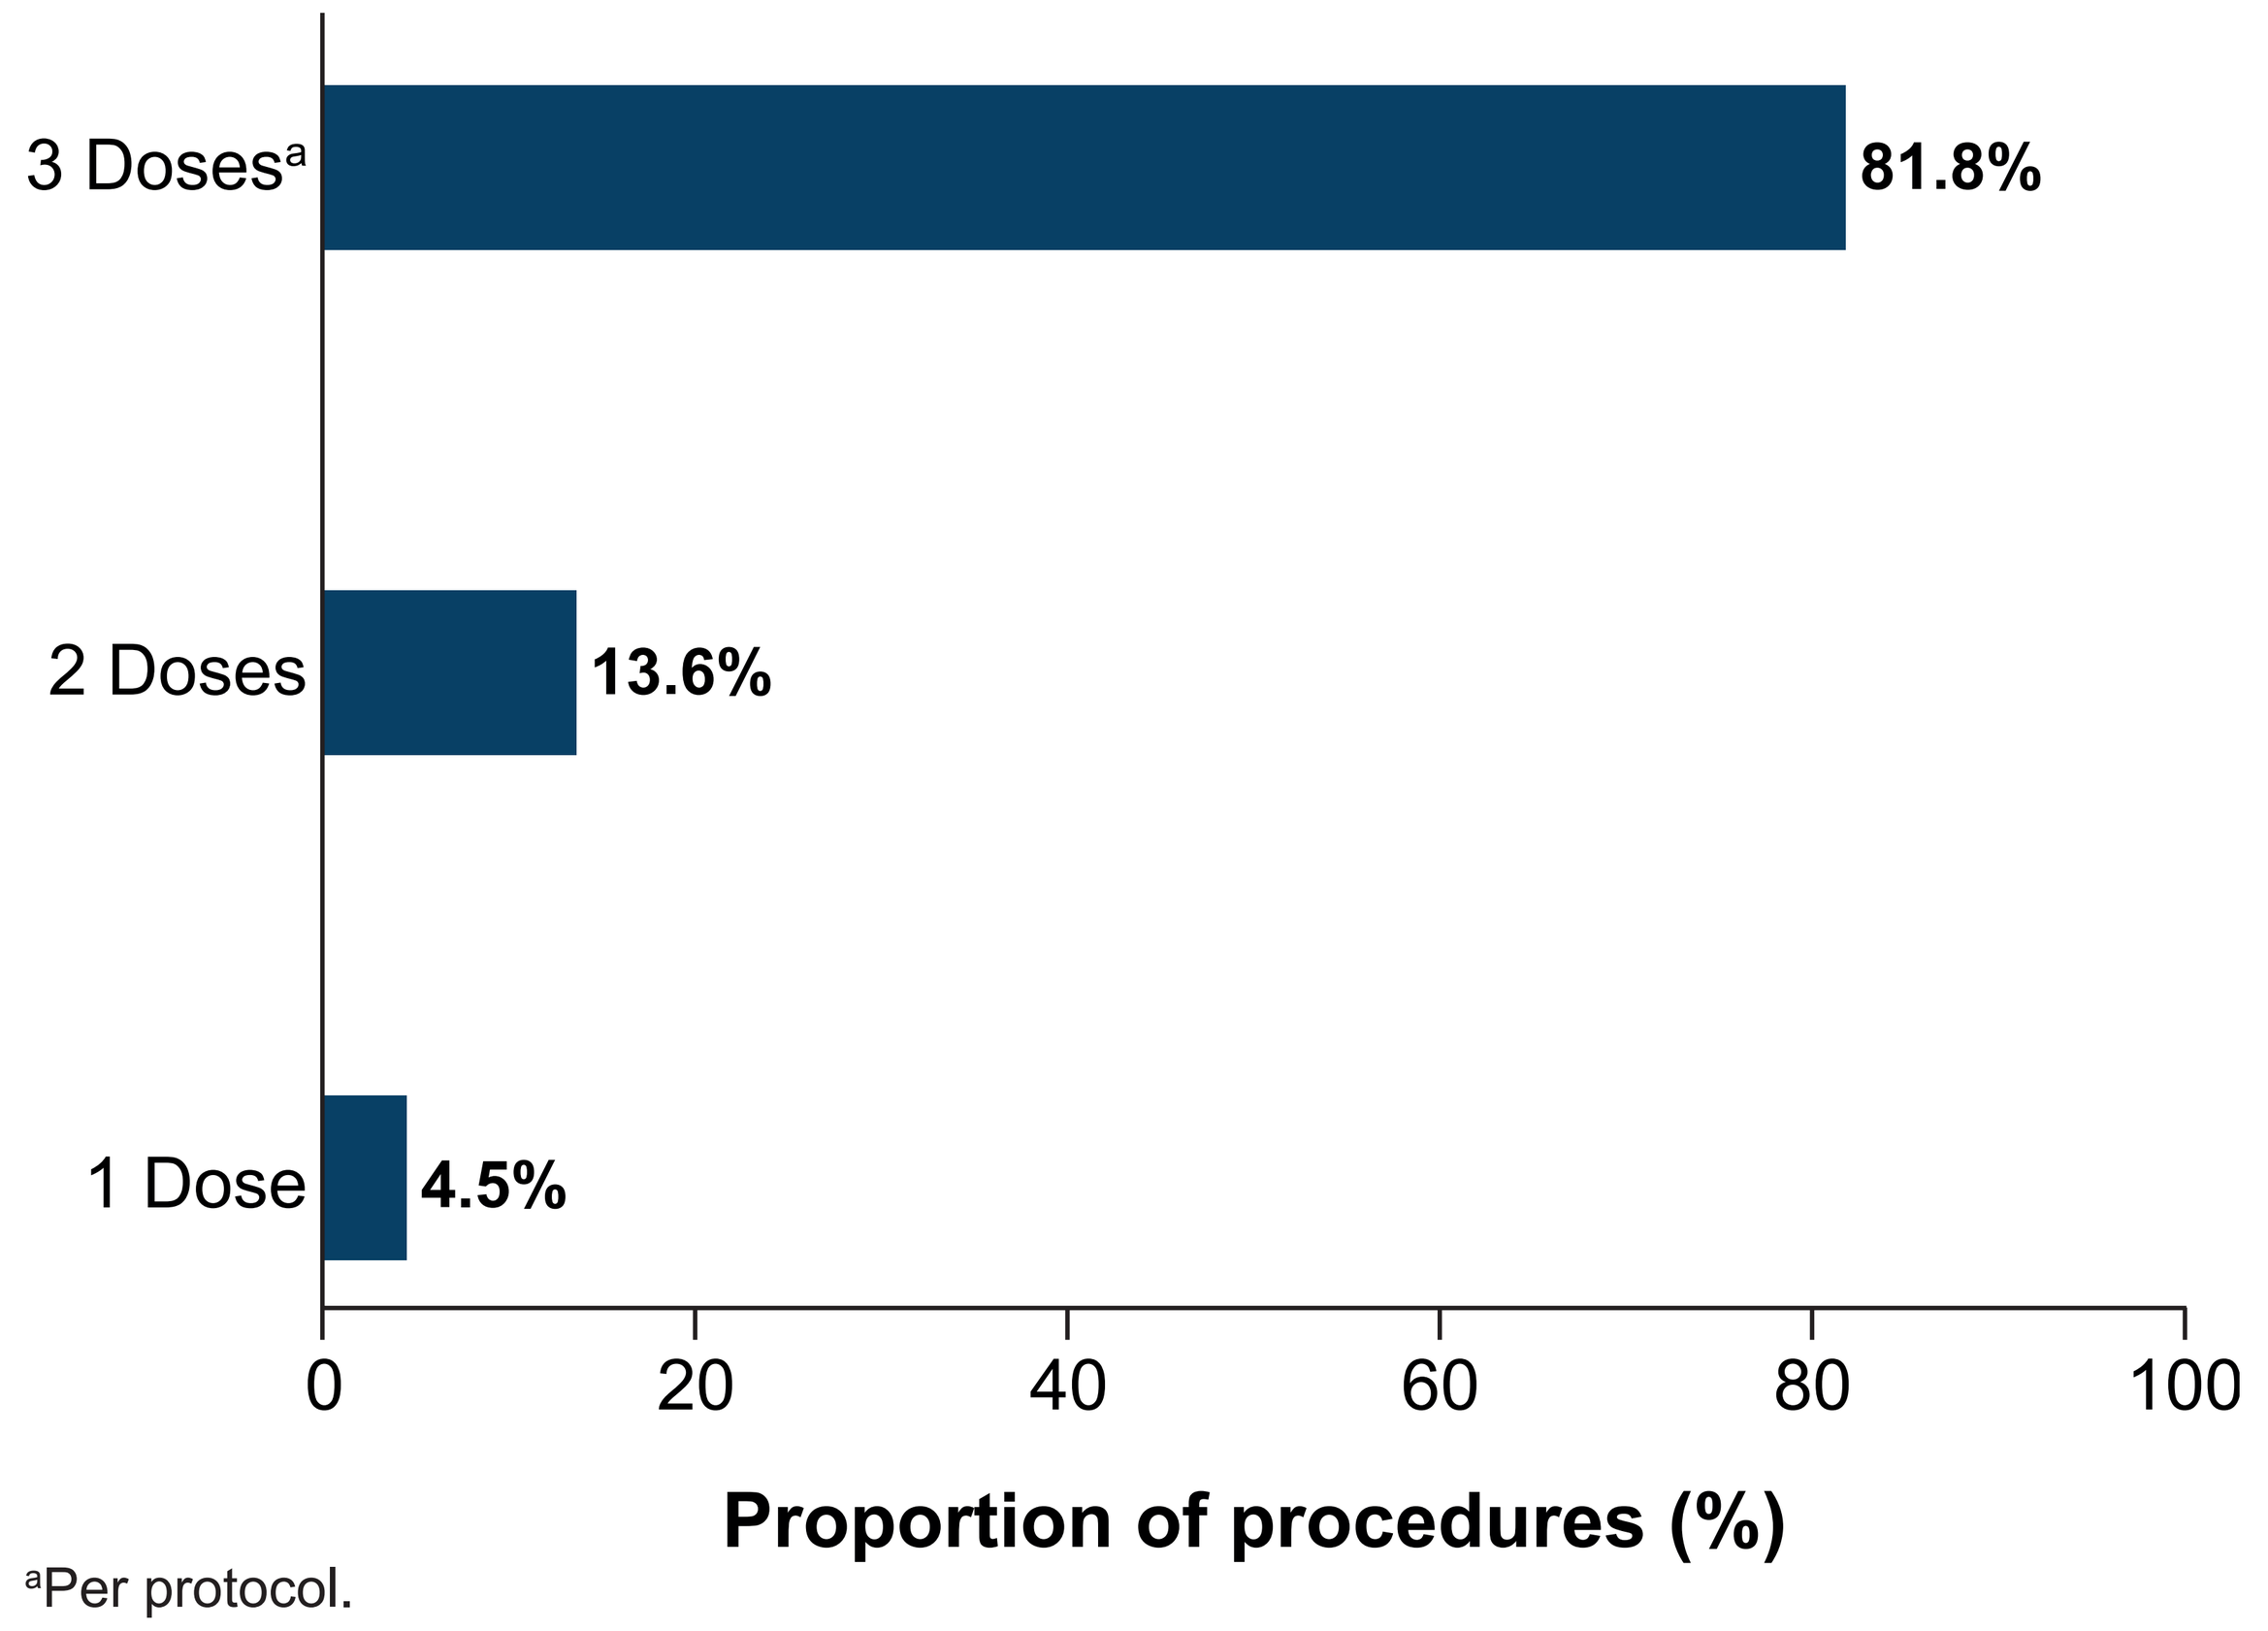
3 Doses[a]
81.8%
2 Doses
13.6%
1 Dose
4.5%
0
20
40
60
80
100
Proportion of procedures (%)

[a]Per protocol.

STP Effectiveness Within 24 Hours

- For the 42 procedures in which 2 or 3 scheduled doses of oral sebetralstat STP were administered, 1 HAE attack (2.3%) occurred following dermal filler injection mild severity) (**Figure 4**)
- For the 2 procedures in which only the first, pre-procedural dose of oral sebetralstat STP was administered, 1 HAE attack was reported following a colonoscopy (attack severity and location unknown)

Figure 4. HAE attack incidence within 24 hours of the procedure

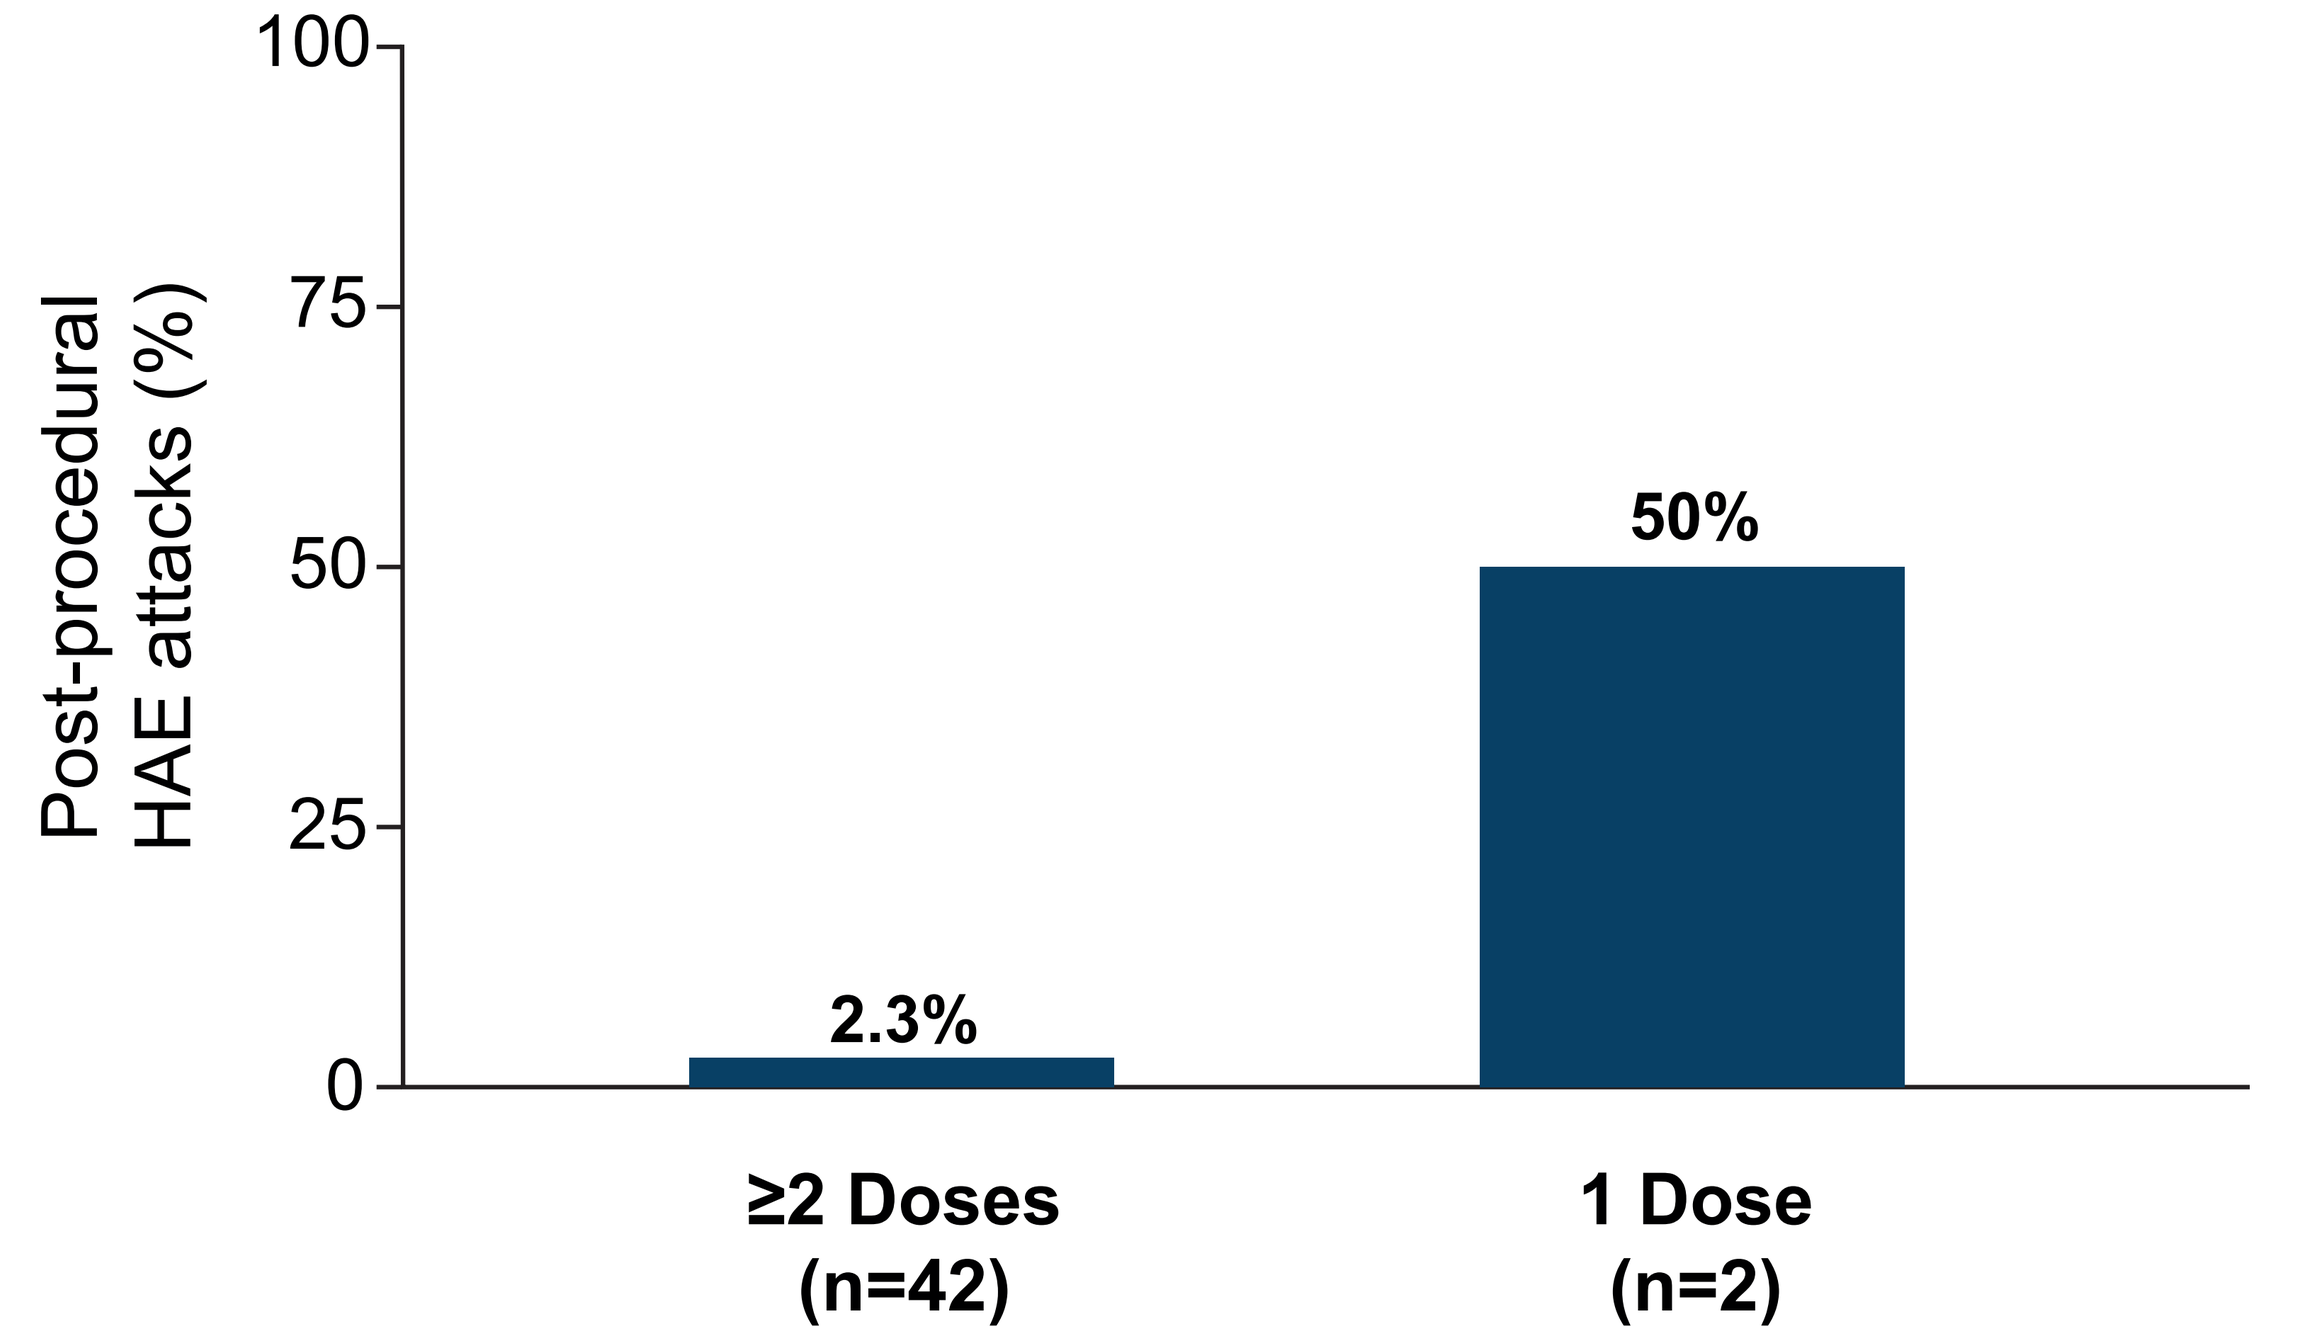
Post-procedural
HAE attacks (%)
100
75
50
25
0
2.3%
50%
≥2 Doses
(n=42)
1 Dose
(n=2)

- Of the 44 total routine medical and dental procedures performed during which sebetralstat was used as STP, 2 (4.5%) procedures were accompanied by an HAE attack within 24 hours

Safety

- Five AEs were reported in 3 patients within 72 hours of administering sebetralstat as STP (**Figure 5**)

Figure 5. Adverse events

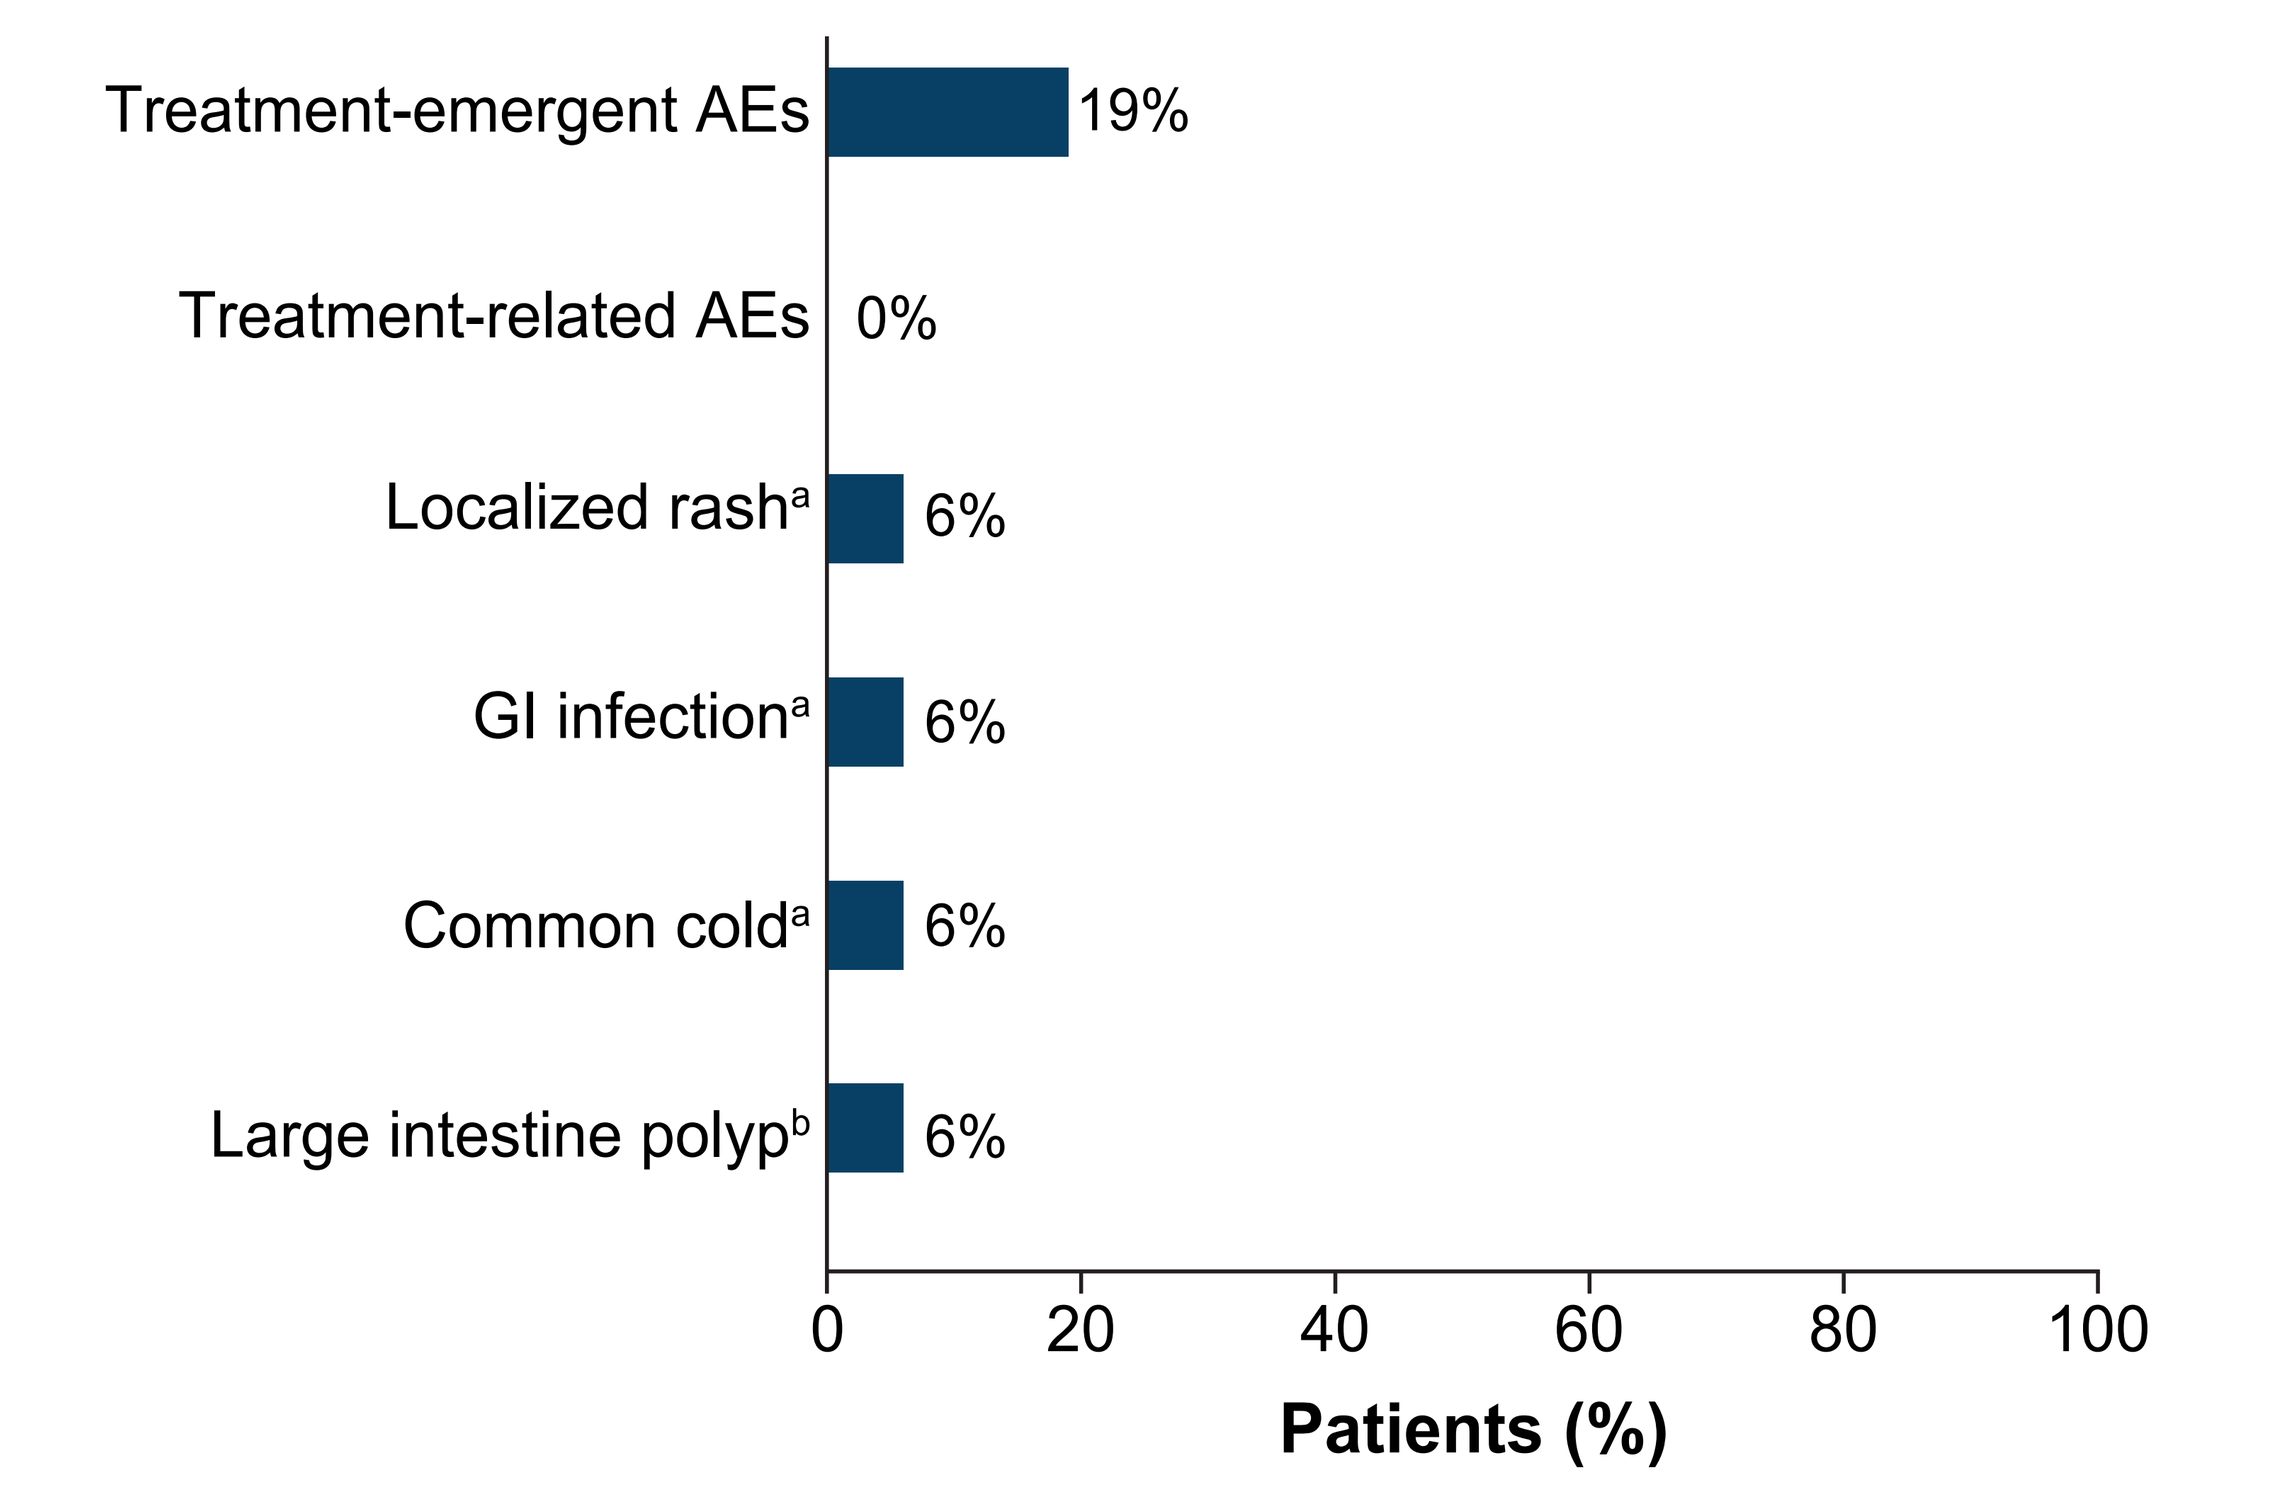
Treatment-emergent AEs
19%
Treatment-related AEs
0%
Localized rash[a]
6%
GI infection[a]
6%
Common cold[a]
6%
Large intestine polyp[b]
6%
0
20
40
60
80
100
Patients (%)

[a]With the use of 3 doses of sebetralstat.
[b]After a single dose of sebetralstat.
AE, adverse event; GI, gastrointestinal.

Limitations to this Study

- Few highly invasive procedures included
- No control arm/comparator
- Limited participant medical history

References

1. Betschel S, et al. *Allergy Asthma Clin Immunol*. 2026;22:24.
2. Martinez-Saguer I, et al. *Clin Transl Allergy*. 2025;15(12):e70126.
3. Nanda MK, et al. *Ann Allergy Asthma Immunol*. 2014;113(2): 198-203.
4. Holmes A, et al. *Front Immunol*. 2024;15:1413547.
5. Ekterly (sebetralstat). Package Insert. KalVista Pharmaceuticals; 2025.
6. Ekterly (sebetralstat). Summary of Product Characteristics (UK). KalVista Pharmaceuticals; 2025.
7. Ekterly (sebetralstat). Summary of Product Characteristics (EU). KalVista Pharmaceuticals; 2025.
8. Ekterly (sebetralstat). Package Insert (Japan). KalVista Pharmaceuticals; 2025.
9. Farkas H, et al. *J Allergy Clin Immunol Pract*. 2025;13:3094-3103.

Acknowledgments

Medical writing support was provided by Josh Rodman, PhD, of ApotheCom, San Francisco, CA, and funded by KalVista Pharmaceuticals, a company of the Chiesi Group.

Disclosures

Maeve O'Connor: is a speaker/consultant/advisor or researcher for AbbVie, AZ, Blueprint, CSL Behring, Grifols, GSK, KalVista, Pharming, Sanofi, and Teva; and Chief Medical Officer of the CIIC.

Presented at the Consortium of Independent Immunology Clinics (CIIC) Fall Conference; October 2-4, 2026 ; Frisco, TX

Copyright © 2026 KalVista Pharmaceuticals, a company of the Chiesi Group. All rights reserved.